Antiviral Drug Efficacy Evaluation Against Pandemic Influenza A (H1N1) Viruses In MDCK Cells

E. Bart Tarbet[1], Brett L. Hurst[1], Dale L. Barnard[1], Y.S. Babu[2], Yousuke Furuta[3], and Donald F. Smee[1]

[1]Institute for Antiviral Research, Utah State University, Logan, UT, [2]BioCryst Pharmaceuticals, Birmingham, AL, USA, [3]Research Laboratories, Toyama Chemical Co., Ltd., 2-4-1 Shimookui, Toyama 930-8508, Japan

ABSTRACT

New and emerging influenza virus strains, such as the pandemic influenza A (H1N1) virus, require constant vigilance for antiviral drug sensitivity and resistance. These studies evaluated eight isolates of pandemic influenza A virus against seven antiviral drugs: oseltamivir, peramivir, zanamivir, T-705 (favipiravir), ribavirin, amantadine and rimantadine. The pandemic viruses tested include isolates from California, New York, Utah, and an oseltamivir-resistant isolate from Hong Kong. In addition, a mouse-adapted A/CA/04/2009 virus was evaluated. EC_{90} values for each drug were determined by virus yield reduction assay in three replicate experiments. A broad range of drug sensitivities was observed among the H1N1 viruses tested. However, significantly higher ($P < 0.05$) EC_{90} values for the neuraminidase inhibitors were only observed for the drug-resistant A/Hong Kong/2369/2009 virus. As expected, all isolates showed resistance to the adamantanes. Dose-response curves also showed that the mouse-adapted strain had consistently higher EC_{90} values compared to its non-adapted counterpart. We also evaluated the effects of antiviral drug combinations on drug-resistant influenza A/Hong Kong/2369/2009 virus. Drug combinations included T-705 plus one of the neuraminidase inhibitors: oseltamivir, peramivir, or zanamivir. Synergism was observed involving three doses of T-705 (1.0, 3.2, and 10.0 µM) for all three neuraminidase inhibitors. Oseltamivir and zanamivir were synergistic with T-705 at three concentrations (3.2, 10, and 32 µM for oseltamivir, or 0.1, 0.32, and 1.0 µM for zanamivir) and peramivir showed synergism with T-705 at two concentrations (0.32 and 1.0 µM). These studies demonstrate that pandemic influenza A virus isolates from different regions remain sensitive in vitro to commonly used antiviral drugs, and that combination chemotherapy holds promise as a treatment strategy for drug-resistant influenza virus. [This work was supported by Contract N01-AI-30048 from the Virology Branch, DMID, NIAID, NIH]

INTRODUCTION

New influenza virus strains, such as pandemic influenza A (H1N1) virus, require constant vigilance for antiviral drug sensitivity and resistance. These studies evaluated nine isolates of pandemic influenza A virus against seven antiviral drugs: Oseltamivir, Peramivir, Zanamivir, Ribavirin, T-705 (Favipiravir), Amantadine and Rimantadine. The pandemic viruses tested include isolates from California, Utah, New York, New Zealand (Southern Hemisphere), and an Oseltamivir-resistant isolate from Hong Kong. In addition, the effectiveness of antiviral drug combinations was evaluated. Combination chemotherapy holds promise as a treatment strategy superior to that of monotherapy, especially for treatment of drug-resistant virus infections.

MATERIALS AND METHODS

Antiviral Compounds: Oseltamivir carboxylate was provided by Dr. Nguyen (Adamas Pharmaceuticals, Emeryville, CA). Peramivir was provided by Dr. Babu (BioCryst Pharmaceuticals, Inc). Zanamivir was obtained from Haorui Pharma-Chem Inc. (Edison, New Jersey). T-705 was provided by Dr. Furuta (Toyama Chemical Co.). Ribavirin was obtained from ICN Pharmaceuticals (Costa Mesa, CA). Amantadine and rimantadine were purchased from Sigma (St. Louis, MO). The compounds were dissolved in cell culture medium for antiviral testing.

Viruses: Each virus was passaged one time in Madin-Darby canine kidney (MDCK) cells and then titrated for antiviral experiments. 1) Influenza A/California/04/2009 (H1N1) virus was received from Dr. Daniel Perez, Department of Veterinary Medicine, University of Maryland, College Park, MD. 2) Influenza A/California/04/2009 (H1N1) was received from Dr. Elena Govorkova, Department of Infectious Diseases, St. Jude Children's Research Hospital, Memphis TN. 3) Influenza A/California/07/2009 (H1N1) virus was provided by the Centers for Disease Control and Prevention (Atlanta, GA). 4) Influenza A/New York/18/2009 (H1N1) virus was provided by the Centers for Disease Control and Prevention (Atlanta, GA). 5) Influenza A/Utah/475/2009 (H1N1) virus, 6) Influenza A/Utah/476/2009 (H1N1) virus, and 7) Influenza A/Utah/727/2009 (H1N1) virus were isolated from pediatric patients at the University of Utah Medical Center, Salt lake City, UT. 8) Influenza A/Hong Kong/2369/2009 (Oseltamivir-resistant H1N1) virus was provided by the Centers for Disease Control and Prevention (Atlanta, GA). 9) Influenza A/Auckland/3/2009 (H1N1) provided by Dr. Angela Luttick, Biota Holdings Limited (Australia).

Cell culture antiviral studies. Antiviral activity of each drug or drug combination was determined in MDCK cells. The assays were completed in 96-well microplates infected with approximately 50 cell culture infectious doses ($CCID_{50}$) of virus. Microplates were visually examined after 3 d of infection and then treated for 2 h with neutral red (0.011%) to quantify the virus-induced cytopathic effects (CPE). Excess dye was rinsed from cells with PBS. The absorbed dye was eluted from the cells by addition of 0.1 ml 50% Sorensen's citrate buffer/50% ethanol added to each well. Optical density (OD) measurements were completed on the microtiter plates at 560 nm. OD readings were converted to percent of uninfected control using an Excel spread sheet. Fifty percent virus-inhibitory concentrations (EC_{50} values) were determined by plotting CPE versus $\log_{10}$ of inhibitor concentration.

Virus yield reduction (VYR) assays were performed in 96-well microplates infected with approximately 50 $CCID_{50}$ of virus by quantifying virus yield. Virus yields were determined from samples collected on day 3 of the infections when untreated control microwells exhibited 100% CPE. To quantify the amount of virus produced in the wells, microplate samples (pooled from replicate wells) were titrated in new 96-well plates of MDCK cells by 10-fold serial dilution. Four wells were used per dilution. The samples were centrifuged for 10 min at 4000 rpm to pellet cell debris. Plates were visually examined for appearance of virus-induced CPE. Wells were indicated as plus or minus virus, and virus titers calculated by endpoint dilution method (1, 2).

Drug combination effects were determined by two methods. One method identified possible regions of synergy in the VYR data based upon the following definitions: <u>synergy</u> defined as a one log (or greater) decrease in virus titer when compared to both individual drugs alone; an <u>additive effect</u> defined as a consistent decrease in virus titer demonstrated in replicate tests; <u>antagonism</u> defined as a one log (or greater) increase in virus titer; an <u>inhibitory effect</u> defined as a consistent increase in virus titer; and indifferent defined as no clear synergy or antagonism. In addition, three replicates are considered a minimum number of VYR tests required to determine synergy, additive, antagonistic, or indifferent effects. A second method determined the effects of drug combinations by the three-dimensional analysis (MacSynergy™ II) of Prichard and Shipman (3).

Statistical analysis: Mean EC_{50} values for the antiviral drugs tested against the nine pandemic (H1N1) viruses were analyzed by one-way (within group) or two-way (between groups) ANOVA followed by Bonferroni's multiple comparison tests using Prism 5.0b (GraphPad Software Inc.).

REFERENCES

1. Smee, D.F., J.H. Huffman, A.C. Morrison, D.L. Barnard, and R.W. Sidwell. 2001. Cyclopentane neuraminidase inhibitors with potent in vitro anti-influenza virus activity. Antimicrob. Agents Chemother. 45:743-748.
2. Reed, L.J., and H. Muench. 1938. A simple method of estimating fifty percent endpoints. Am. J. Hyg. 27:493-497.
3. Prichard, M. N., and C. Shipman, Jr. 1990. A three dimensional model to analyze drug-drug interactions. Antiviral Res. 14:181-206.

Table 1: Pandemic Influenza A (H1N1) Virus *In Vitro* Testing Results

Virus	Effective Concentration (µM)													
	Oseltamivir		Peramivir		Zanamivir		T-705		Ribavirin		Amantadine		Rimantadine	
	EC_{50}	EC_{90}	EC_{50}	EC_{90}	EC_{50}	EC_{90}	EC_{50}	EC_{90}	EC_{50}	EC_{90}	EC_{50}	EC_{90}	EC_{50}	EC_{90}
CA/04/2009	0.12 ± 0.09	0.07	0.004 ± 0.001	0.01	0.02 ± 0.007	0.07 ± 0.08	2.7 ± 2.6	3.7 ± 1.4	16.5 ± 0.7	19.3 ± 3.7	>100	>100	>10	>10
CA/04/2009 (Mouse Adapted)	0.3 ± 0.09	1.4 ± 0.5	0.11 ± 0.02	0.83 ± 0.25	0.3 ± 0.09	0.93 ± 1.1	6.8 ± 4.2	9.2 ± 0.6	18.5 ± 0.7	23.3 ± 3.2	>100	>100	>10	>10
CA/07/2009	0.28 ± 0.12	0.32 ± 0.08	0.028 ± 0.04	0.48 ± 0.25	0.08 ± 0.004	0.4 ± 0.1	3.9 ± 0.9	4.1 ± 0.45	9.1 ± 1.3	15.2 ± 1.6	>100	>100	>10	>10
NY/18/2009	0.76 ± 0.16	1.8 ± 0.3	0.006	0.06 ± 0.06	0.09 ± 0.03	0.21 ± 0.16	4.3 ± 1.6	5.1 ± 0.6	14 ± 2.8	15.5 ± 4.4	>100	>100	>10	>10
UT/475/2009	0.2 ± 0.03	0.40 ± 0.05	0.024	0.11 ± 0.09	0.12 ± 0.06	0.35 ± 0.08	1.9	1.6 ± 0.7	17.5 ± 0.7	23.3 ± 8.9	>100	>100	>10	>10
UT/476/2009	0.99 ± 0.45	1.24 ± 0.2	0.054 ± 0.04	0.2 ± 0.1	0.54 ± 0.23	0.73 ± 0.9	5.2 ± 3.2	6.1 ± 1.1	20.5 ± 3.5	16.1 ± 5.5	>100	>100	>10	>10
UT/727/2009	0.68 ± 0.25	2.3 ± 0.9	0.013 ± 0.004	1.3 ± 2.3	0.12 ± 0.05	0.25 ± 0.07	4.3 ± 2.9	5.5 ± 4.1	17.5 ± 0.7	25.0 ± 5.6	>100	>100	>10	>10
Auckland/3/2009 (Southern Hemisphere)	1.1 ± 0.74	3.9 ± 0.23	0.03 ± 0.01	0.13 ± 0.6	0.54 ± 0.45	0.73 ± 0.18	7.8 ± 1.9	9.3 ± 1.8	19 ± 5.7	18.1 ± 3.8	>100	>100	>10	>10
HK/2369/2009 (Oseltamivir-Resistant)	>10	>10	>10	4.48 ± 2.4	1.4 ± 1.2	2.1 ± 1.4	7.8	10.8 ± 4.6	20 ± 2.8	18.2 ± 2.5	>100	>100	>10	>10

