Therapeutic Effect of Peramivir (S-021812, BCX-1812) after Single Intravenous Treatment of Mice Infected with Influenza A and B Viruses

V-1065

R. YOSHIDA, M. KODAMA, M. KOBAYASHI, M. KITANO, A. SATO AND Y. YAMANO

Shionogi & Co., Ltd., Osaka, JAPAN

SHIONOGI & CO., LTD.
Institute for Medical Science
2-5-1 Mishima, Settsu-shi
Osaka, Japan 566-0022
Phone: +81-6-6382-0924
Fax: +81-6-6382-2598
ryu.yoshida@shionogi.co.jp

ABSTRACT

Background
Peramivir is a novel inhibitor of influenza neuraminidase (NA). The therapeutic effect of peramivir and other anti-influenza drugs was investigated using a mouse model.

Methods
NA inhibitory activity was monitored using fluorescence assay. The inhibitory activity against viral replication was examined using MDCK cells. The therapeutic effects of peramivir were investigated using the virus-infected mouse model and compared with that of oseltamivir.

Results
Peramivir showed strong *in vitro* inhibitory activity against NA activity and virus replication of human influenza A viruses of various subtypes and B viruses. In the lethal mouse model infected with human influenza A virus of subtype H1N1 or H3N2, peramivir administered as a single (qd for 1 day) intravenous (iv) dosing increased the number of surviving mice in a dose-dependent manner and led to almost 80 - 100% survival at 3 mg/kg against the tested virus strains. In contrast, no significant improvement in survival was observed after oral dose of oseltamivir phosphate (qd for 1 day) at 10 mg/kg. Twice-daily oral administration for 5 days (bid for 5 days) and more than 50 mg/kg as a total dose was required for oseltamivir phosphate to achieve almost 100% survival. Similar results were obtained with the human influenza B virus-infected mouse model. Peramivir iv administration (qd for 1 day) showed significant improvement in survival at less than 3 mg/kg. Oral administration bid for 5 days and 5 - 50 mg/kg as a total dose was required for oseltamivir phosphate to show significant improvement in survival.

Conclusions
These data demonstrate that peramivir has strong *in vitro* inhibitory activity against human influenza A and B viruses, and that peramivir iv treatment (qd for 1 day) has a significant therapeutic effect in the lethal mouse model, which is superior to that of oseltamivir phosphate.

INTRODUCTION

- Peramivir is a novel inhibitor of influenza neuraminidase (NA) and shows strong affinity to influenza neuraminidase and a slow off-rate (S. Bantia et al. Antiviral Research 2006).
- This suggests that peramivir can inhibit NA activity for a long period and that a single-dose IV treatment of peramivir would be effective against influenza infection.
- Thus, we investigated the *in vitro* efficacy of peramivir against various types of influenza A and B viruses including recently isolated viruses.
- We also investigated the *in vivo* therapeutic efficacy of qd dosing of peramivir in mice infected with various influenza A and B viruses.

MATERIALS AND METHODS

Viruses, cell line and compounds
Laboratory strains of influenza virus were obtained from the American Type Culture Collection (Manassas, VA, USA). Clinical isolates of influenza virus were obtained from Sendai Medical Center (Sendai, Japan) and Osaka Prefectural Institute of Public Health (Osaka, Japan). The Madin-Darby canine kidney (MDCK) cells were obtained from the American Type Culture Collection and maintained in Eagle's minimum essential medium (MEM) containing 5% fetal bovine serum. Peramivir, oseltamivir carboxylic acid, oseltamivir phosphate and zanamivir were used.

Mice
BALB/c mice (6 weeks old) were obtained from Charles Rivers Laboratories, Japan, Inc. They were quarantined for 7 days to eliminate the possibility of infection.

NA inhibition assay
Whole viruses inactivated by NP-40 were used as a source of NA activity. 2'-(4-Methylumbelliferyl)-a-D-N-acethlneuraminic acid (MUNANA) was used as substrate. The virus, a compound and MUNANA (final concentration, 10 mM) were mixed in a reaction buffer, and incubated at 37℃ for 30 min. The 50% inhibitory concentration (IC_{50}) was determined by measuring the fluorometric intensity of 4-methylumbelliferon released from MUNANA.

Inhibition of viral replication *in vitro*
A confluent monolayer of MDCK cells was infected with viruses, incubated at 37℃ for 1 hour, washed to remove free viruses, and overlaid with a medium containing agar. Three days after incubation at 37℃, the cells were stained with neutral red. The IC_{50} was determined by counting the plaque number.

Therapeutic effects and inhibition of viral replication in mouse model
Mice were infected intranasally with a 100% lethal dose of virus under ketamine anesthetization. In the study for therapeutic efficacy, peramivir was administered iv (qd for 1 day), and oseltamivir phosphate was administrated orally (bid for 5 days or qd for 1 day). Vehicle (0.5% methylcellulose)-treated control mice were included in the same treatment schedule with repeated dosing of oseltamivir phosphate. The surviving mice were observed daily and the 50% effective dose on survival (ED_{50}) was calculated at 14 days post inoculation. In the study on inhibition of viral replication, both peramivir and oseltamivir carboxylic acid were administered intravenously (qd for 1 day). The vehicle (saline) was also administered intravenously (qd for 1 day) as a control. The viral titer of the lungs collected from infected mice from 1 to 4 days after virus inoculation was measured using MDCK cells.

RESULTS

Inhibitory activity on NA activity (Laboratory strains)

