Antiviral Susceptibility of Influenza Viruses After Therapy (Rx) with Peramivir (PVR) Vs. Placebo (PBO) or Oseltamivir (OSE) in Influenza Subjects in Phase 2 and 3 Trials

Jaime E. Hernandez, MD,
BioCryst Pharmaceuticals,
4505 Emporer Blvd,
Durham, NC, 27703, USA
919-859-7921 (phone)
919-851-1416 (fax)
jhernandez@biocryst.com)

J. E. Hernandez[1], MD, M. G. Ison[2], MD, MS, G. Atiee[3], MD, S. Kohno[4] W. J. Alexander[1], MD, J. Elder[5], PhD, P. Collis[1], PhD

[1]BioCryst Pharmaceuticals, Durham, NC, USA, [2]Northwestern University Feinberg School of Medicine, Chicago, IL USA, [3]HealthCare Discoveries, LLC., San Antonio, TX, USA, [4]Nagasaki University, Japan, [5]PharPoint Res., Wilmington, NC, USA

INTRODUCTION

- Early treatment with NAIs is recommended for subjects hospitalized with influenza[1]
- Concerns about emergence of resistance were raised after most seasonal H1N1 influenza A isolates in 2008 were found to be resistant to oseltamivir (OSE)[2], most with the H275Y mutation
- There are few reports of H275Y isolates after peramivir (PVR) treatment
- Peramivir is an intravenous neuraminidase inhibitor (NAI) approved in Japan and South Korea for treatment of influenza and in Phase 3 global trials to evaluate treatment of influenza patients in the hospital setting
- Peramivir has in vitro activity superior to OSE and zanamivir (ZAN) against wild-type strains of influenza[3,4]
- Resistance of seasonal H1N1 influenza A to PVR is only selected after multiple cycles of in vitro passage[5]
- In 3 completed randomized double-blind Phase 2 and 3 trials of IV and IM peramivir, subjects treated with peramivir demonstrated reductions in viral shedding and time to alleviation of influenza symptoms similar to those treated with oseltamivir[6] and superior to placebo[7,8]
- With viral susceptibility data from 4 peramivir trials now available, we have combined these data to assess the potential development of resistance to peramivir in patients with influenza treated with peramivir

METHODS

Study Design and Treatments

Peramivir Clinical Trials with Virology Susceptibility Data

Study:	BCX1812-211	0722T0621	BCX1812-201	BCX1812-303
Phase	2	2	2	3
Design	R, DB, PG	R, DB, PG	R, DB, DD, PG	R, PG
Years Conducted	2006-08	2007-08	2007-08	2009-10
Countries	7	1	7	5
Sites	151	75	84	59
Peramivir	150 mg IM 300 mg IM	300 mg IV 600 mg IV	200 mg IV 400 mg IV	300 mg IV BID 600 mg IV QD
Control	Placebo	Placebo	Oseltamivir 75 mg BID	None
Treatment Duration	Single dose	Single dose	5 days	5-10 days

IV=intravenous, IM=intramuscular, R=randomized, DB=double blind, PG=parallel group, DD=double dummy

- 4 completed randomized phase 2 and 3 studies conducted from December 2006 to October 2010
- Eligible patients were randomly assigned to treatment with peramivir (PVR) IV or IM, oral oseltamivir, or matched placebo

Study Subjects

- Adult or adolescent inpatients or outpatients
- Symptoms consistent with diagnosis of acute influenza infection
- Confirmation of influenza A or B infection of influenza A or B infection by Rapid Antigen Test prior to randomization except for study BCX1812-303

Study Assessments

- Virus was collected from nasopharyngeal swabs at baseline and post treatment
- Influenza A subtype and B infection at baseline was determined by RT-PCR, culture, or serology
- H275Y status was assessed by pyrosequencing in H1N1 specimens
- Seasonal A/H1N1 isolates without the H275Y mutation by pyrosequencing were considered wild type (WT)
- Neuraminidase enzyme inhibition (NAI) assay was performed on virus MDCK culture supernatants by MUNANA technique to assess the IC_{50} of PVR, oseltamivir (OSE) and zanamivir (ZAN) at baseline and post treatment. IC_{50} values could not be determined for all virus subtypes that were confirmed by RT-PCR, culture or serology
- Genotypic sequencing at baseline and post treatment was performed in 2 populations:
 - IC_{50} subset: Paired isolates with change in NAI IC_{50} from baseline to last timepoint > mean + 2 SD, or > 3 fold
 - Delayed clearance: Subjects with positive culture post-treatment at day 5 or day 9