Figure 1: Statistical Analysis of *In Vitro* Testing Results

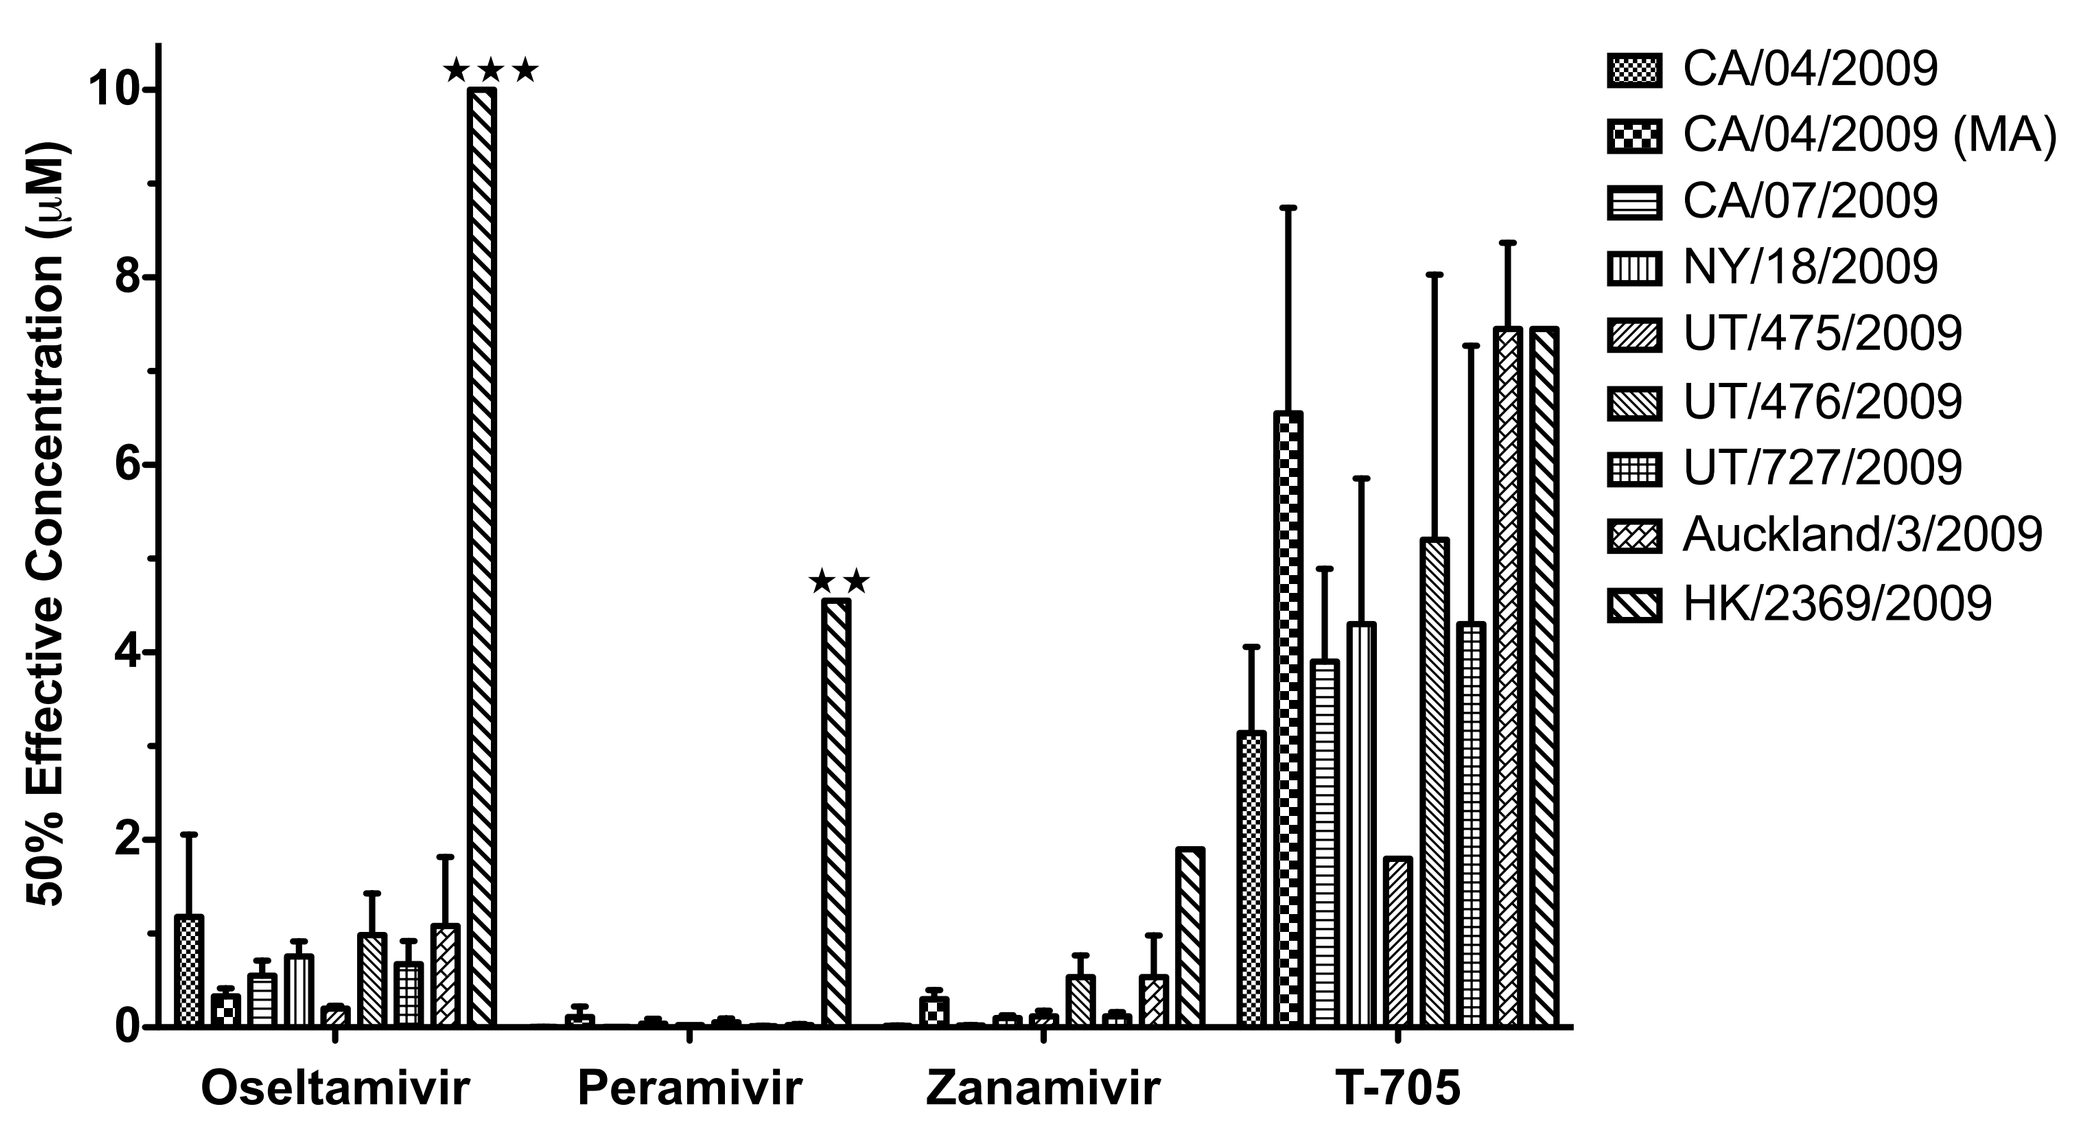
50% Effective Concentration (µM)
★★★
★★
Oseltamivir
Peramivir
Zanamivir
T-705
CA/04/2009
CA/04/2009 (MA)
CA/07/2009
NY/18/2009
UT/475/2009
UT/476/2009
UT/727/2009
Auckland/3/2009
HK/2369/2009