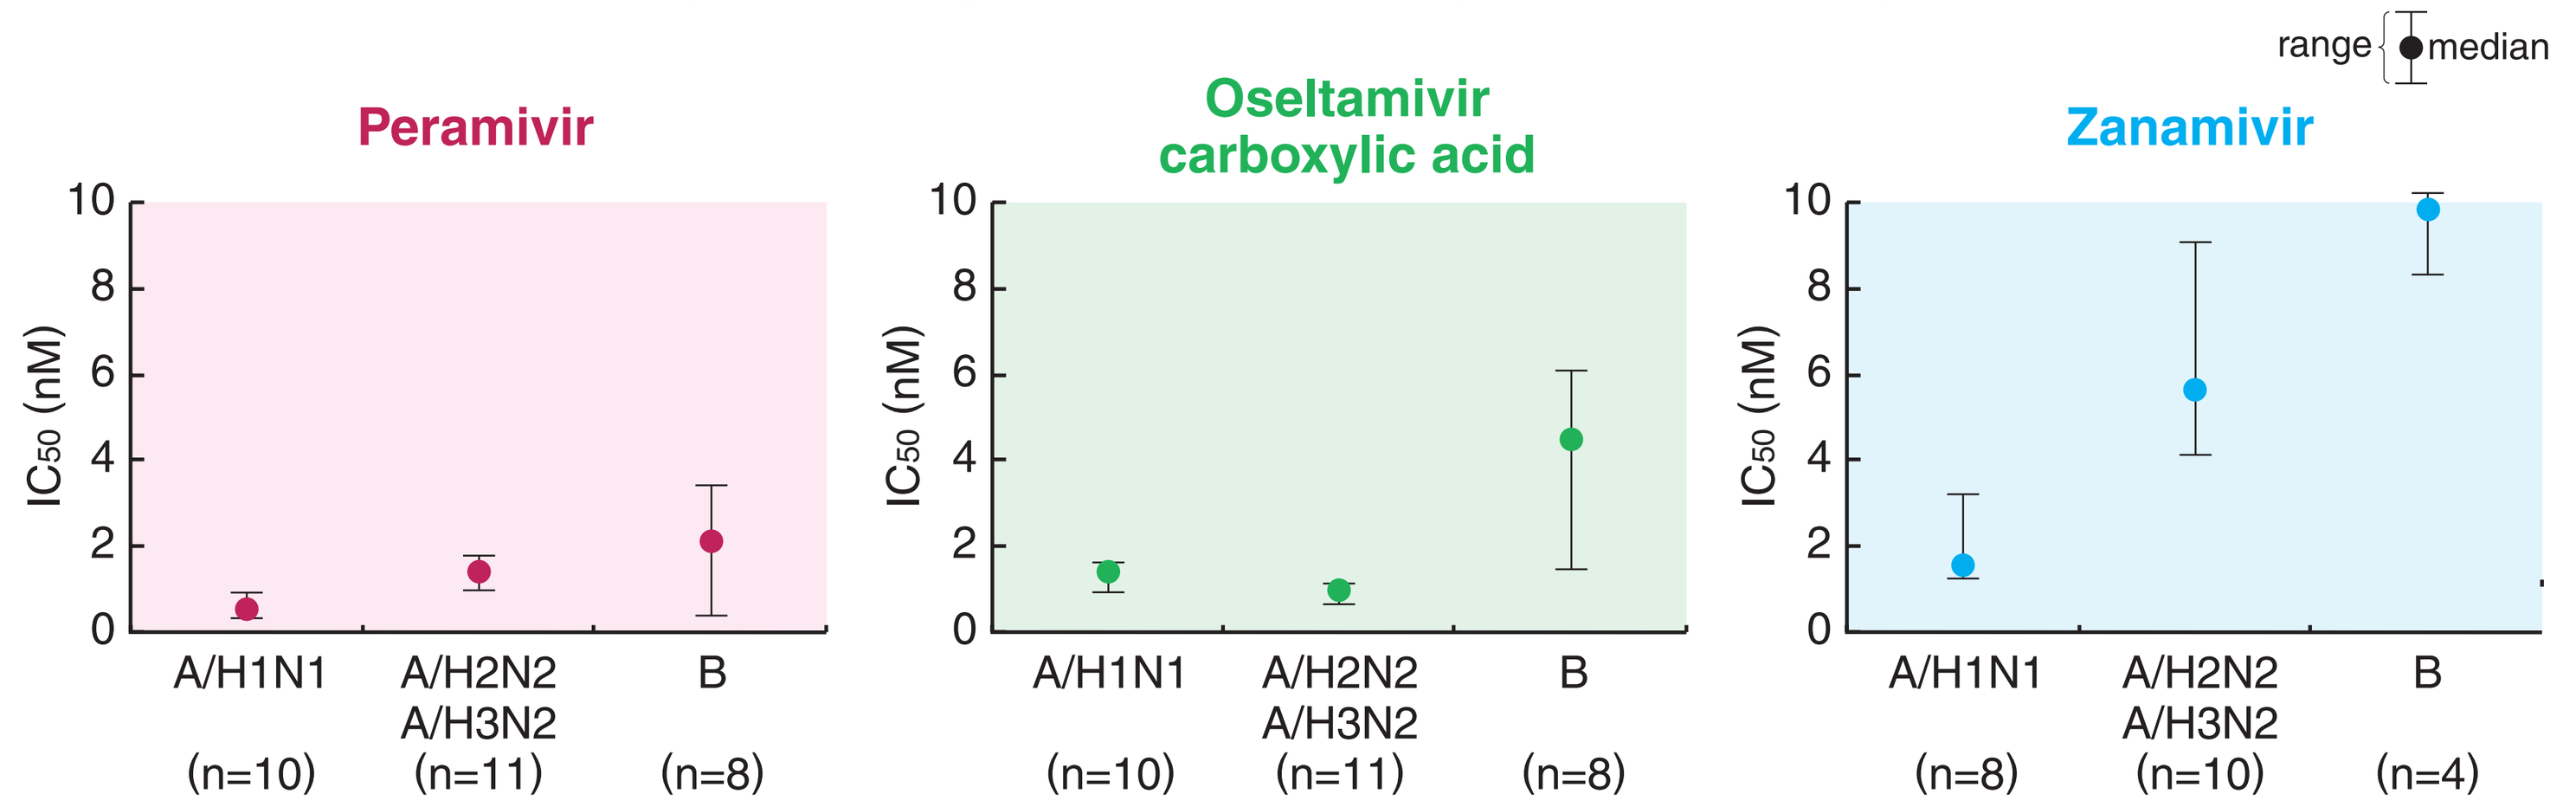
range
median
Peramivir
Oseltamivir carboxylic acid
Zanamivir
IC_{50} (nM)
A/H1N1 (n=10)
A/H2N2 A/H3N2 (n=11)
B (n=8)
A/H1N1 (n=10)
A/H2N2 A/H3N2 (n=11)
B (n=8)
A/H1N1 (n=8)
A/H2N2 A/H3N2 (n=10)
B (n=4)

Inhibitory activity on NA activity (Clinical isolates)