RESULTS

Study Subjects

Subjects in Peramivir Clinical Trials by Dose and Influenza Strain at Baseline

Study:	BCX1812-211	0722T0621	BCX1812-201	BCX1812-303	Total
Enrolled Subjects	344 outpatients	300 outpatients	137 inpatients	234 inpatients	1015
Treated Subjects	PVR 150 IM SD = 113 PVR 300 IM SD = 115 Placebo IM SD = 114	PVR 300 IV SD = 99 PVR 600 IV SD = 99 Placebo IV SD = 100	PVR 200 QD IV 5d = 45 PVR 400 QD IV 5d = 46 OSE 75 BID PO = 46	PVR 300 BID IV 5-10d = 114 PVR 600 BID IV 5-10d = 116	1007
Subjects with A/sH1N1 WT*	PVR 150 IM SD = 32 PVR 300 IM SD = 21 Placebo IM SD = 30	PVR 300 IV SD = 74 PVR 600 IV SD = 69 Placebo IV SD = 72	PVR 200 QD IV 5d = 5 PVR 400 QD IV 5d = 7 OSE 75 BID PO = 6		316
Subjects with A/H3N2*	PVR 150 IM SD = 50 PVR 300 IM SD = 57 Placebo IM SD = 53	PVR 300 IV SD = 21 PVR 600 IV SD = 25 Placebo IV SD = 24	PVR 200 QD IV 5d = 20 PVR 400 QD IV 5d = 26 OSE 75 BID PO = 22		298
Subjects with B*	PVR 150 IM SD = 19 PVR 300 IM SD = 25 Placebo IM SD = 24	PVR 300 IV SD = 2 PVR 600 IV SD = 1 Placebo IV SD = 0	PVR 200 QD IV 5d = 15 PVR 400 QD IV 5d = 6 OSE 75 BID PO = 11	PVR 300 BID IV 5-10d = 3 PVR 600 BID IV 5-10d = 2	108
Subjects with A/Indeterminate*	PVR 150 IM SD = 3 PVR 300 IM SD = 2 Placebo IM SD = 2	PVR 300 IV SD = 2 PVR 600 IV SD = 2 Placebo IV SD = 4	PVR 200 QD IV 5d = 0 PVR 400 QD IV 5d = 0 OSE 75 BID PO = 1	PVR 300 BID IV 5-10d = 10 PVR 600 BID IV 5-10d = 16	42
Subjects with A/sH1N1H275Y**			PVR 200 QD IV 5d = 1 PVR 400 QD IV 5d = 1 OSE 75 BID PO = 1		3
Subjects with 2009 A/H1N1*				PVR 300 BID IV 5-10d = 43 PVR 600 BID IV 5-10d = 51	94
Subjects with Indeterminate*				PVR 300 BID IV 5-10d = 1 PVR 600 BID IV 5-10d = 1	2

IV=intravenous, IM=intramuscular, SD=single dose, PO=oral
*Determined by RT PCR, culture or paired serology, either by a local or central lab
**Determined by pyrosequencing

- 1007 subjects were treated once or over 5-10 days with PVR delivered IM or IV or OSE delivered PO
- Two studies were placebo controlled
- Influenza was confirmed at baseline and the strain identified by RT PCR, culture or paired serology in 863 subjects