Figure 2: Effect of Mouse Adaptation (MA) on CA/04/2009 Virus

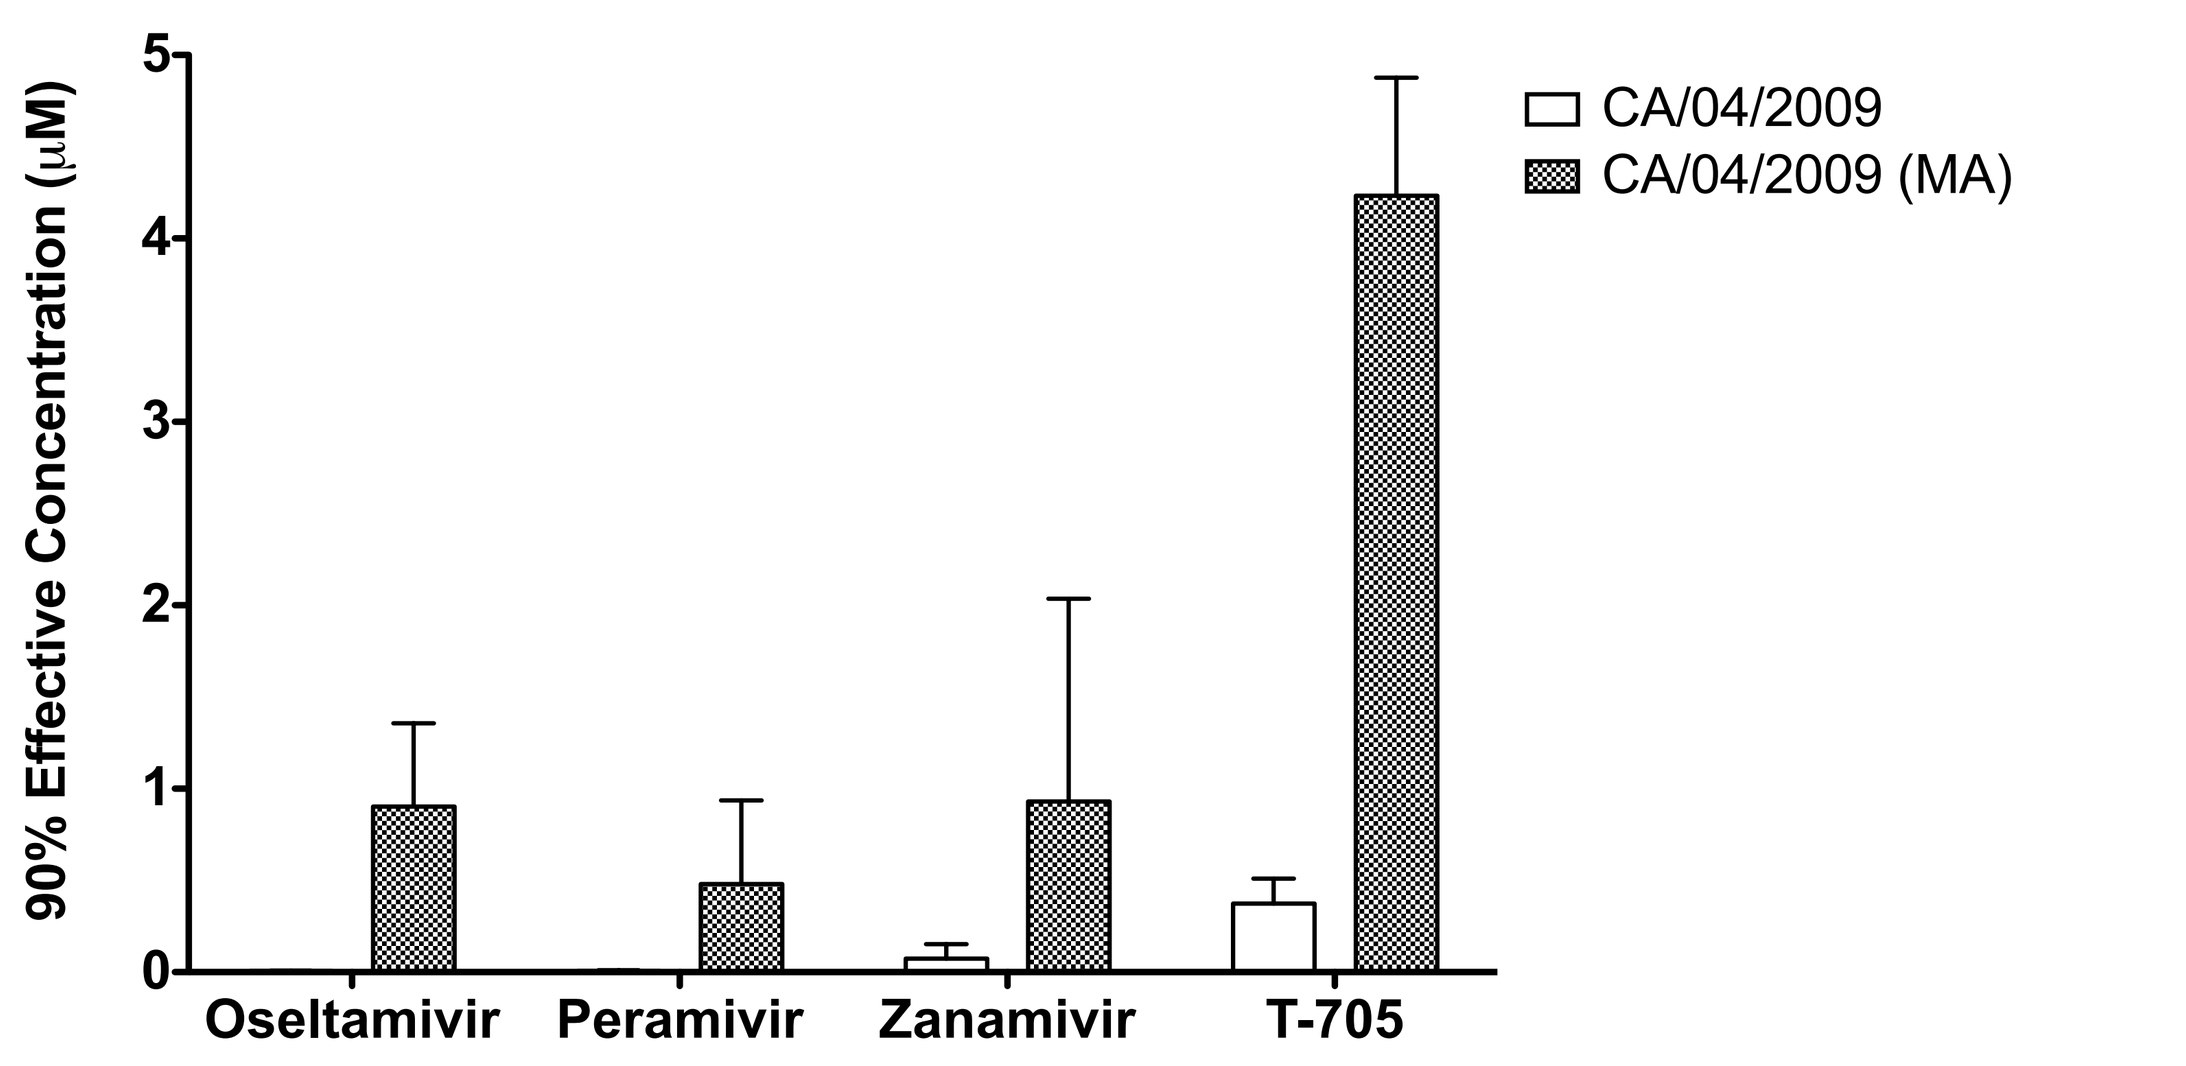
90% Effective Concentration (µM)
Oseltamivir
Peramivir
Zanamivir
T-705
CA/04/2009
CA/04/2009 (MA)

Figure 3: Reduction in Virus Yield by Oseltamivir

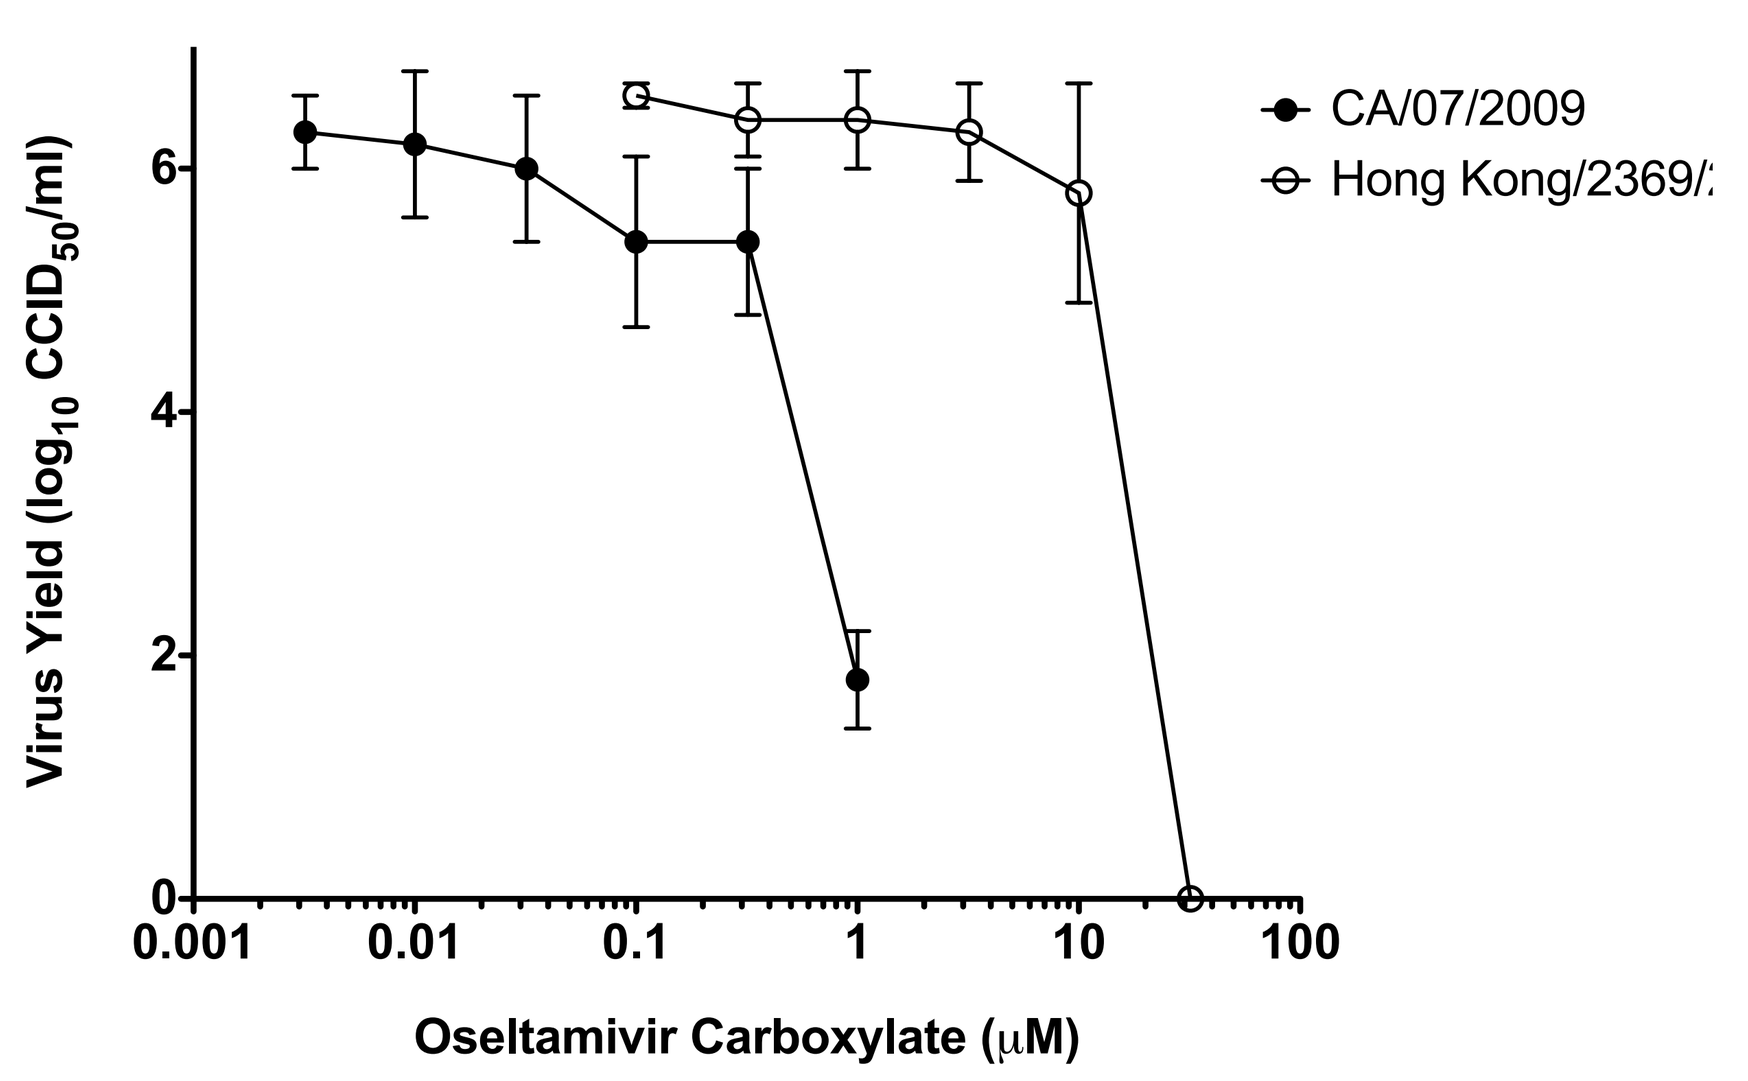
Virus Yield (log10 CCID50/ml)
Oseltamivir Carboxylate (µM)
CA/07/2009
Hong Kong/2369/