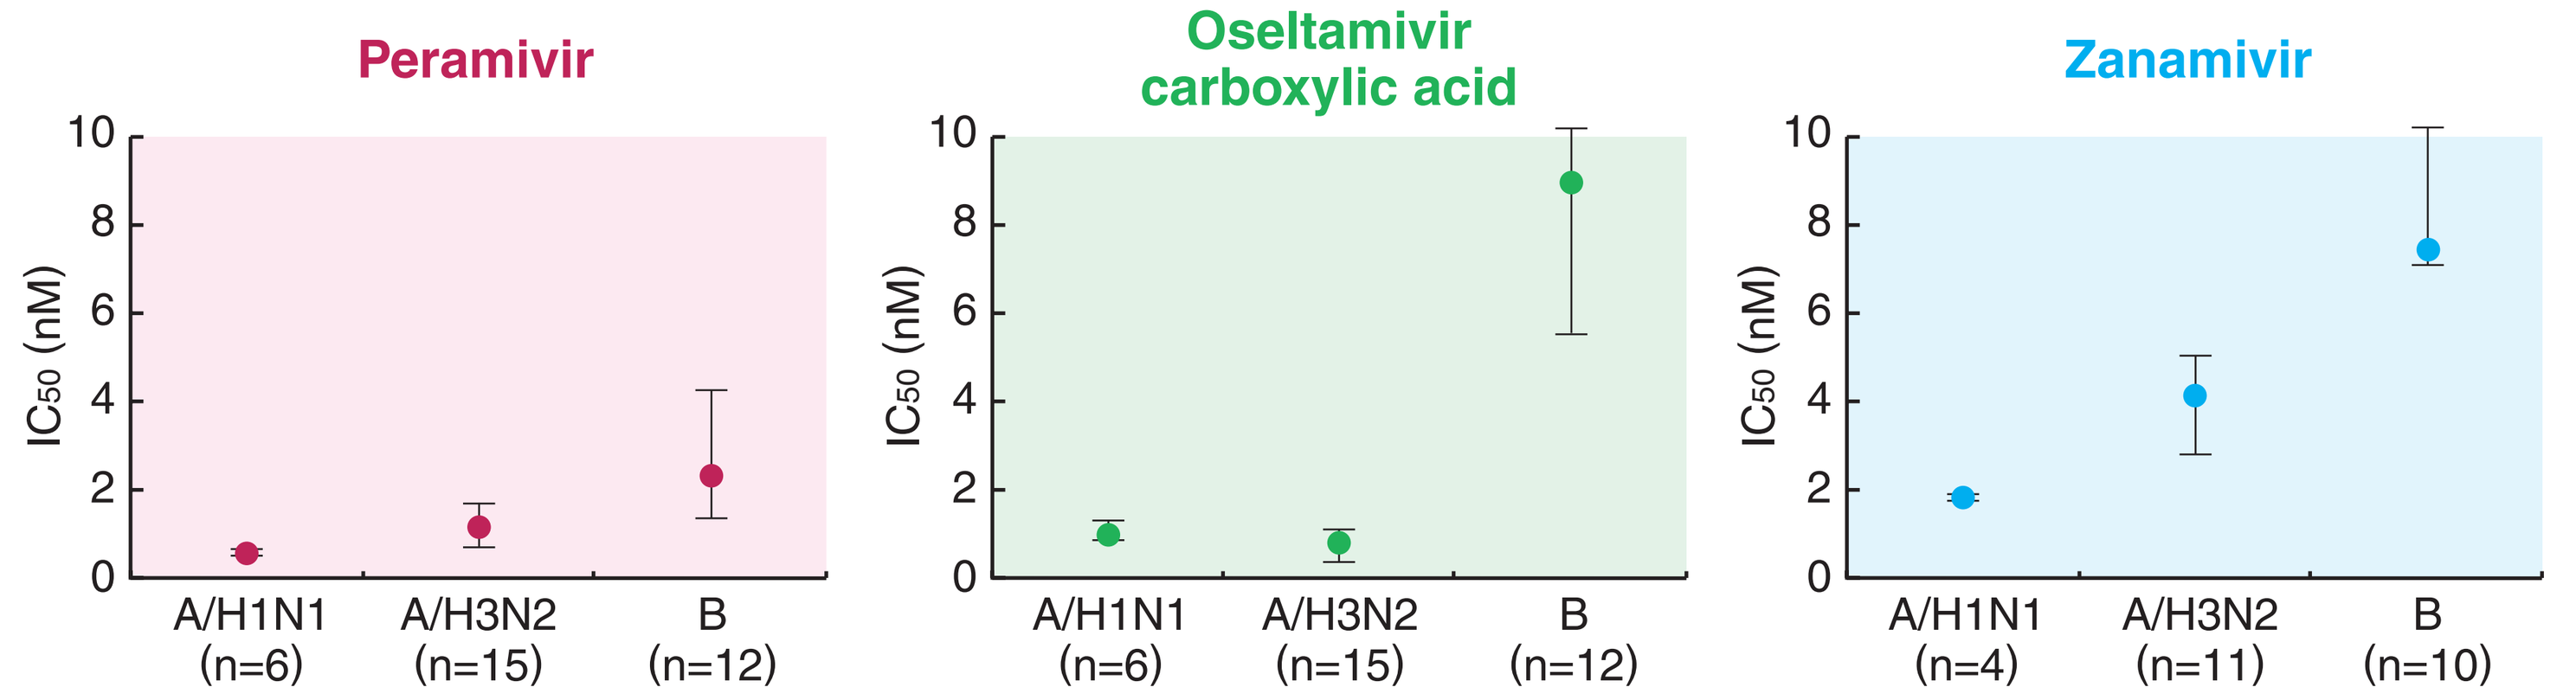
Peramivir
Oseltamivir carboxylic acid
Zanamivir
IC_{50} (nM)
A/H1N1 (n=6)
A/H3N2 (n=15)
B (n=12)
A/H1N1 (n=6)
A/H3N2 (n=15)
B (n=12)
A/H1N1 (n=4)
A/H3N2 (n=11)
B (n=10)

- Peramivir inhibited NA activity of various influenza A and B viruses including laboratory strains and clinical isolates with nano molar potency.
- IC_{50}s of peramivir against influenza B viruses were lower than that of oseltamivir carboxylic acid and zanamivir.

Inhibitory activity on *in vitro* viral replication activity

type	subtype	strain	IC_{50} (nM)		
			Peramivir	Oseltamivir carboxylic acid	Zanamivir
A	H1N1	A/WS/33	15	60	37
A	H1N1	A/PR/8/34	0.93	140	5.8
A	H3N2	A/Victoria/3/75	0.36	0.78	4.6
B	-	B/Lee/40	25	31	21
B	-	B/Maryland/1/59	26	42	29

- Peramivir inhibited virus replication activity of influenza A and B viruses at a lower range of IC_{50} than that of oseltamivir carboxylic acid and zanamivir.