Viral Susceptibility at Baseline

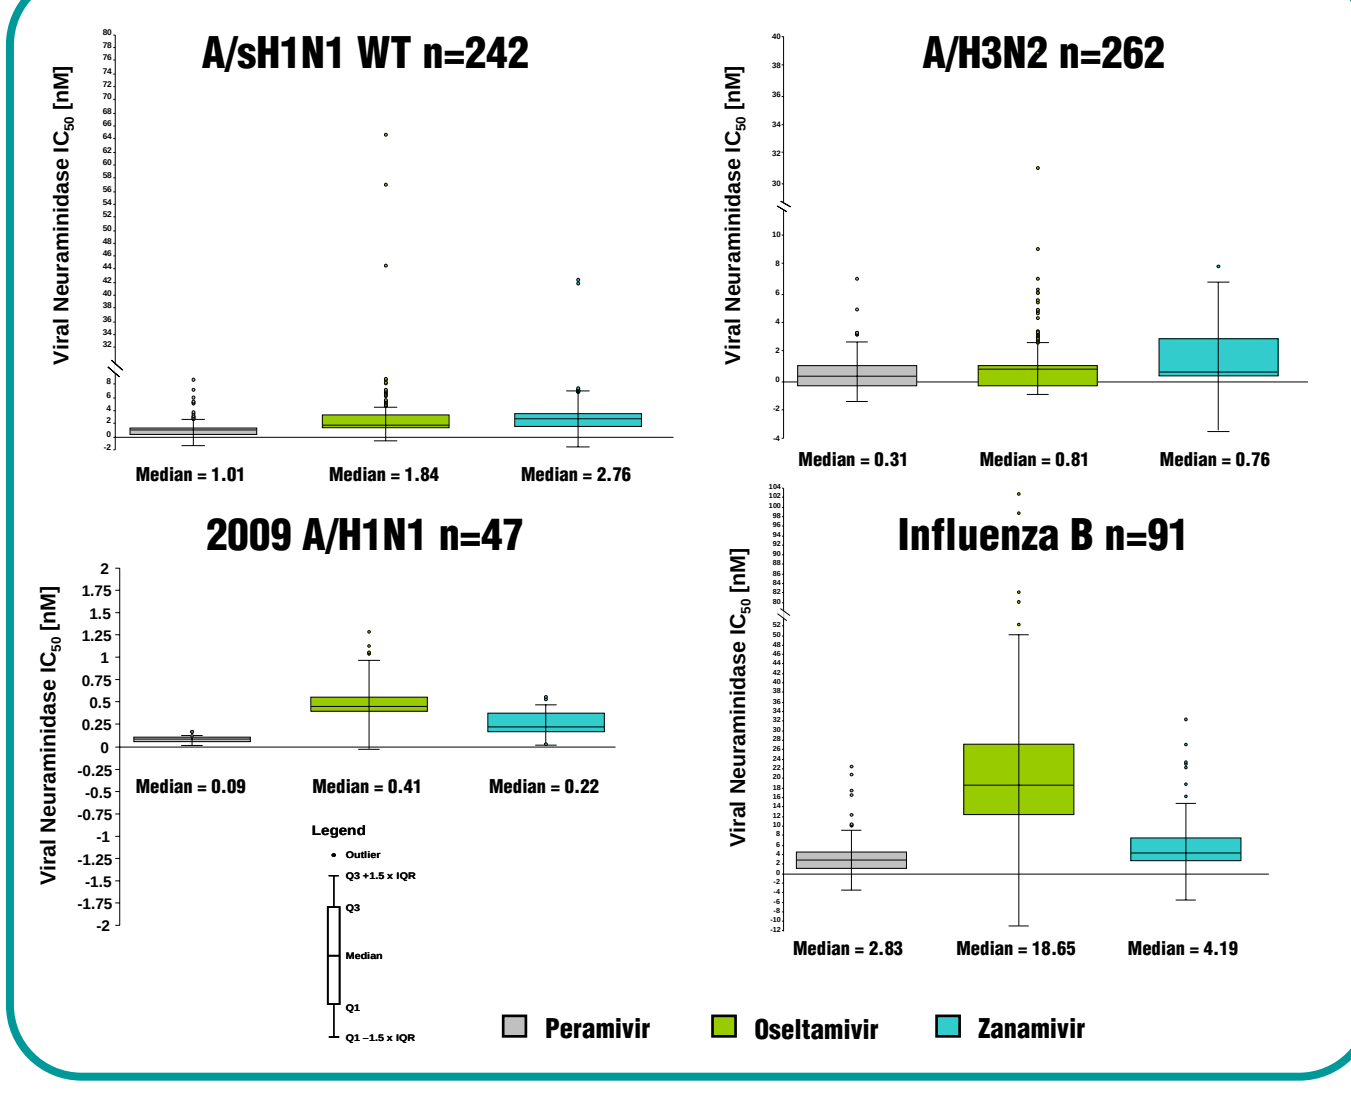

A/sH1N1 WT n=242
Viral Neuraminidase IC_{50} [nM]
Median = 1.01
Median = 1.84
Median = 2.76
A/H3N2 n=262
Median = 0.31
Median = 0.81
Median = 0.76
2009 A/H1N1 n=47
Median = 0.09
Median = 0.41
Median = 0.22
Influenza B n=91
Median = 2.83
Median = 18.65
Median = 4.19
Legend
Outlier
Q3 +1.5 x IQR
Q3
Median
Q1
Q1 –1.5 x IQR
Peramivir
Oseltamivir
Zanamivir

- Mean and median IC_{50} values for all influenza A and B viruses isolated at baseline were lowest for peramivir compared to the two other NAIs
- 3 isolates in the 201 study had the H275Y mutation at baseline; they had high IC_{50} values for oseltamivir and intermediate IC_{50} values for peramivir

Change in Viral Susceptibility

Seasonal A/H1N1 WT

Treatment Group		Peramivir	Oseltamivir	Zanamivir
		NAI, median (min, max)		
PBO, n=81	IC_{50} (nM) of last positive culture	0.90 (0.01, 2.98)	1.63 (0.23, 13.02)	2.55 (0.06, 32.64)
	Fold change from baseline*	0.90 (0.04, 14.00)	0.96 (0.19, 7.92)	0.95 (0.12, 40.00)
PVR 150 IM, SD, n=20	IC_{50} (nM) of last positive culture	0.22 (0.01, 8.34)	0.95 (0.10, 95.45)	0.54 (0.08, 10.39)