Figure 4: Reduction in Virus Yield by Peramivir

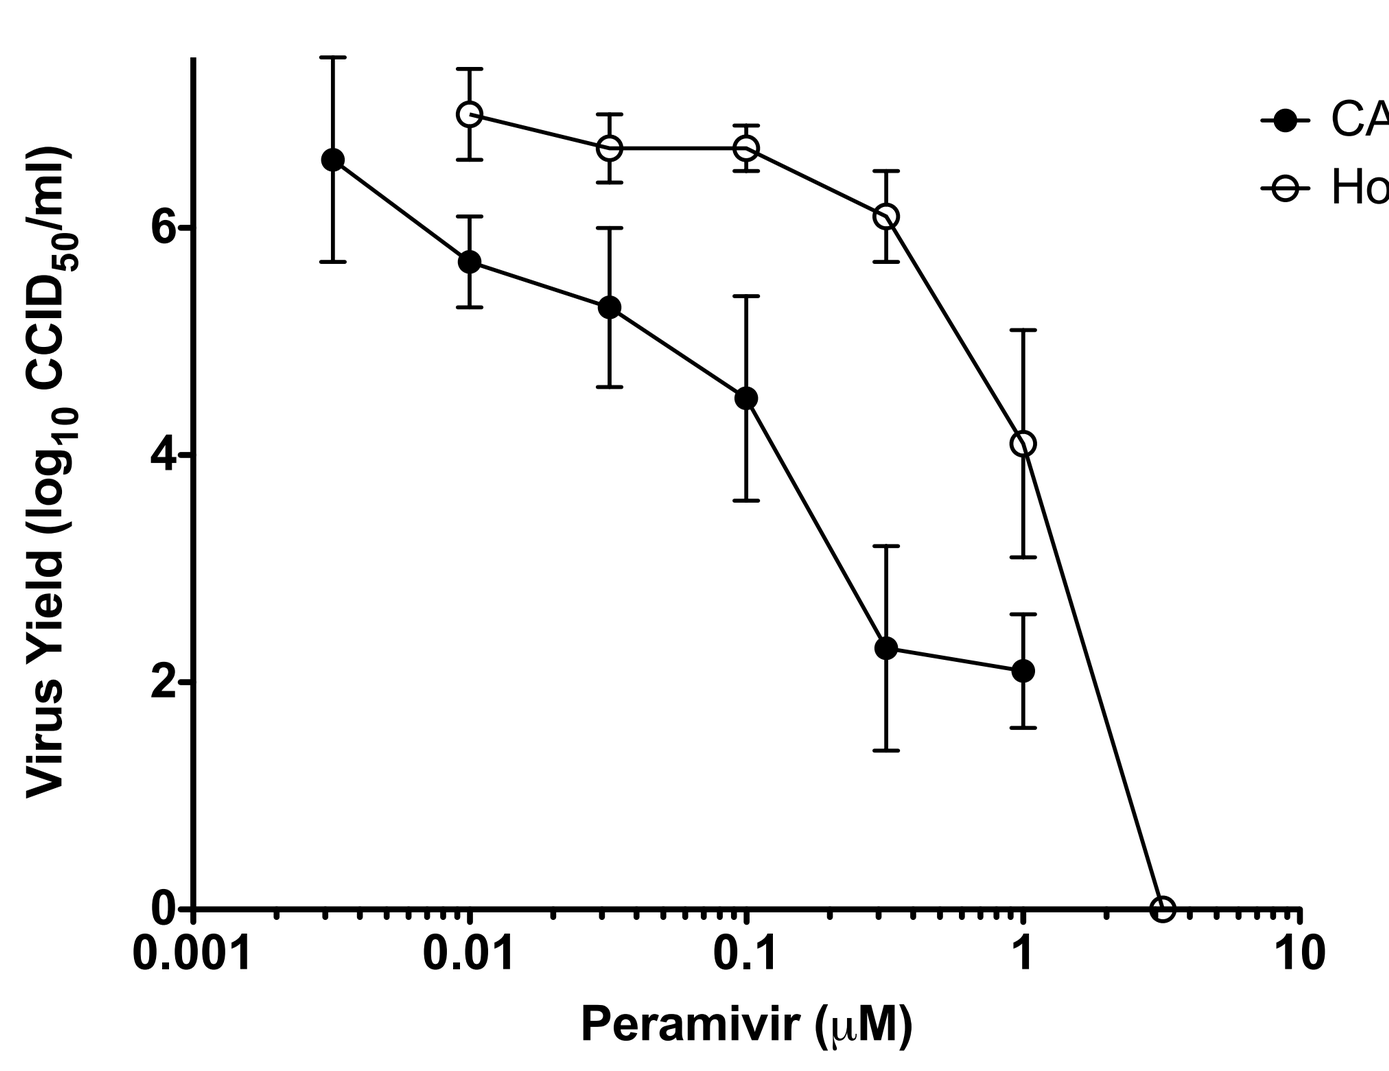
Virus Yield (log10 CCID50/ml)
Peramivir (µM)
CA/07/2009
Hong Kong/236

Figure 5: Reduction in Virus Yield by Zanamivir

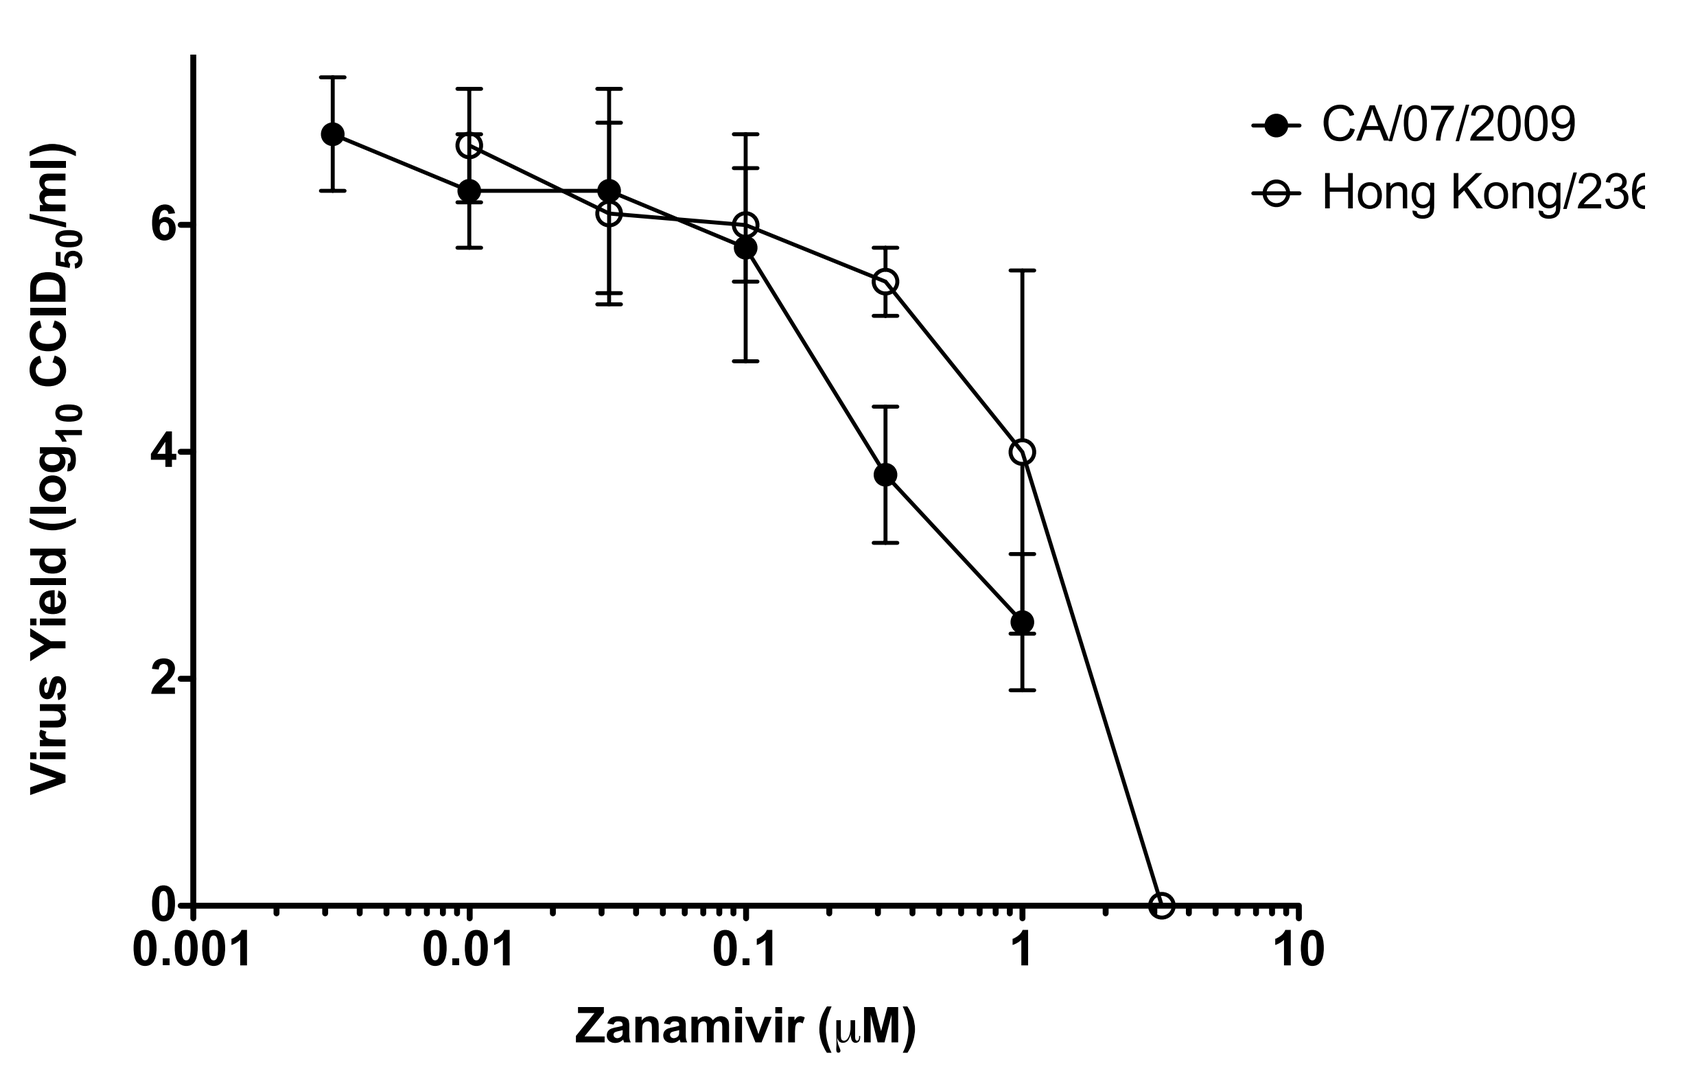
Virus Yield (log10 CCID50/ml)
Zanamivir (µM)
CA/07/2009
Hong Kong/236

