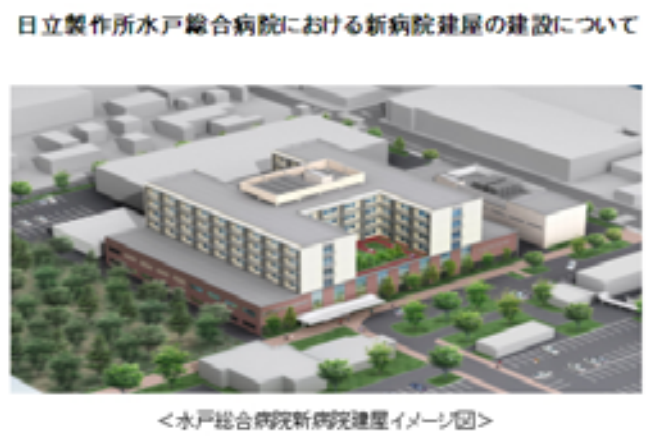
日立製作所水戸総合病院における新病院建屋の建設について
<水戸総合病院新病院建屋イメージ図>

HITACHI
Inspire the Next

Serum Antibody Titres Against Influenza Virus In Pediatric Patients After Treating With Peramivir

1 Department of Pediatrics, Hitachi, Ltd., Hitachinaka General Hospital, Hitachinaka, Japan
2 Department of Pediatrics, Shizuoka Kosei Hospital, Shizuoka, Japan

Toshihiro Tanaka, MD[1,2], Yoji Nagai, MD, PhD[1]

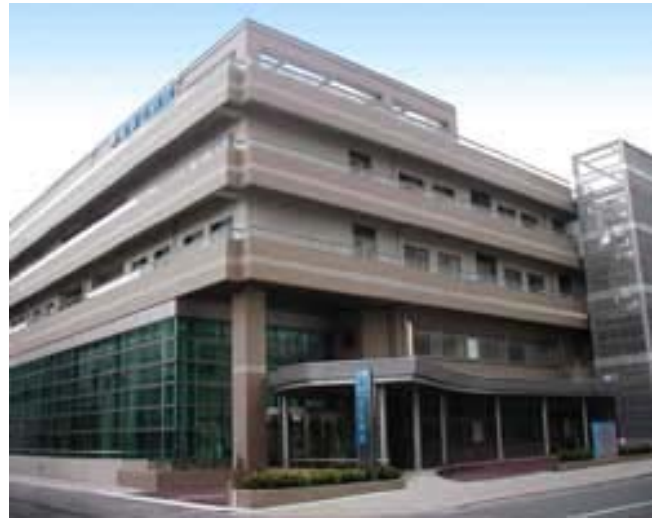

静岡厚生連 静岡厚生病院

Background

Influenza virus infection is known to be basically self-limiting; however, serious conditions, such as pneumonia with respiratory failure and encephalopathy, can occur especially in high-risk patients, including children, and thorough treatments are required. Two types of neuraminidase inhibitors, oseltamivir (Tamiflu®) and zanamivir (Relenza®), have been used to treat influenza virus infection in this decade.

Peramivir (Rapiacta®, Shionogi & Co., Ltd. / BioCryst), a new neuraminidase inhibitor, was approved for adults and a clinical trial for children was conducted in Japan during the 2009-2010 influenza season. Regardless of patients' condition, peramivir can be given intravenously. The data from the trials showed that peramivir is effective and safe enough to use in both adults and children with influenza virus infection. It is highly effective and the time to recover from the infection is shortened after the administration of mostly one dose.

Because peramivir is highly effective and seems to produce rapid recovery from the infection, it was uncertain whether a patient obtains serum antibody against influenza virus after treating with it.

Objectives

To assess whether patients obtain serum antibody against influenza virus after treating with peramivir in spite of its immediate effect and patients' quick recovery.

Methods

A multicenter and open-label clinical trial without a control group was conducted in children with influenza virus infection during the Pandemic H1N1/09 influenza epidemic in Japan in order to evaluate the efficacy and safety of peramivir. Peramivir was given intravenously at a dosage of 10 mg/kg (600 mg maximum) once daily.

Blood samples were provided before and after the administration and serum was separated from each sample and analyzed for the titre of antibody against influenza virus in the hemagglutination inhibition (HI) test. This study was conducted besides the trial.