	Fold change from baseline*	1.91 (0.21, 69.50)	1.15 (0.12, 28.81)	0.76 (0.08, 14.60)
PVR 300 IM SD, n=19	IC_{50} (nM) of last positive culture	0.14 (0.01, 9.68)	1.72 (0.04, 24.37)	1.04 (0.21, 5.44)
	Fold change from baseline*	1.08 (0.04, 29.33)	1.40 (0.34, 36.92)	1.15 (0.49, 23.54)
PVR 300 IV SD, n=57	IC_{50} (nM) of last positive culture	1.34 (0.68, 5.48)	2.22 (1.09, 10.20)	3.40 (1.74, 22.40)
	Fold change from baseline*	0.95 (0.58, 2.64)	0.95 (0.41, 2.64)	0.97 (0.31, 3.53)
PVR 600 IV SD, n=49	IC_{50} of last positive culture	1.21 (0.79, 30.30)	1.86 (0.10, 94.00)	2.48 (1.62, 5.15)
	Fold change from baseline*	1.14 (0.74, 24.82)	1.00 (0.04, 2.88)	0.99 (0.48, 1.85)
PVR 200 IV, n=5	IC_{50} of last positive culture	0.05 (0.01, 0.31)	1.11 (0.68, 2.31)	0.82 (0.08, 2.49)
	Fold change from baseline*	0.79 (0.16, 1.00)	0.60 (0.45, 1.31)	0.79 (0.28, 1.71)
PVR 400 IV, n=5	IC_{50} of last positive culture	0.40 (0.01, 0.88)	2.24 (0.89, 2.65)	1.00 (0.42, 2.00)
	Fold change from baseline*	0.88 (0.57, 1.31)	0.98 (0.33, 2.60)	0.68 (0.55, 1.42)
OSE, n=6	IC_{50} of last positive culture	2.23 (0.14, 4.20)	2.65 (0.91, 3.41)	1.72 (0.11, 2.53)
	Fold change from baseline*	1.86 (0.26, 5.19)	1.27 (0.21, 2.38)	1.06 (0.08, 1.98)