Figure 6: Reduction in Virus Yield by T-705

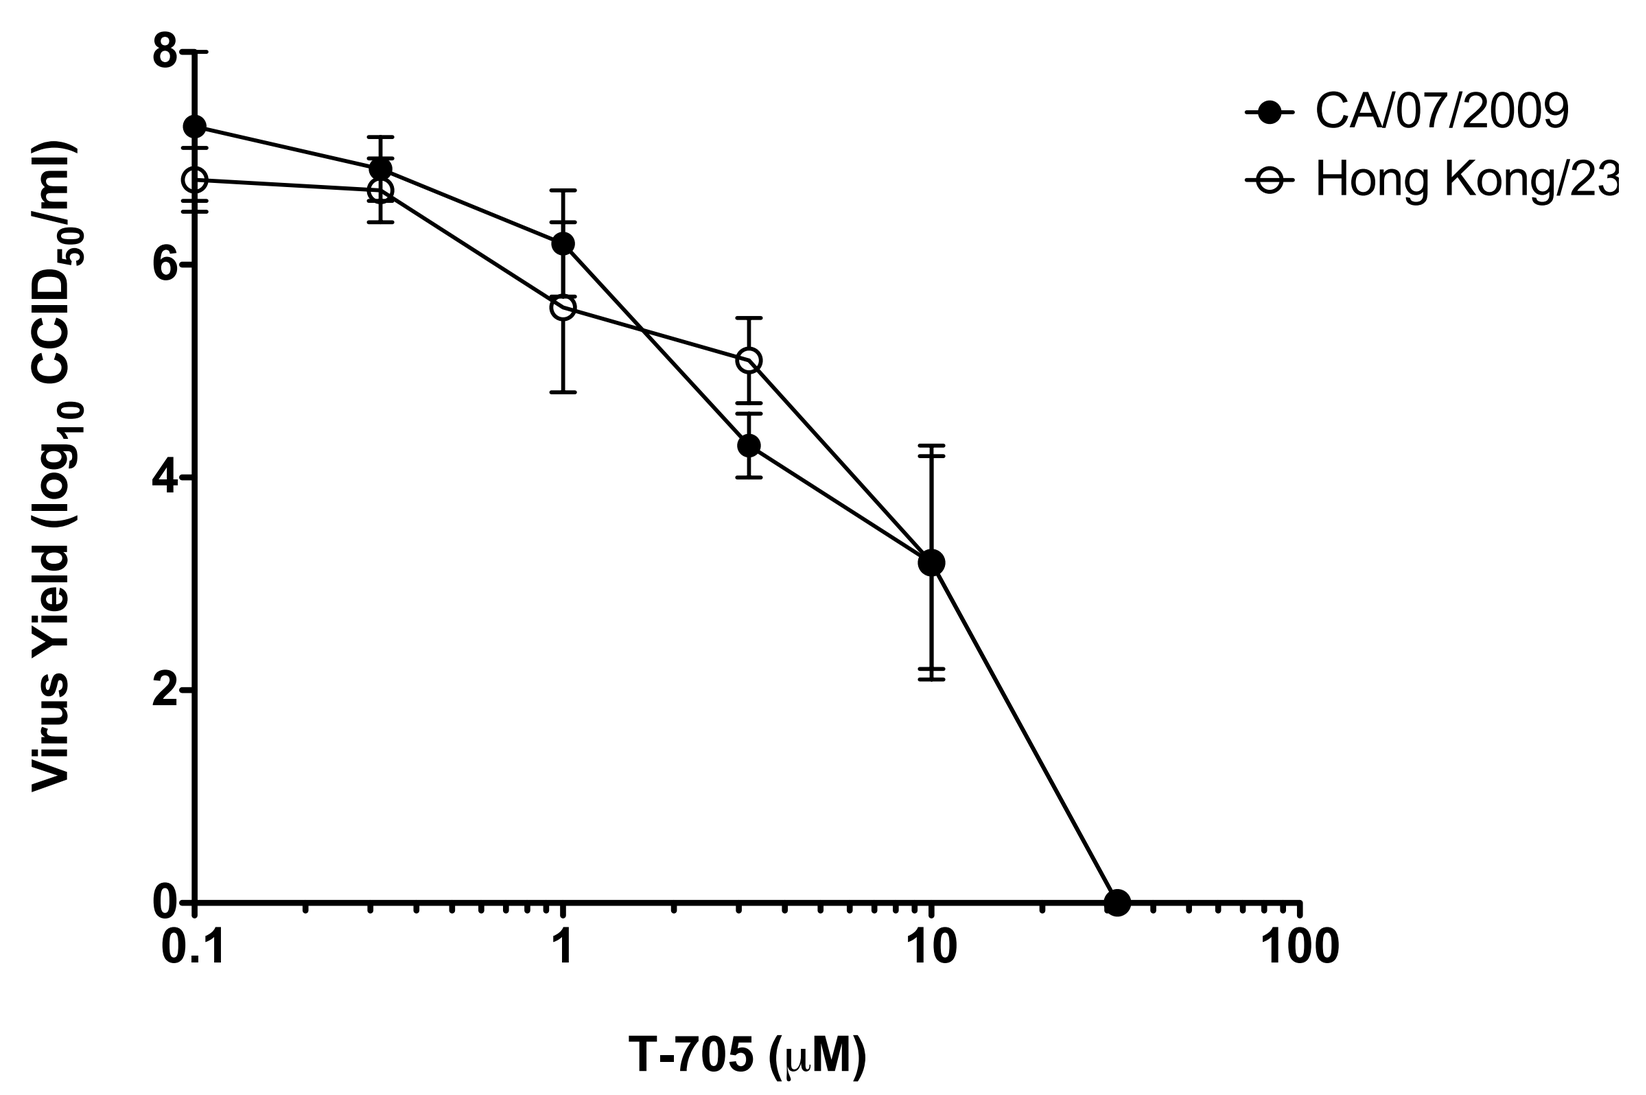
Virus Yield (log10 CCID50/ml)
T-705 (µM)
CA/07/2009
Hong Kong/23

Figure 7: Effect of Oseltamivir and T-705 on CA/07/2009 Virus

Oseltamivir carboxylate (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) T-705 (µM)						
	32	10	3.2	1	0.32	0.1	0
1.0	0	0	0	0	0	0	0
0.32	0	0	0	1.7 ± 1.6	3.2 ± 0.4	3.9 ± 0.5	4.4 ± 0.5
0.1	0	0	1.6 ± 1.8	3.0 ± 1.1	3.8 ± 0.4	5.2 ± 0.6	4.7 ± 0.6
0.032	0	0	2.2 ± 1.3	3.6 ± 0.8	5.1 ± 0.6	5.4 ± 0.3	5.6 ± 0.7
0.01	0	0	2.3 ± 1.5	4.2 ± 0.6	5.3 ± 0.7	5.6 ± 0.1	5.8 ± 0.5
0.0032	0	1.3 ± 1.3	2.7 ± 1.0	4.3 ± 0.7	5.9 ± 0.6	6.1 ± 0.3	6.0 ± 0.5
0	0	2.2 ± 1.1	3.3 ± 0.3	5.2 ± 0.5	5.9 ± 0.3	6.3 ± 0.7	6.2 ± 0.1

Oseltamivir carboxylate EC_{90} = 0.17 µM
T-705 EC_{90} = 1.0 µM

Range of Synergy by VYR analyses:
Oseltamivir (0.01 to 0.32 µM) and
T-705 (0.32 to 10 µM)

Synergy Plot:
Synergy = 28.6
(95% Confidence Interval = 44 - 13)
Antagonism = -1.97
(95% Confidence Interval = 0 - -4)

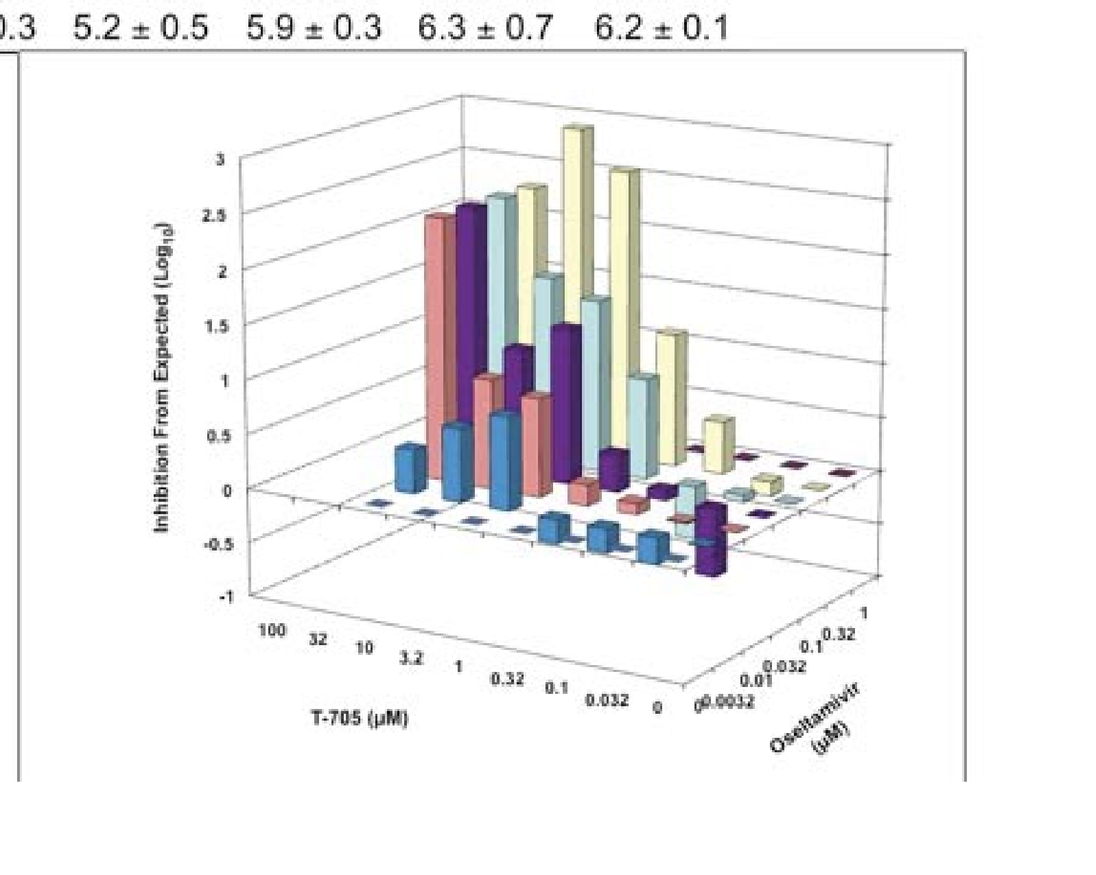
Inhibition From Expected (Log10)
T-705 (µM)
Oseltamivir (µM)

Figure 8: Effect of Peramivir and T-705 on CA/07/2009 Virus

Peramivir (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) T-705 (µM)						
	32	10	3.2	1	0.32	0.1	0
1.0	0.0	0.0	0.0	0.0	1.4 ± 2.5	1.3 ± 2.2	1.2 ± 2.1
0.32	0.0	0.0	0.0	0.0	1.2 ± 2.0	1.4 ± 2.5	1.3 ± 2.3
0.1	0.0	0.0	0.6 ± 1.0	0.4 ± 0.7	2.3 ± 1.1	3.3 ± 3.1	3.5 ± 2.0
0.032	0.0	0.0	0.3 ± 0.4	1.5 ± 1.3	3.1 ± 1.0	4.3 ± 1.8	4.3 ± 0.7
0.01	0.0	0.3 ± 0.4	0.9 ± 1.6	2.2 ± 0.6	3.3 ± 0.9	4.7 ± 1.1	4.7 ± 0.4
0.0032	0.0	0.0	1.7 ± 1.5	2.5 ± 1.1	4.1 ± 0.6	5.1 ± 1.3	5.6 ± 0.9
0	0.0	1.2 ± 1.3	2.6 ± 1.0	5.2 ± 1.0	5.3 ± 0.6	5.7 ± 0.5	5.8 ± 0.6

Peramivir EC_{90} = 0.009 µM
T-705 EC_{90} = 1.2 µM

Range of Synergy by VYR analyses:
Peramivir (0.0032 to 1.0 µM) and
T-705 (0.32 to 10 µM)

Synergy Plot:
Synergy = 35.13
(95% Confidence Interval = 55 - 16)
Antagonism = -1.01
(95% Confidence Interval = not signif.)