Subjects

Inclusion

Patients who;

1. were 28 d/o and older & younger than 16 y/o.
2. had fever (38.0ºC and higher) and showed positive result in a rapid diagnostic test.
3. were diagnosed within 48 hours of onset (37.5ºC and higher).
4. gave written consent by parents.

Exclusion

Pediatric patients who;

1. had neurological symptoms, such as convulsion and delirium.
2. were treated with steroids and/or other immunosupressants for their medical condition.

Results 1

Characteristics of subjects in our Hosp.

Case	Sex	Age	Onset to Tx	Dose	Detected Virus
1	M	15.5 y/o	5.6 hr	1	Pandemic H1N1/09
2	M	2.8 y/o	25.8 hr	1	Pandemic H1N1/09
3	M	5.7 y/o	18.2 hr	1	Pandemic H1N1/09
4	F	4.8 y/o	6.9 hr	1	Pandemic H1N1/09
5	F	1.8 y/o	8.3 hr	1	Pandemic H1N1/09
6	F	4 m/o	8.7 hr	2	Pandemic H1N1/09
7	F	1.6 y/o	20.2 hr	1	Pandemic H1N1/09
8	F	1.2 y/o	16.1 hr	1	Pandemic H1N1/09
9	F	6 m/o	28.6 hr	1	Pandemic H1N1/09
	Min.	4 m/o	5.6 hr	1	
	Max.	15.5 y/o	28.6 hr	2	
	Median	1.8 y/o	16.1 hr	1	

Results 2

Serum HI titres against Pandemic H1N1/09 virus

Case	Before Tx	Next day of Tx	1 wk after Tx	1-2 mo after Tx
1	-	-	-	1: 80
2	-	< 1:10	-	1:160
3	-	< 1:10	-	1: 40
4	-	< 1:10	-	1:160
5	< 1:10	-	1:10	1: 80
6	-	-	< 1:10	1:640
7	< 1:10	< 1:10	1:10	1:320
8	< 1:10	< 1:10	< 1:10	1: 40
9	< 1:10	< 1:10	-	1:160

* Some data were missing because remaining blood samples in the clinical trial were used for the analysis.

Conclusions

Although peramivir produces rapid recovery from influenza virus infection, it does not seem to interfere with obtaining antibodies in blood.

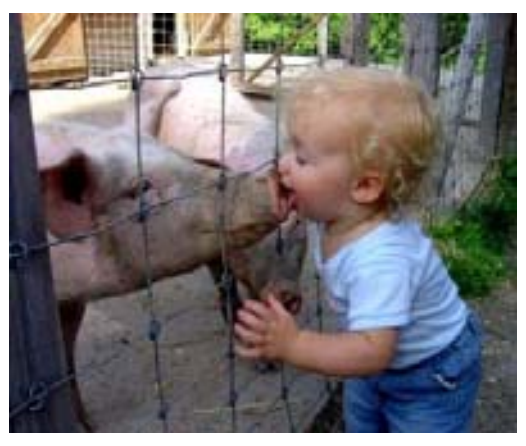

Acknowledgements

- Mr. Mitsunori Yasugi
- Ms. Nobuko Kanazawa, RN
- Mr. Takanori Kato
- Mr. Toshiichi Seki

This project is supported by;

- SRL,Inc.
- Ethic Co.,Ltd.
- Medical Business Unit, Hitachi Chemical Co., Ltd.

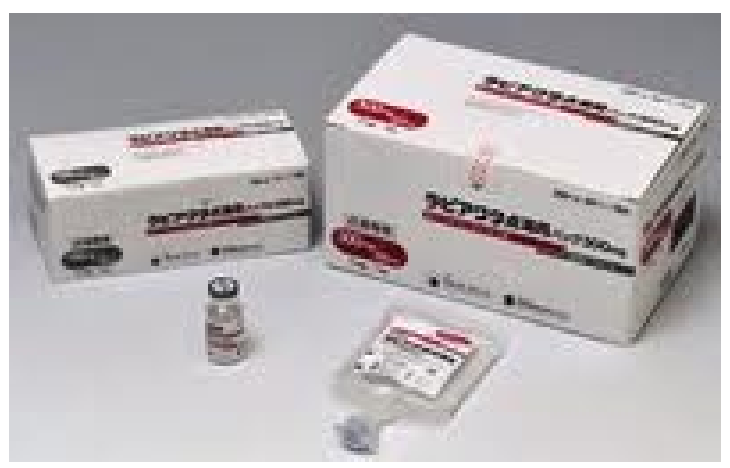