*Values = 1 indicate no change, values >1 indicate a fold increase, values <1 indicate a fold decrease

A/H3N2

Treatment Group		Peramivir	Oseltamivir	Zanamivir
		NAI, median (min, max)		
PBO, n=72	IC_{50} (nM) of last positive culture	0.34 (0.01, 2.63)	0.70 (0.06, 20.19)	0.80 (0.03, 8.30)
	Fold change from baseline*	1.04 (0.16, 18.00)	0.99 (0.16, 23.60)	1.07 (0.14, 25.38)
PVR 150 IM, SD, n=44	IC_{50} (nM) of last positive culture	0.17 (0.02, 1.78)	0.50 (0.02, 3.99)	0.71 (0.05, 3.56)
	Fold change from baseline*	1.21 (0.11, 53.00)	1.06 (0.07, 6.23)	1.40 (0.25, 141.00)
PVR 300 IM SD, n=44	IC_{50} (nM) of last positive culture	0.21 (0.04, 4.65)	0.51 (0.11, 13.09)	0.82 (0.04, 14.15)
	Fold change from baseline*	1.60 (0.28, 59.00)	1.22 (0.26, 36.36)	1.40 (0.08, 36.20)
PVR 300 IV SD, n=21	IC_{50} (nM) of last positive culture	1.21 (1.04, 1.67)	0.84 (0.75, 1.24)	2.97 (2.50, 4.22)
	Fold change from baseline*	0.80 (0.42, 1.28)	0.76 (0.42, 1.23)	0.86 (0.61, 1.27)
PVR 600 IV SD, n=24	IC_{50} of last positive culture	1.41 (0.88, 1.92)	1.06 (0.49, 1.66)	3.41 (2.61, 4.56)
	Fold change from baseline*	0.97 (0.47, 1.21)	0.96 (0.42, 1.40)	0.89 (0.70, 1.22)
PVR 200 IV 5d, QD, n=16	IC_{50} of last positive culture	0.33 (0.06, 4.83)	1.36 (0.09, 34.93)	0.58 (0.17, 5.56)
	Fold change from baseline*	0.95 (0.33, 21.95)	1.23 (0.03, 36.39)	0.85 (0.09, 5.10)
PVR 400 IV 5d, QD, n=21	IC_{50} of last positive culture	0.45 (0.04, 2.33)	1.76 (0.05, 10.21)	0.73 (0.13, 4.20)
	Fold change from baseline*	0.86 (0.04, 2.69)	0.99 (0.21, 7.59)	0.87 (0.03, 6.80)
OSE PO 5d BID, n=20	IC_{50} of last positive culture	0.60 (0.13, 7.10)	1.17 (0.26, 6.22)	1.02 (0.20, 4.17)
	Fold change from baseline*	0.90 (0.32, 177.50)	1.01 (o0.10, 4.49)	0.65 (0.07, 25.80)

*Values = 1 indicate no change, values >1 indicate a fold increase, values <1 indicate a fold decrease

2009 A/H1N1

Treatment Group		Peramivir	Oseltamivir	Zanamivir
		NAI, median (min, max)		
PVR 300 IV 5-10d BID, n=22	IC_{50} (nM) of last positive culture	0.09 (0.06, 31.02)	0.37 (0.23, 282.40)	0.23 (0.08, 0.39)
	Fold change from baseline*	1.00 (0.58, 3102.00)	1.00 (0.39, 882.50)	1.05 (0.29, 1.56)
PVR 600 IV 5-10d QD, n=25	IC_{50} of last positive culture	0.10 (0.07, 0.13)	0.38 (0.28, 0.93)	0.25 (0.11, 0.34)
	Fold change from baseline*	0.95 (0.67, 1.63)	0.92 (0.62, 1.69)	1.00 (0.28, 1.69)

*Values = 1 indicate no change, values >1 indicate a fold increase, values <1 indicate a fold decrease