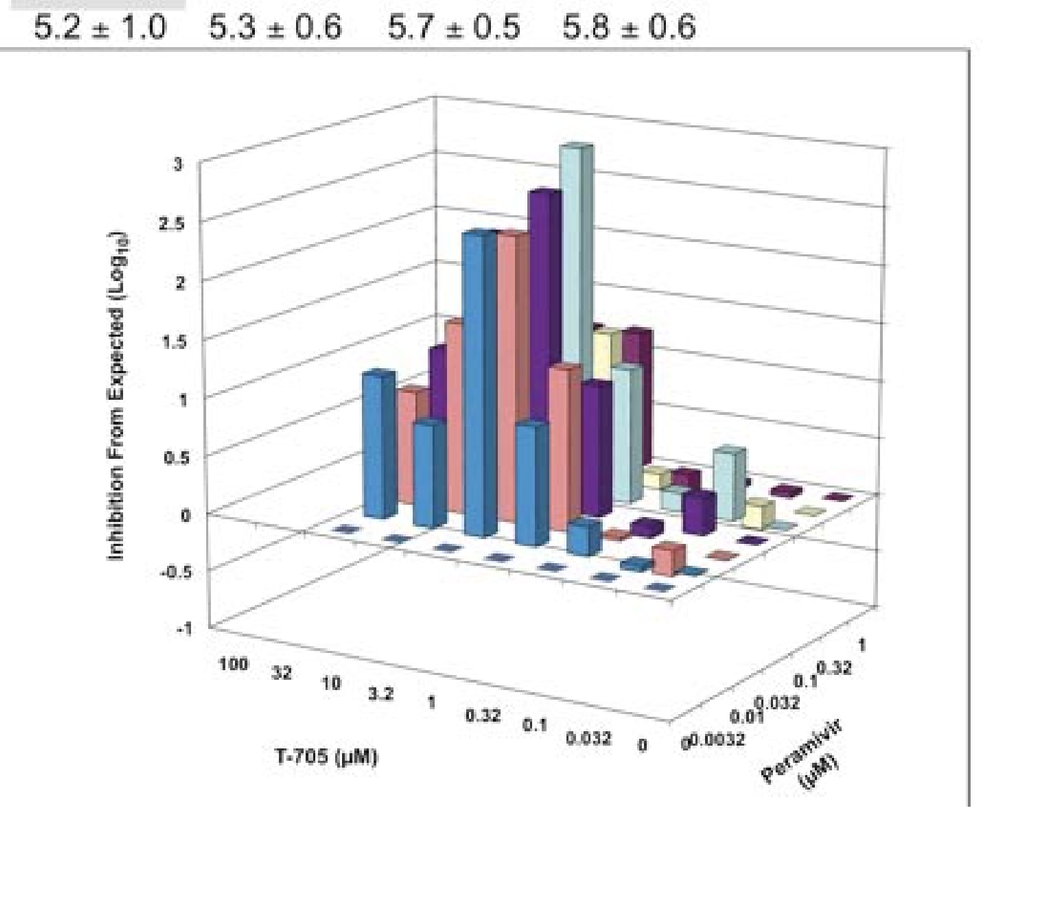
Inhibition From Expected (Log10)
T-705 (µM)
Peramivir (µM)

Figure 9: Effect of Zanamivir and T-705 on CA/07/2009 Virus

Zanamivir (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) T-705 (µM)						
	32	10	3.2	1	0.32	0.1	0
1.0	0	0.3 ± 0.4	0	0.8 ± 1.3	1.1 ± 1.9	1.2 ± 2.0	1.5 ± 2.6
0.32	0	0.3 ± 0.4	0	1.4 ± 1.8	2.0 ± 1.8	2.8 ± 2.5	2.8 ± 2.6
0.1	0	0.0	0.6 ± 1.0	3.4 ± 0.8	3.8 ± 1.6	5.0 ± 1.4	4.8 ± 1.0
0.032	0	0.3 ± 0.4	0.4 ± 0.7	3.8 ± 1.3	4.3 ± 0.7	5.3 ± 1.4	5.3 ± 0.9
0.01	0	0.3 ± 0.4	1.7 ± 0.1	4.0 ± 0.7	4.5 ± 0.9	5.2 ± 0.9	5.3 ± 0.5
0.0032	0	0.7 ± 0.6	1.9 ± 0.7	3.8 ± 0.8	4.9 ± 0.7	5.3 ± 0.5	5.8 ± 0.5
0	0	0.5 ± 0.5	3.3 ± 0.7	4.7 ± 1.0	5.3 ± 0.6	5.9 ± 0.6	5.9 ± 0.7

Zanamivir EC_{90} = 0.067 µM
T-705 EC_{90} = 0.61 µM

Range of Synergy by VYR analyses:
Zanamivir (0.0032 to 1.0 µM) and
T-705 (0.32 to 3.2 µM)

Synergy Plot:
Synergy = 32.9
(95% Confidence Interval = 59 - 6)
Antagonism = -0.62
(95% Confidence Interval = not signif.)

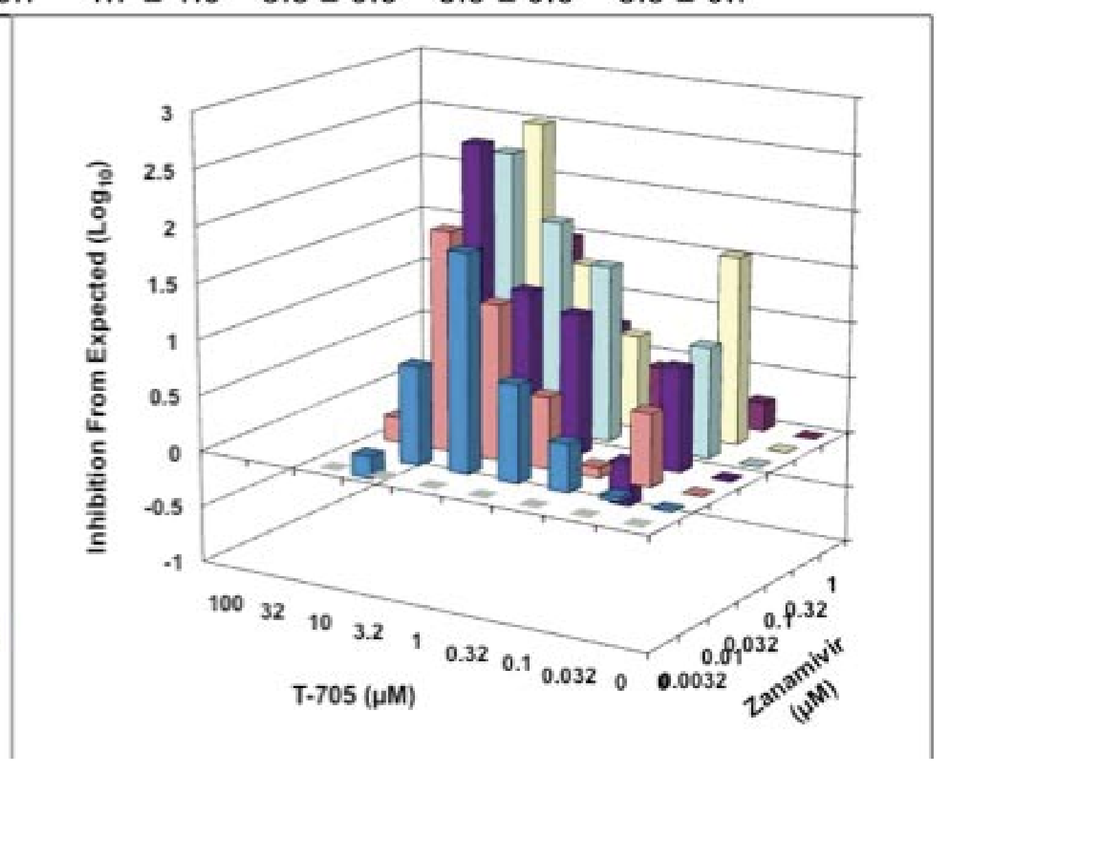
Inhibition From Expected (Log10)
T-705 (µM)
Zanamivir (µM)

Figure 10: Effect of Oseltamivir and Zanamivir on CA/07/2009 Virus

Oseltamivir Carboxylate (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) Zanamivir (µM)						
	1.0	0.32	0.1	0.032	0.01	0.0032	0
1.0	0	1.2 ± 2.0	1.0 ± 1.7	1.2 ± 2.0	1.2 ± 2.1	1.5 ± 2.6	1.4 ± 2.2
0.32	2.9 ± 2.5	4.2 ± 0.4	4.2 ± 0.5	4.1 ± 0.5	4.5 ± 0.8	4.2 ± 0.5	4.3 ± 0.4
0.1	3.2 ± 2.7	3.9 ± 0.7	4.0 ± 0.7	4.9 ± 0.5	4.9 ± 0.5	4.9 ± 1.0	4.8 ± 0.2
0.032	3.1 ± 2.7	4.6 ± 0.3	4.8 ± 0.6	5.0 ± 0.4	5.0 ± 0.4	5.2 ± 0.5	5.4 ± 0.6
0.01	2.3 ± 2.0	4.1 ± 1.2	4.6 ± 0.8	4.7 ± 0.9	5.1 ± 1.0	5.2 ± 0.6	5.2 ± 0.6
0.0032	2.5 ± 2.2	4.1 ± 0.7	4.3 ± 0.5	4.7 ± 0.9	5.1 ± 1.3	5.1 ± 0.5	5.5 ± 0.4
0	4.3 ± 0.3	4.7 ± 0.6	4.7 ± 0.3	5.1 ± 0.4	5.2 ± 0.7	5.4 ± 0.3	5.7 ± 0.2

Oseltamivir carboxylate EC_{90} = 0.12 µM
Zanamivir EC_{90} = 0.1 µM

Range of Synergy by VYR analyses:
Oseltamivir (0.0032 to 1.0 µM) and
Zanamivir (1.0 µM)

Synergy Plot:
Synergy = 12.78
(95% Confidence Interval = 24 - 1)
Antagonism = -1.93
(95% Confidence Interval = not signif.)

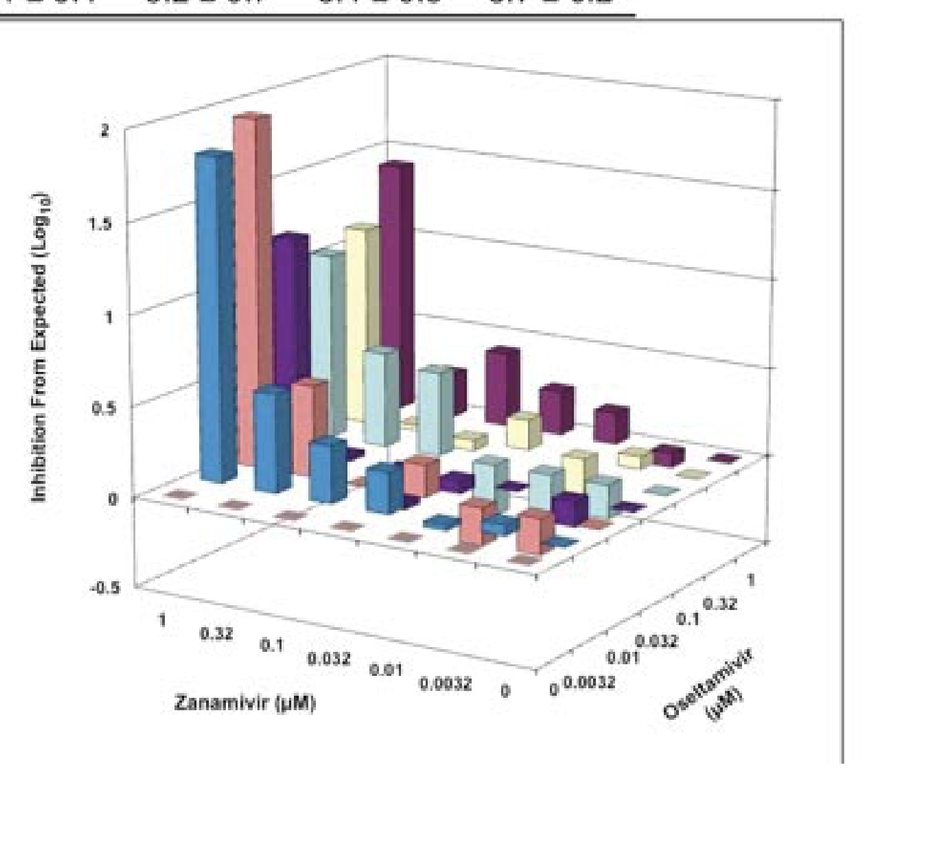
Inhibition From Expected (Log10)
Zanamivir (µM)
Oseltamivir (µM)