Therapeutic effect on A/Victoria/3/75-infected mice

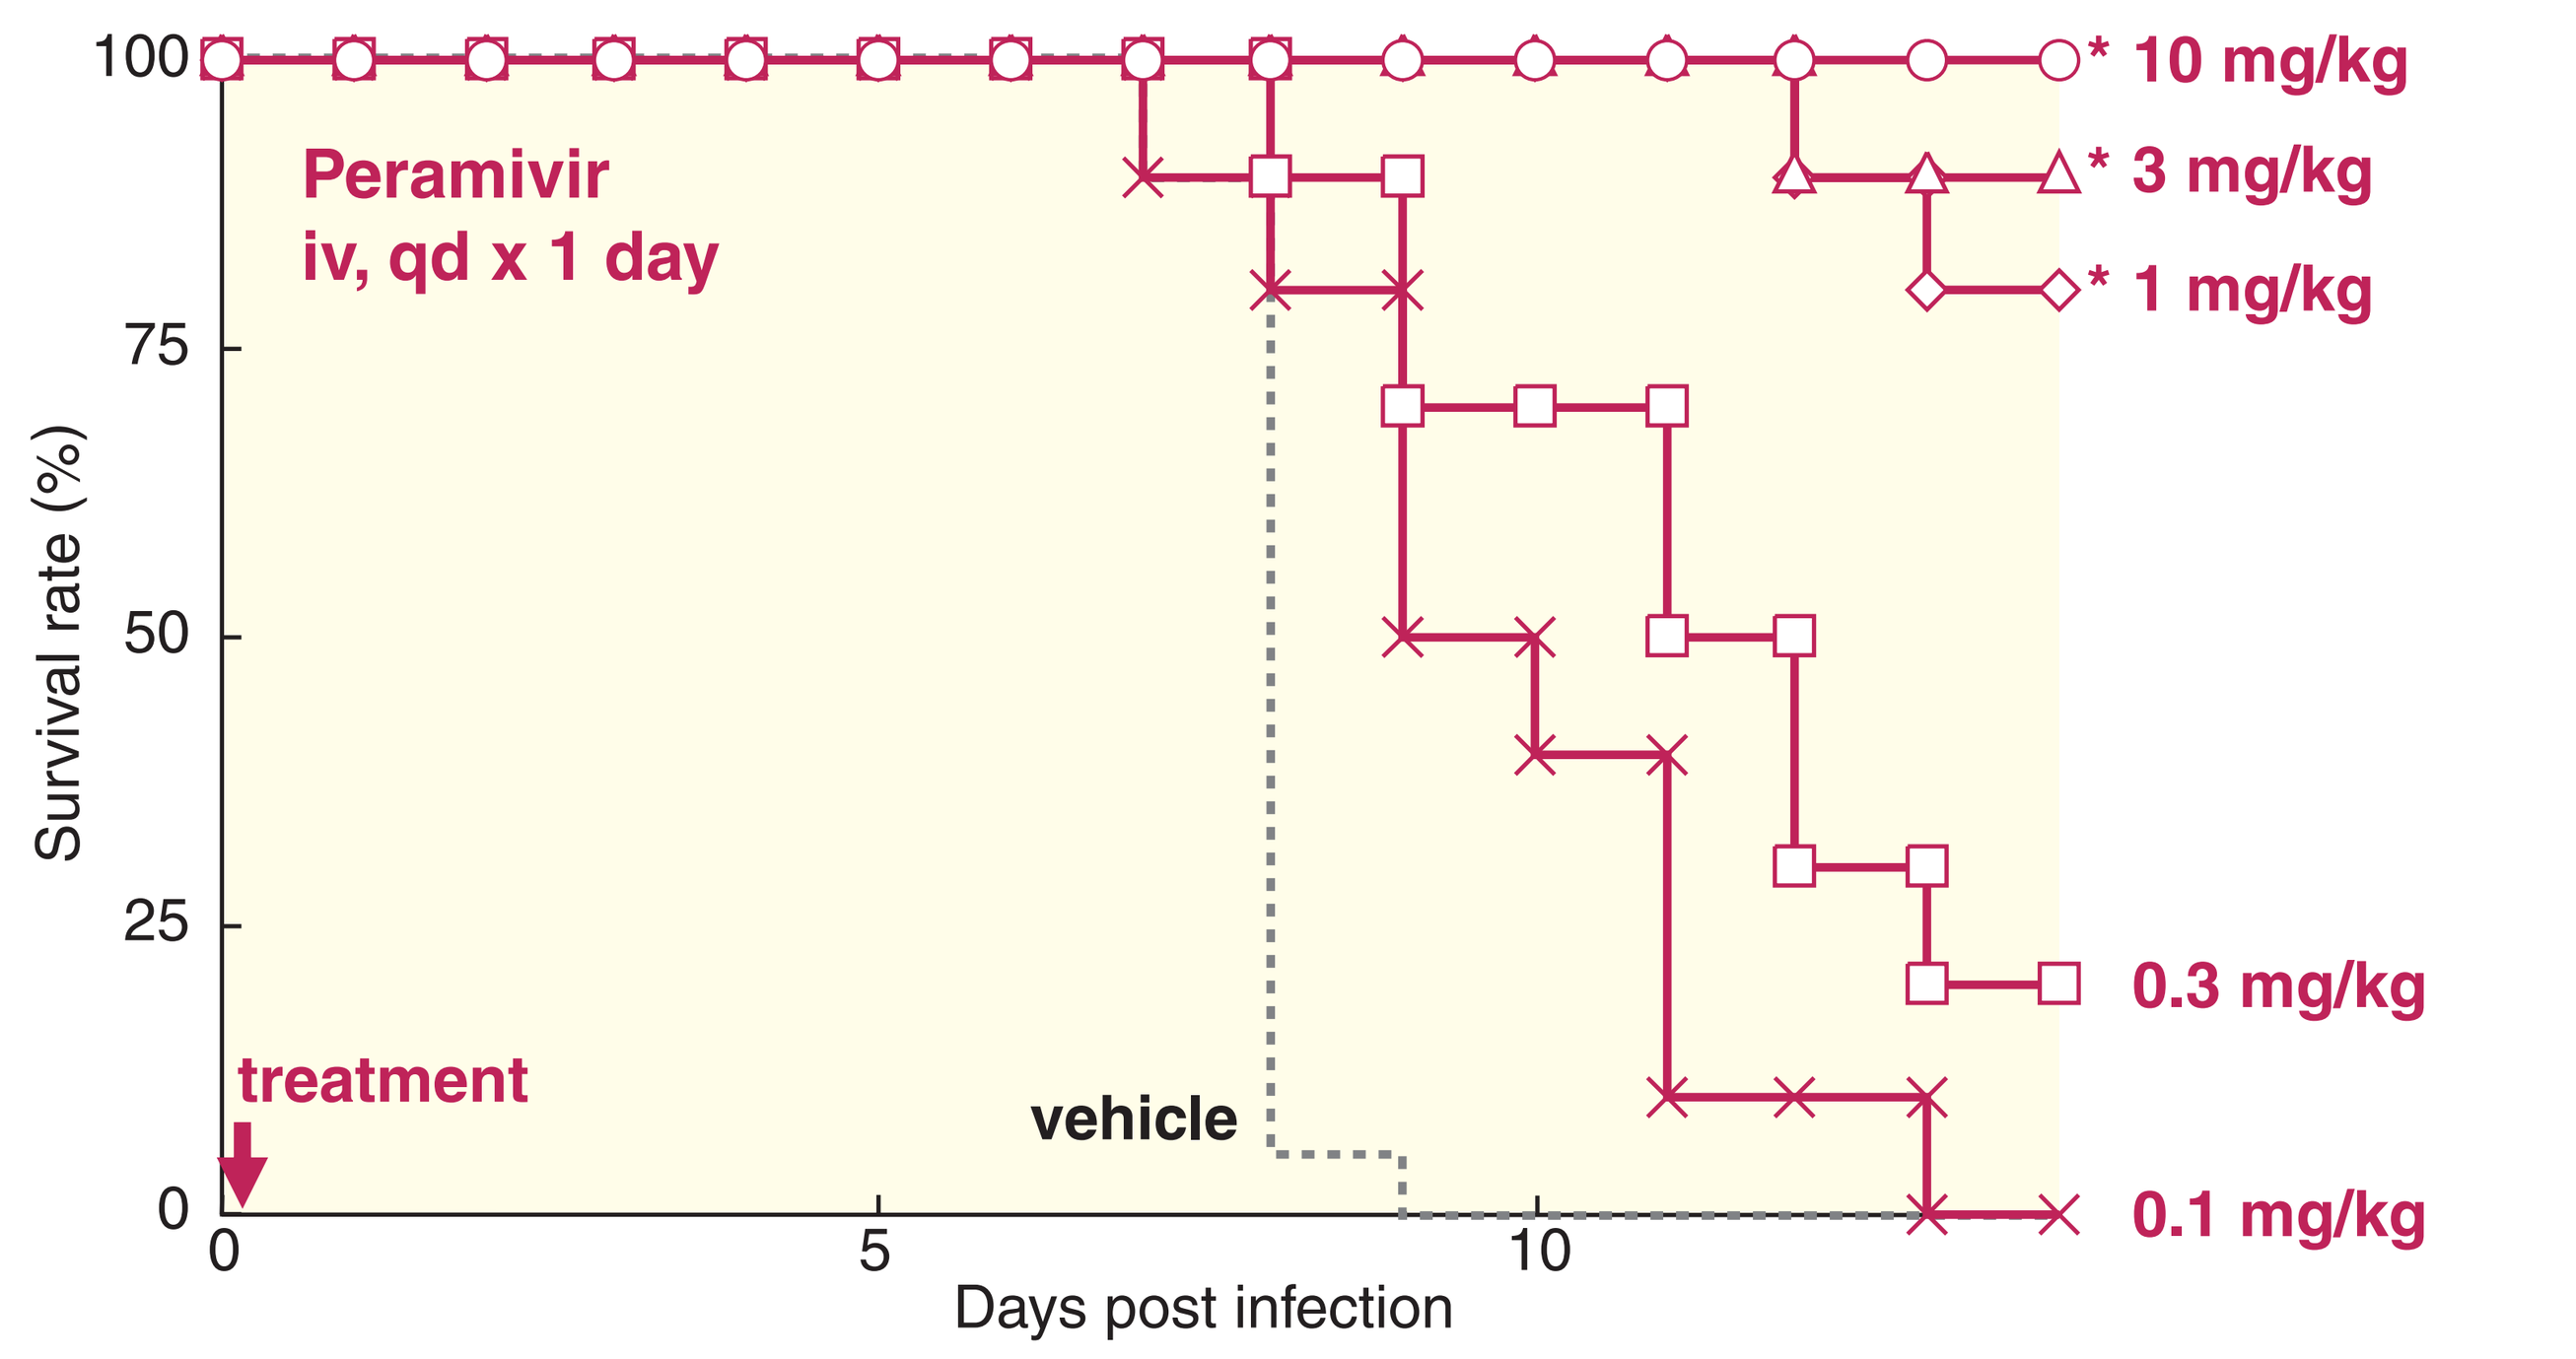
Peramivir
iv, qd x 1 day
* 10 mg/kg
* 3 mg/kg
* 1 mg/kg
0.3 mg/kg
0.1 mg/kg
treatment
vehicle
Survival rate (%)
Days post infection