Influenza B

Treatment Group		Peramivir	Oseltamivir	Zanamivir
		NAI, median (min, max)		
PBO, n=19	IC_{50} (nM) of last positive culture	2.53 (0.02, 6.30)	17.63 (0.06, 54.63)	2.81 (0.02, 11.97)
	Fold change from baseline*	1.12 (0.50, 3.04)	1.15 (0.24, 2.52)	1.03 (0.30, 2.41)
PVR 150 IM, SD, n=16	IC_{50} (nM) of last positive culture	2.75 (1.02, 11.69)	15.04 (3.98, 32.95)	5.24 (1.90, 14.91)
	Fold change from baseline*	1.03 (0.21, 2.08)	0.94 (0.24, 1.19)	1.06 (0.36, 2.71)
PVR 300 IM SD, n=21	IC_{50} (nM) of last positive culture	1.74 (0.29, 3.41)	13.12 (0.53, 32.57)	2.83 (0.52, 8.80)
	Fold change from baseline*	0.96 (0.03, 15.25)	0.94 (0.13, 4.08)	0.92 (0.37, 6.50)
PVR 300 IV SD, n=2	IC_{50} (nM) of last positive culture	2.89 (2.64, 3.14)	8.52 (7.28, 9.75)	8.98 (7.75, 10.20)
	Fold change from baseline*	1.03 (1.01, 1.05)	0.99 (0.88, 1.09)	0.97 (0.91, 1.03)
PVR 600 IV SD, n=1	IC_{50} of last positive culture	2.49	7.07	7.86
	Fold change from baseline*	0.89	0.97	0.91
PVR 200 IV 5d, QD, n=15	IC_{50} of last positive culture	4.42 (0.41, 17.60)	21.02 (0.33, 61.25)	4.06 (0.50, 37.65)
	Fold change from baseline*	0.87 (0.04, 2.91)	0.81 (0.03, 1.23)	0.78 (0.11, 1.96)
PVR 400 IV 5d, QD, n=6	IC_{50} of last positive culture	2.58 (0.06, 6.26)	30.52 (4.62, 71.51)	2.55 (1.67, 10.83)
	Fold change from baseline*	0.90 (0.02, 3.85)	0.70 (0.44, 1.97)	0.85 (0.50, 1.69)
OSE PO 5d BID, n=11	IC_{50} of last positive culture	6.89 (0.26, 12.91)	41.92 (8.17, 88.63)	5.90 (2.11, 21.21)
	Fold change from baseline*	0.78 (0.10, 5.43)	0.91 (0.41, 22.73)	0.82 (0.43, 8.66)

*Values = 1 indicate no change, values >1 indicate a fold increase, values <1 indicate a fold decrease

- In pairs of influenza isolates obtained at baseline and up to 9 days post therapy, there were no meaningful decreases in susceptibility in any treatment group