Figure 11: Effect of Oseltamivir and T-705 on HK/2639/2009 Virus

Oseltamivir Carboxylate (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) T-705 (µM)								
	100	32	10	3.2	1	0.32	0.1	0.032	0
32	0	0	0	0.9 ± 1.6	1.2 ± 2.0	1.8 ± 3.2	2.1 ± 3.6	1.5 ± 2.6	1.8 ± 3.0
10	0	0	0	1.8 ± 1.5	2.9 ± 2.5	4.4 ± 1.0	4.8 ± 1.0	4.9 ± 1.2	4.8 ± 0.9
3.2	0	0	1.2 ± 2.0	2.5 ± 1.5	3.8 ± 1.3	5.3 ± 1.1	5.2 ± 0.4	5.0 ± 0.7	5.3 ± 0.4
1.0	0	0	1.1 ± 1.9	3.5 ± 1.0	4.3 ± 1.4	5.5 ± 1.0	5.5 ± 1.0	5.5 ± 1.0	5.4 ± 0.4
0.32	0	0	1.3 ± 2.2	3.7 ± 1.0	4.4 ± 0.9	5.7 ± 0.5	5.8 ± 0.6	6.1 ± 0.3	5.4 ± 0.3
0.1	0	0	1.1 ± 1.9	3.8 ± 0.9	4.8 ± 0.5	5.5 ± 0	5.8 ± 0.6	5.8 ± 0.4	5.6 ± 0.1
0	0	0	1.1 ± 2.0	3.9 ± 0.9	4.8 ± 0.4	5.7 ± 0.4	5.6 ± 0.4	5.8 ± 0.3	5.7 ± 0.3

Oseltamivir carboxylate EC_{90} = 11.0 µM
T-705 EC_{90} = 1.2 µM

Range of Synergy by VYR analyses:
Oseltamivir (3.2 to 32 µM) and
T-705 (1.0 to 10 µM)

Synergy Plot:
Synergy = 12.8
(95% Confidence Interval = 23 - 2)
Antagonism = -4.8
(95% Confidence Interval = not signif.)

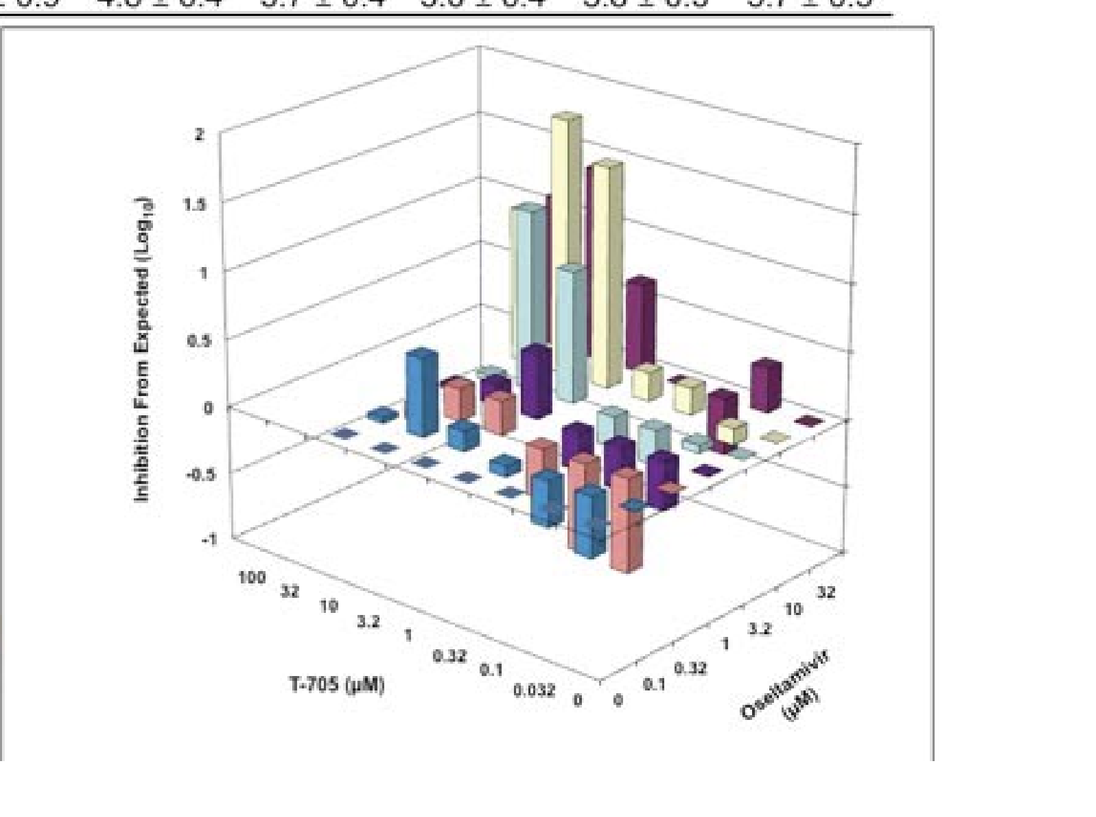
Inhibition From Expected (Log10)
T-705 (µM)
Oseltamivir (µM)

Figure 12: Effect of Peramivir and T-705 on HK/2639/2009 Virus

Peramivir (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) T-705 (µM)								
	100	32	10	3.2	1	0.32	0.1	0.032	0
3.2	0	0	0	0	0	0	0	0	0
1.0	0	0	0	0.9 ± 1.6	2.0 ± 1.8	3.4 ± 0.6	4.1 ± 0.5	4.4 ± 0.6	4.1 ± 1.0
0.32	0	0	0	2.5 ± 0.2	4.2 ± 0.4	4.9 ± 0.5	4.8 ± 0	5.0 ± 0.4	5.1 ± 0.4
0.1	0	0	0.8 ± 1.4	3.3 ± 0.5	4.5 ± 0.3	5.4 ± 0.1	5.5 ± 0.3	5.4 ± 0.3	5.7 ± 0
0.032	0	0	1.1 ± 1.9	3.4 ± 0.3	4.2 ± 0.4	5.2 ± 0.6	5.7 ± 0.1	5.8 ± 0.4	5.7 ± 0.3
0.01	0	0	1.3 ± 2.2	3.5 ± 0.3	4.8 ± 0.4	5.6 ± 0.6	6.1 ± 0.6	5.8 ± 0.4	6.0 ± 0.4
0	0	0	1.2 ± 2.1	3.6 ± 0.9	4.8 ± 0.4	5.4 ± 0.4	5.8 ± 0.7	5.7 ± 0.1	6.0 ± 0.3

Peramivir EC_{90} = 0.35 µM
T-705 EC_{90} = 0.65 µM

Range of Synergy by VYR analyses:
Peramivir (3.2 to 10 µM) and
T-705 (1.0 to 10 µM)

Synergy Plot:
Synergy = 11.7
(95% Confidence Interval = 20 - 3)
Antagonism = -2.47
(95% Confidence Interval = not signif.)

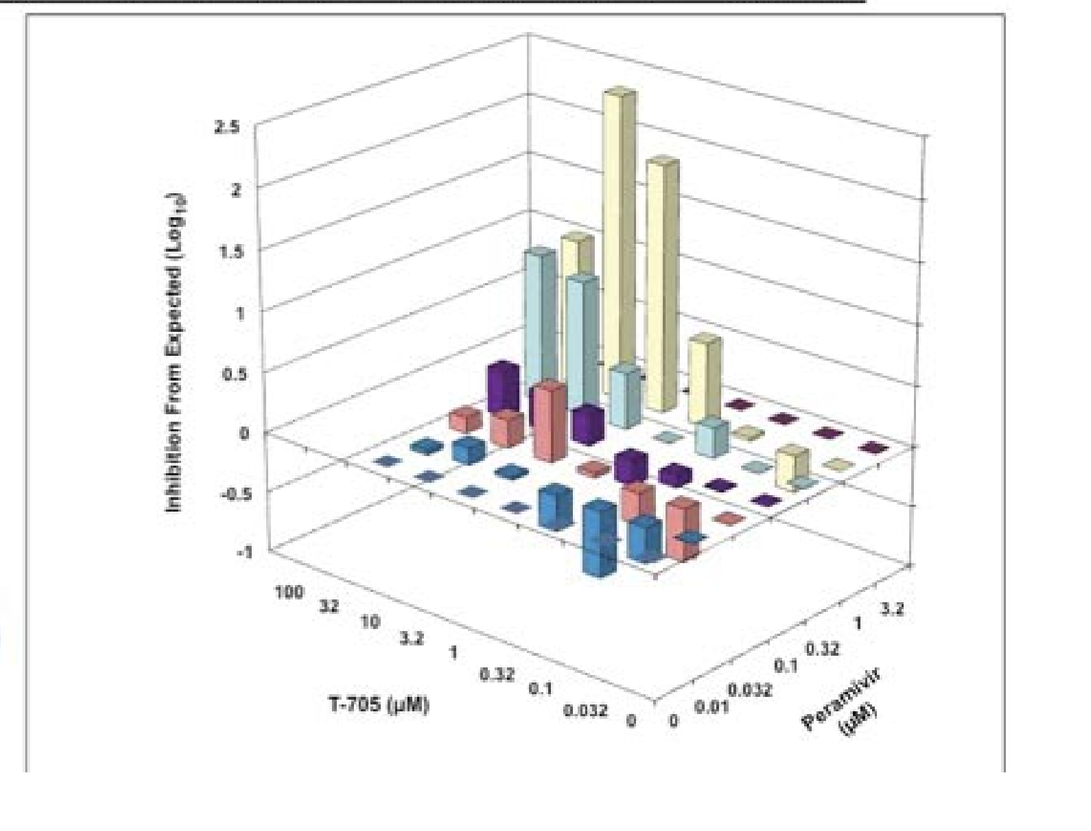
Inhibition From Expected (Log10)
T-705 (µM)
Peramivir (µM)

Figure 13: Effect of Zanamivir and T-705 on HK/2639/2009 Virus

Zanamivir (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) T-705 (µM)								
	100	32	10	3.2	1	0.32	0.1	0.032	0
3.2	0	0	0	0	0	0	0	0	0
1.0	0	0	0	0.8 ± 1.3	1.8 ± 1.6	2.7 ± 2.4	2.9 ± 2.5	2.8 ± 2.4	3.0 ± 2.6
0.32	0	0	0	2.9 ± 0.6	3.5 ± 0.3	4.5 ± 0.8	4.9 ± 0.5	4.8 ± 0.4	4.5 ± 0.3
0.1	0	0	0.9 ± 1.6	2.9 ± 0.5	3.8 ± 0	4.9 ± 0.5	5.1 ± 0.3	5.5 ± 0.3	5.0 ± 0.5
0.032	0	0	1.1 ± 1.9	3.3 ± 0.4	4.3 ± 0	5.3 ± 0.5	5.7 ± 0.1	5.9 ± 0.3	5.1 ± 0.8
0.01	0	0	1.2 ± 2.0	3.6 ± 0.8	4.5 ± 0.8	5.6 ± 0.3	5.8 ± 0	5.6 ± 0.1	5.7 ± 0.5
0	0	0	1.2 ± 2.0	4.1 ± 0.4	4.6 ± 0.8	5.7 ± 0.3	5.8 ± 0.3	6.0 ± 0.4	5.7 ± 0.6

Zanamivir EC_{90} = 0.22 µM
T-705 EC_{90} = 0.9 µM

Range of Synergy by VYR analyses:
Zanamivir (1.0 to 32 µM) and
T-705 (1.0 to 10 µM)