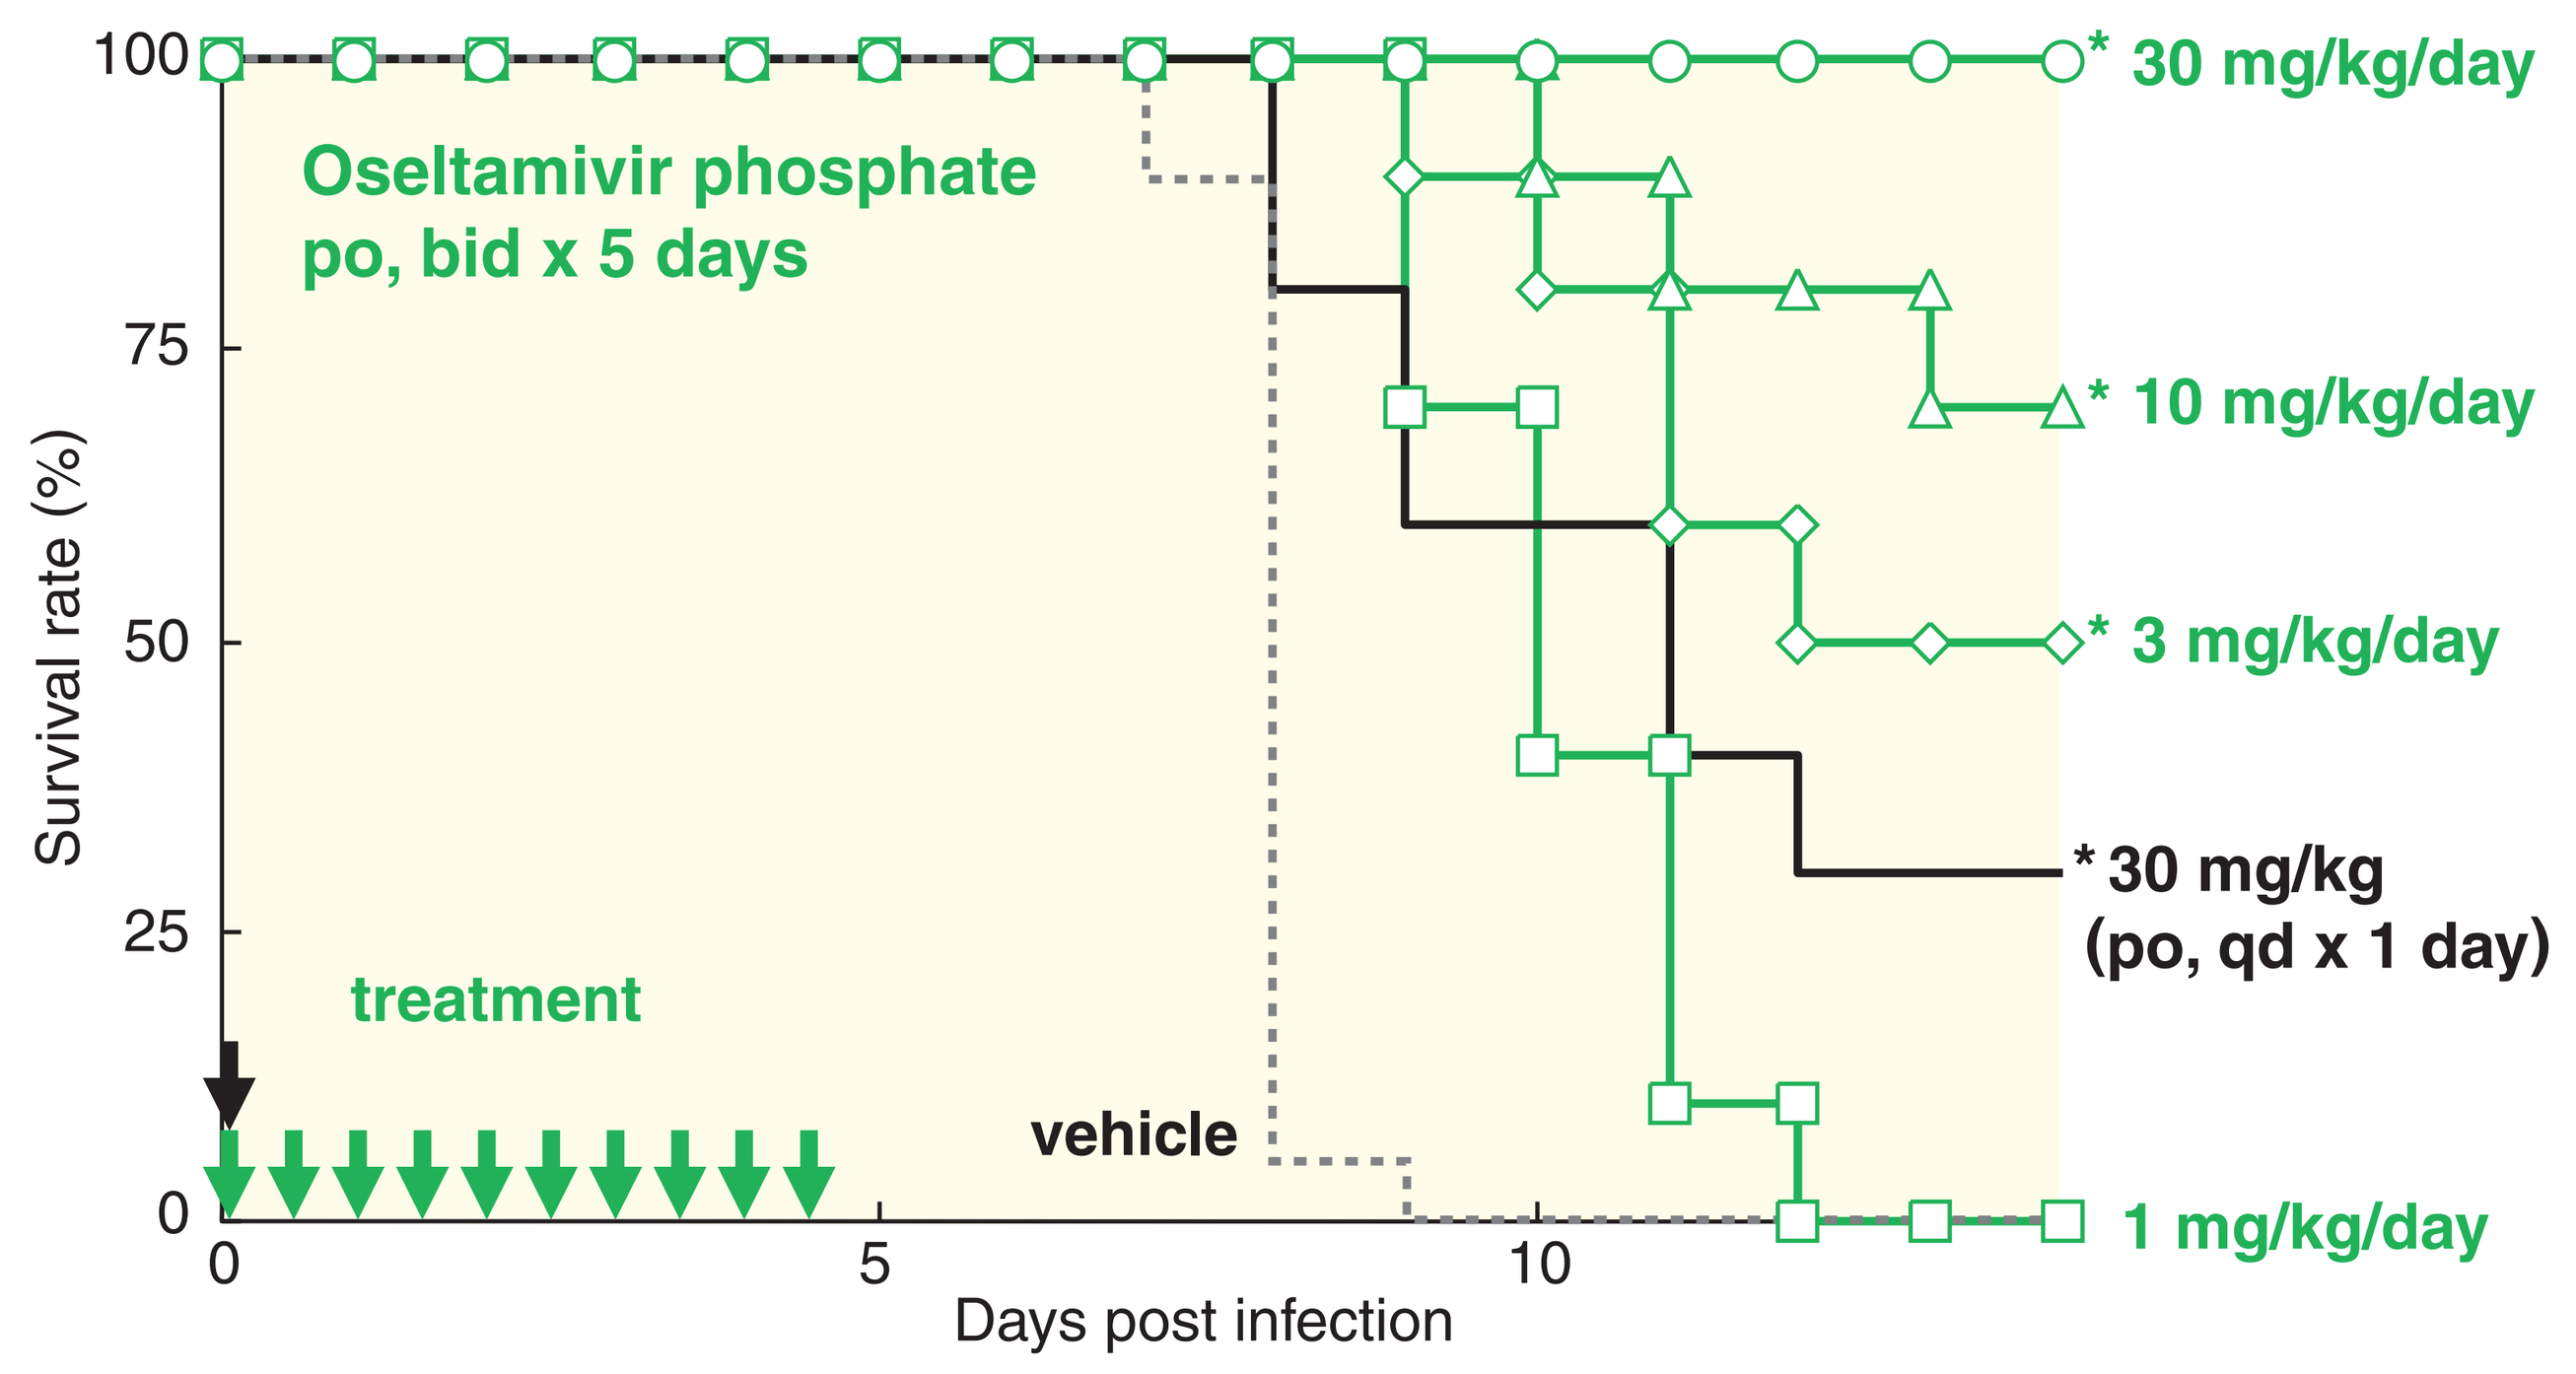
Oseltamivir phosphate
po, bid x 5 days
* 30 mg/kg/day
* 10 mg/kg/day
* 3 mg/kg/day
* 30 mg/kg
(po, qd x 1 day)
1 mg/kg/day
treatment
vehicle
Survival rate (%)
Days post infection