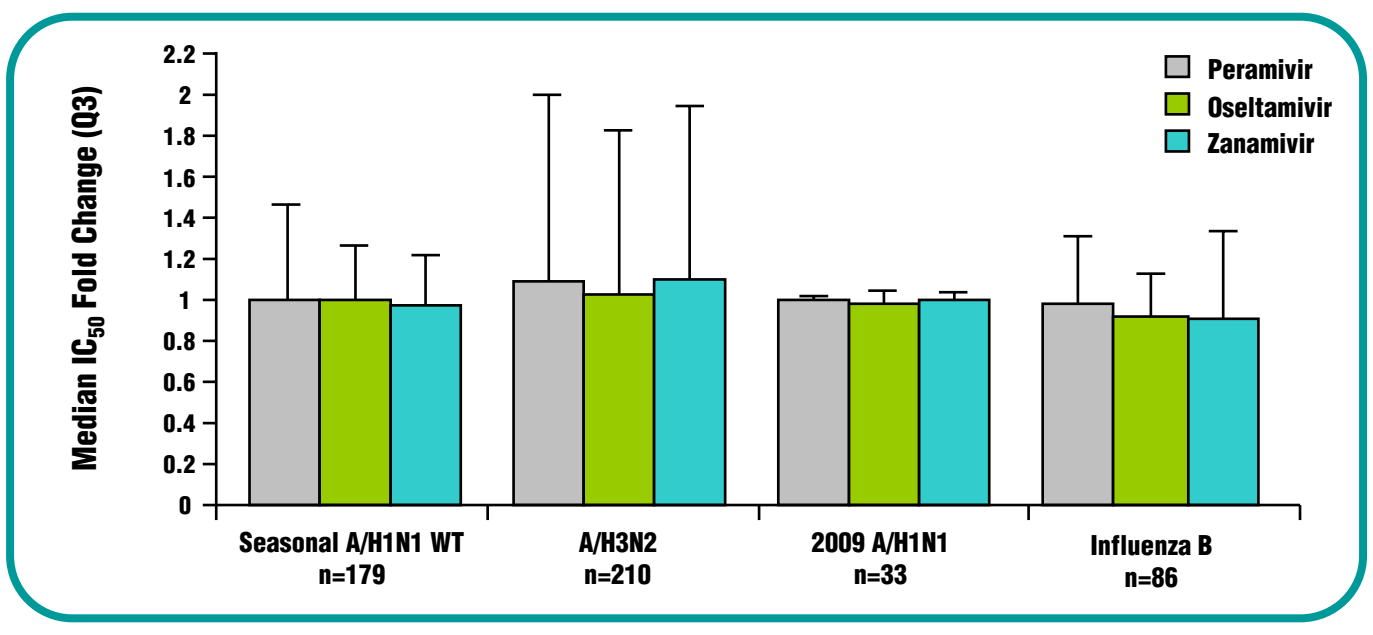

Median IC_{50} Fold Change (Q3)
Peramivir
Oseltamivir
Zanamivir
Seasonal A/H1N1 WT n=179
A/H3N2 n=210
2009 A/H1N1 n=33
Influenza B n=86

- When the subjects from all 4 studies and treatment groups are assessed, there were no meaningful changes in viral susceptibility for subjects with any viral influenza strain

Genotypic Analysis

Subjects in Peramivir Clinical Trials with Post-Baseline Treatment-Emergent H275Y Mutation

Subject	Study	Treatment	Sex	Age	Influenza Subtype	Peramivir IC_{50} (nM) Baseline	Peramivir IC_{50} (nM) Post-Rx*	Fold Change from BL
662-007	211	PVR 150 IM single dose	F	20	A/H1N1	0.12	8.34	70
671-004**	211	PVR 300 IM single dose	F	27	A/H1N1	5.52	1.09	0.2
080-1	621	PVR 600 IV single dose	F	38	A/H1N1	0.97	14.5	15
086-5	621	PVR 600 IV single dose	F	39	A/H1N1	1.72	31.5	18
135-6	621	PVR 600 IV single dose	F	21	A/H1N1	1.35	27.6	20
148-4	621	PVR 600 IV single dose	M	22	A/H1N1	1.12	27.8	25
163-5	621	PVR 600 IV single dose	F	25	A/H1N1	1.47	30.3	21
143-003	303	PVR 300 BID IV 5-10 d	F	67	2009 A/H1N1	0.01	31.02	3102

IV = intravenous, IM = intramuscular, BL = baseline
*Last positive culture
**Last positive culture for this subject (Day 5) did not yield a result for sequence analysis. Sequence performed on Day 3 specimen

- Genotypic analyses of the limited number of isolates from single dose studies (paired isolates with change in IC_{50} > mean + 2 SD, n=31; subjects with viral shedding post baseline, n=17) found 7 subjects (1.6%) with treatment-emergent mutations, all of which were H275Y substitutions in the NA gene. No additional known resistance mutations were identified. In study 621, there was no difference in clinical outcomes when subjects with the H275Y mutation post therapy were compared to those without (data not shown).[9]
- In the 5-10 day studies, genotyping found only 1 subject (0.5%) with a treatment-emergent H275Y mutation out of those tested (paired isolates with change in IC_{50} > mean + 2 SD, n=13; subjects with viral shedding post baseline, n=18); that subject had received oseltamivir treatment prior to the study. No additional known resistance mutations were identified.