- Peramivir iv dosing (qd for 1 day) at more than 1 mg/kg significantly increased the survival rate compared with the vehicle-treated group (*p<0.05).
- Oseltamivir phosphate oral dosing (bid for 5 days) at more than 3 mg/kg/day showed significant increase of the survival rate compared with the vehicle-treated group (*p<0.05).
- Oseltamivir phosphate oral dosing (qd for 1 day) at 30 mg/kg showed significant increase of the survival rate compared with the vehicle-treated group (*p<0.05). However, it did not lead to more than 50% prevention of death.

Comparison of therapeutic effect on mice infected by various influenza viruses

type	subtype	strain	ED_{50}		
			Peramivir (iv, single) [mg/kg]	Oseltamivir phosphate (po, bid for 5 days) [mg/kg/day]	Oseltamivir phosphate (po, single) [mg/kg]
A	H1N1	A/WS/33	1.5	3.0	>10
A	H1N1	A/PR/8/34	0.4	6.7	>30
A	H3N2	A/Victoria/3/75	0.6	4.3	>30
B	-	B/Lee/40	1.0	7.8	>30
B	-	B/Maryland/1/59	0.1	1.7	>10

Starting time of administration was immediately after virus inoculation except for A/WS/33 (48hr after virus inoculation)

- ED_{50} values of peramivir iv dosing (qd for 1 day) against various influenza-infected mice ranged from 0.14 to 1.5 mg/kg, which was markedly lower than that of oseltamivir phosphate oral dosing (bid for 5 days) (ranging from 8.5 to 39 mg/kg in total dose).