DISCUSSION

- The development of viral resistance is a matter of concern
- The H275Y mutation has been prevalent in OSE-resistant seasonal influenza A strains, although there have been few reports with peramivir treatment
- Currently, approximately 1% of 2009 H1N1 influenza A strains with oseltamivir resistance have been reported; however, one mutation (S247N) caused a small shift in susceptibility to oseltamivir and zanamivir, but not to peramivir[10]
- Combining viral susceptibility data from 4 large clinical trials, there were no meaningful changes in any treatment group or with any influenza strain
- 8 treatment-emergent H275Y mutations were observed (approximately 1% of those with confirmed influenza and paired samples), all from the subset with paired isolates with change in NAI IC_{50} from baseline to last timepoint > mean + 2 SD

CONCLUSIONS

- Baseline mean IC_{50}s for influenza A/H1N1 (wild type), A/H3N2, 2009 A/H1N1, and influenza B were as previously reported with the order being peramivir<zanamivir<oseltamivir
- In this study combining viral susceptibility data from 4 large clinical trials, little change in IC_{50} values was observed with treatment
- 8 treatment-emergent mutations were observed (approx. 1%)
- There was little evidence of resistance in influenza viruses after treatment with up to 10 days of peramivir in patients with confirmed influenza

ACKNOWLEDGMENTS

The authors acknowledge the study participants, investigators, study site personnel, and BioCryst/Shionogi study teams who participated in these 4 clinical trials; HHS/BARDA for study support (Advanced Development Contract HHS0100200700032C); and the assistance of Elizabeth Field, PhD, in preparing the poster, which was funded by BioCryst.

REFERENCES

1. Lee N, Choi KW, Chan PK, et al. Outcomes of adults hospitalized with severe influenza. Thorax 2010; 65:510-5
2. Hurt AC, Ernest J, Deng YM, et al. Emergence and spread of oseltamivir-resistant A(H1N1) influenza viruses in Oceania, South East Asia and South Africa. Antiviral Res 2009; 83:90-3
3. Boivin G, Goyette N. Susceptibility of recent Canadian influenza A and B virus isolates to different neuraminidase inhibitors. Antiviral Res 2002; 54:143-7
4. Gubareva LV, Trujillo A, Okomo-Adhiambo M, et al. Comprehensive assessment of 2009 pandemic influenza A (H1N1) virus drug susceptibility in vitro. Antiviral Ther 2010; 15:1151–9
5. Baz M, Abed Y, Boivin G. Characterization of drug-resistant recombinant influenza A/H1N1 viruses selected in vitro with peramivir and zanamivir. Antiviral Res 2007; 74:159-62.
6. Ison MG, McGeer AJ, Hui DS, et al. Safety and efficacy of multiple-day treatment with intravenous peramivir or oral oseltamivir in hospitalized adults with acute influenza. Presented at XI International Symposium on Respiratory Viral Infections, Bangkok, February 2009
7. Kohno S, Kida H, Mizuguchi M, et al. Efficacy and safety of intravenous peramivir for treatment of seasonal influenza virus infection. Antimicrob Agents Chemother 2010; 54:4568-74.
8. Alexander WJ. Peramivir: An investigational antiviral therapy for seasonal and pandemic infuenza. Presented at X International Symposium on Respiratory Viral Infections, March 2008
9. Yoshida R, Kohno S, Kida H, Sugaya N. Neuraminidase sequence analysis and susceptibilities to neuraminidase (NA) inhibitors of influenza virus isolated from peramivir clinical studies. Presented at the International Union of Microbiological Societies 2011 Congress. 2011 Sept 11-16. Sapporo, Japan.
10. Hurt AC, Lee RT, Leang SK, et al. Increased detection in Australia and Singapore of a novel influenza A(H1N1)2009 variant with reduced oseltamivir and zanamivir sensitivity due to a S247N neuraminidase mutation. Eurosurv 2011:16(23)

Supported by HHS/BARDA Advanced Development Contract HHS0100200700032C

ISIRV 2011, Rio de Janeiro