Inhibitory effect on viral replication activity in mice

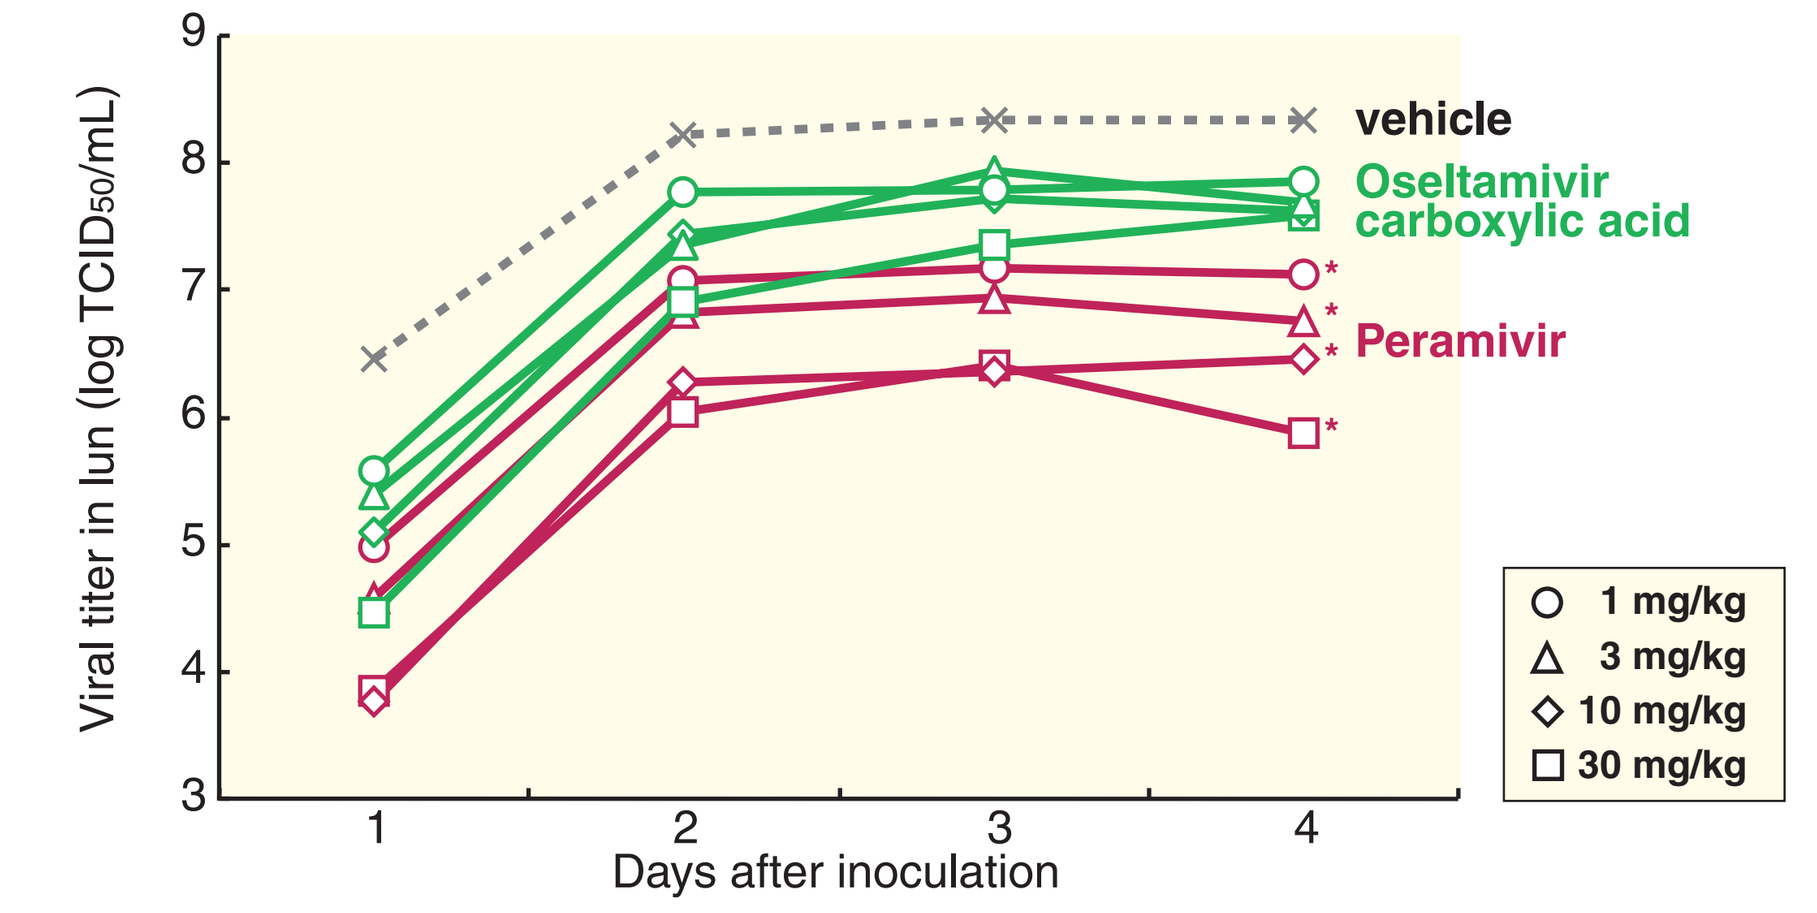
vehicle
Oseltamivir carboxylic acid
Peramivir
1 mg/kg
3 mg/kg
10 mg/kg
30 mg/kg
Viral titer in lun (log $TCID_{50}$/mL)
Days after inoculation

- Peramivir iv dosing (qd for 1 day) at 1-30 mg/kg significantly reduced the virus titers in lungs by 1-2 log $TCID_{50}$/mL compared with that in the control group from day 1 to 4 of post-virus inoculation.
- The inhibitory effect of peramivir on virus titers was significantly stronger than that of the oseltamivir carboxylic acid group at the same dose (*p<0.01).

CONCLUSIONS

- Peramivir strongly inhibited NA activity of various influenza A and B viruses. Regarding influenza B viruses, the efficacy of peramivir was more potent than that of oseltamivir carboxylic acid and zanamivir.
- Peramivir inhibited replication of influenza A and B viruses at a lower range of IC_{50} than those of oseltamivir carboxylic acid and zanamivir.
- Intravenous treatment with peramivir (qd for 1 day) had a significant therapeutic effect in lethal mouse models, which was superior to oral dosing of oseltamivir phosphate (bid for 5 days).
- Intravenous treatment with peramivir (qd for 1 day) had a significant inhibitory effect in viral replication in mice lungs, which was superior to the same treatment with oseltamivir carboxylic acid.

ACKNOWLEDGEMENT

- We thank Dr. Hidekazu Nishimura of Sendai Medical Center (Sendai, Japan) and Dr. Tetsuo Kase of Osaka Prefectural Institute of Public Health (Osaka, Japan) for providing clinical isolates of influenza virus for this study.

12-15 September 2009, SAN FRANCISCO 49th ICAAC