Synergy Plot:
Synergy = 11.64
(95% Confidence Interval = 20 - 4)
Antagonism = -3.67
(95% Confidence Interval = -1 - -6)

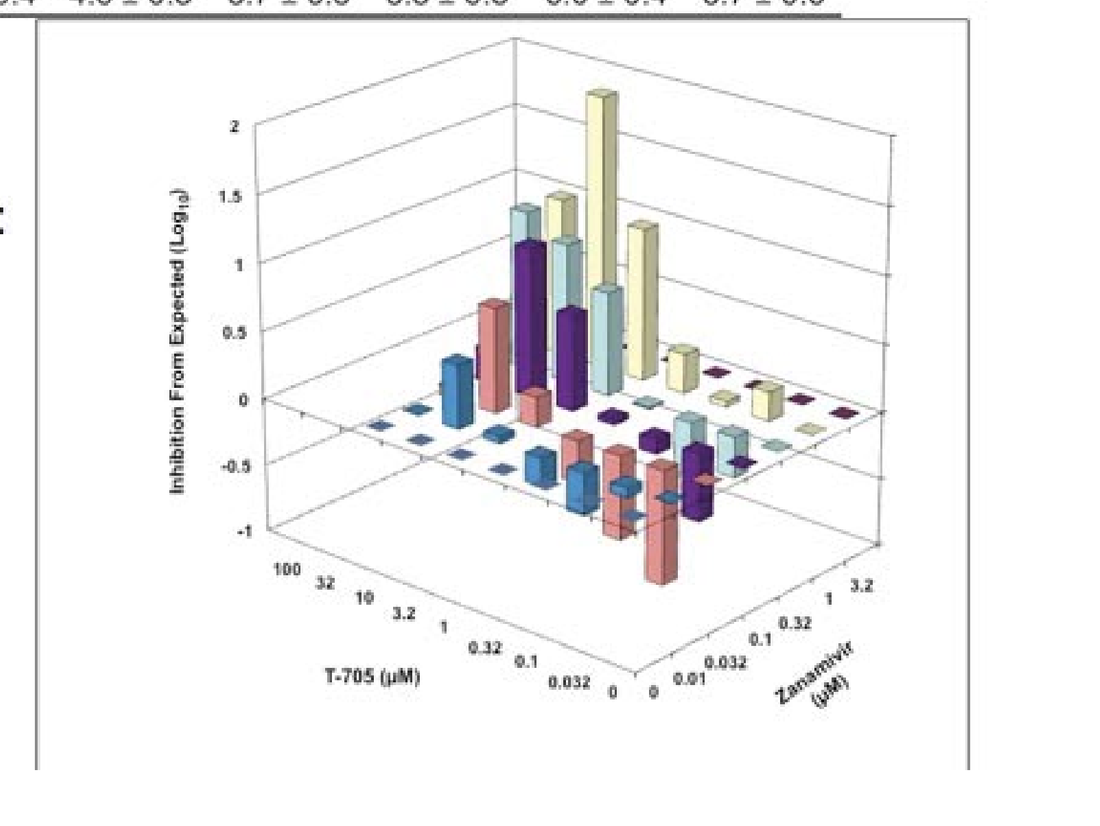
Inhibition From Expected (Log10)
T-705 (µM)
Zanamivir (µM)

Figure 14: Effect of Oseltamivir and Zanamivir on HK/2639/2009 Virus

Oseltamivir Carboxylate (µM)	Virus Titer ($\log_{10}$ $CCID_{50}$/0.1 ml) Zanamivir (µM)						
	3.2	1.0	0.32	0.1	0.032	0.01	0
32	0	1.5 ± 2.6	1.3 ± 2.2	1.2 ± 2.0	1.2 ± 2.0	1.5 ± 2.6	1.5 ± 2.6
10	0	4.1 ± 1.0	3.7 ± 0.1	4.3 ± 0.4	4.3 ± 0.5	5.1 ± 0.5	5.1 ± 0.4
3.2	1.2 ± 2.0	4.2 ± 0.8	3.8 ± 0.4	4.8 ± 0.4	4.9 ± 0.3	5.6 ± 0.3	5.1 ± 0.1
1.0	1.4 ± 2.5	4.7 ± 0.9	4.0 ± 0.4	4.9 ± 0.3	5.3 ± 0.1	5.3 ± 0.5	5.4 ± 0.1
0.32	1.3 ± 2.2	4.1 ± 0.6	4.3 ± 0.5	5.2 ± 0.6	5.1 ± 0.5	5.4 ± 0.6	5.4 ± 0.3
0.1	1.2 ± 2.0	4.2 ± 0.6	4.3 ± 0.5	5.0 ± 0.4	5.2 ± 0.6	5.7 ± 0.5	5.5 ± 0.3
0	1.6 ± 2.8	4.4 ± 0.7	4.6 ± 0.3	5.1 ± 0.5	5.4 ± 0.3	5.7 ± 0.3	5.6 ± 0.2

Oseltamivir carboxylate EC_{90} = 17.0 µM
Zanamivir EC_{90} = 0.32 µM

Range of Synergy by VYR analyses:
Oseltamivir (10 to 32 µM) and
Zanamivir (3.2 µM)

Synergy Plot:
Synergy = 9.95
(95% Confidence Interval = 17 - 3)
Antagonism = -2.24
(95% Confidence Interval = not signif.)

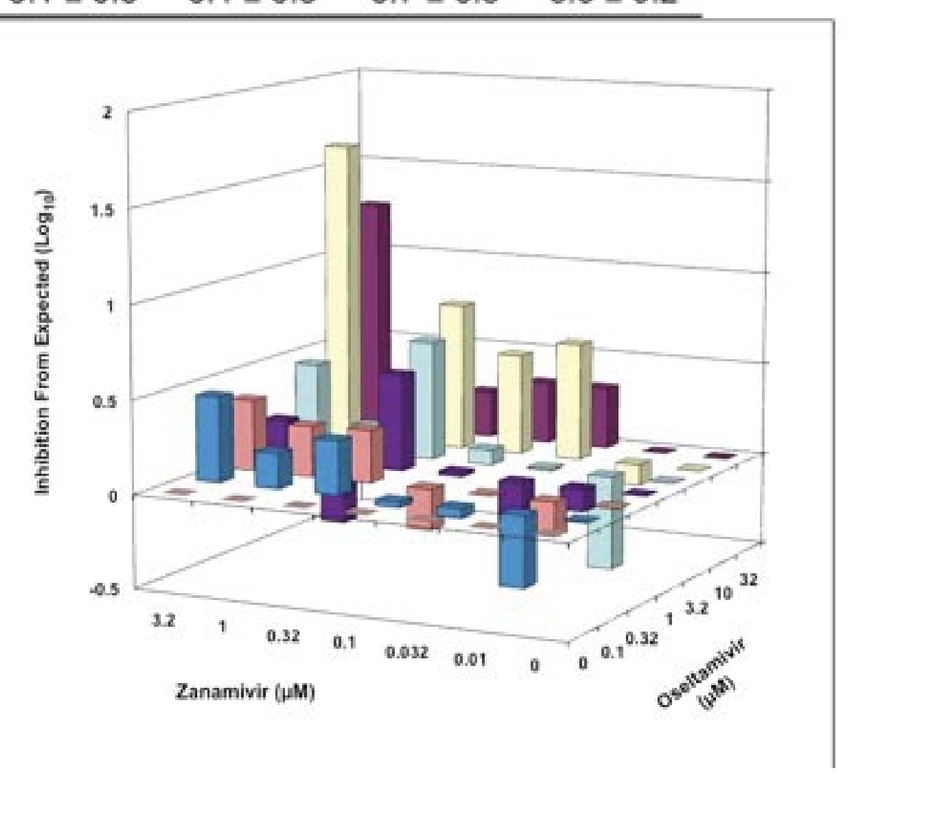
Inhibition From Expected (Log10)
Zanamivir (µM)
Oseltamivir (µM)

RESULTS

These experiments determined EC_{50} and EC_{90} values for seven antiviral drugs against nine isolates of pandemic influenza A (H1N1) virus. The antivirals tested included: Oseltamivir, Peramivir, Zanamivir, T-705 (Favipiravir), Ribavirin, Amantadine and Rimantadine.

Mean EC_{50} and EC_{90} values from three replicate experiments are shown in Table 1. These data indicate a range of sensitivities to the neuraminidase inhibitors among the H1N1 viruses tested. The mean EC_{50} values for Oseltamivir ranged from 0.2 to 1.2 µM for all viruses except the Oseltamivir-resistant A/Hong Kong/2369/2009, which had an EC_{50} of > 10 µM. Mean EC_{50} values for Peramivir ranged from 0.004 to 0.12 µM for all viruses except A/Hong Kong/2369/2009, which had a mean EC_{50} of > 10 µM. The mean EC_{50} values for Zanamivir ranged from 0.02 to 0.54 µM for all viruses except A/Hong Kong/2369/2009, which had a mean EC_{50} of 1.4 µM. The mean EC_{50} values for T-705 had a much smaller range, from 1.9 to 7.8 µM, for all viruses. Mean EC_{50} values for ribavirin ranged from 9.1 to 20.5 µM for all viruses. As expected, all pandemic H1N1 viruses tested showed resistance to the Adamantanes.

Figure 1 shows results from statistical analysis (two-way ANOVA) of the mean EC_{50} values for the antivirals tested. Significant increases (★★★$P < 0.01$ and ★★$P < 0.05$) in the EC_{50} values of the neuraminidase inhibitors were only observed against the drug-resistant A/Hong Kong/2369/2009 virus, and then only for Oseltamivir and Peramivir.

Figure 2 shows effects of mouse adaptation on the A/CA/04/2009 virus.The EC_{90} values for the mouse-adapted virus were higher for all drugs when compared to non-adapted virus.

Figures 3 through 6 show dose response curves for A/CA/07/2009 and drug-resistant A/Hong Kong/2369/2009 viruses using Oseltamivir, Peramivir, Zanamivir and T-705, respectively. The Oseltamivir dose response curves show an increase in EC_{90} from 0.32 to 14 µM (43.8-fold increase) for the drug resistant virus compared to drug sensitive virus. The EC_{90} for Peramivir increased from 0.48 to 4.5 µM (9.3-fold increase) against the drug resistant virus, while Zanamivir showed an increase from 0.4 to 2.11 µM (5.3-fold increase). T-705 showed no significant difference in EC_{90} for the two viruses as would be expected.

Results of combination drug studies are presented in Figures 7 through 10 for A/CA/07/2009 and Figures 11 through 14 for Oseltamivir-resistant A/Hong Kong/2369/2009. Tables of VYR results are represented in the upper panel of each Figure. The shaded areas in each table indicate possible regions of synergy for the combinations based on definitions described in the Materials and Methods. The lower panel of each Figure depicts 3-dimensional analyses of the VYR data using MacSynergyTM II. Both methods of analyses yielded similar results.

Figure 7 and 11 show results for the Oseltamivir plus T-705 combination. A possible region of synergy ranging over four concentrations was observed for both Oseltamivir (0.01 to 0.32 µM) and T-705 (0.32 to 10.0 µM) against A/CA/07/2009; and three concentrations for both Oseltamivir (3.2 to 32 µM) and T-705 (1.0 to 10 µM) against A/Hong Kong/2369/2009.

Figures 8 and 12 show results for the Peramivir plus T-705 combination. This combination of drugs produced a possible region of synergy ranging over six concentrations for Peramivir (0.0032 to 1.0 µM) and four concentrations for T-705 (0.32 to 10.0 µM) against A/CA/07/2009; and only two concentrations for Peramivir (0.32 to 1.0 µM) and three concentrations for T-705 (1.0 to 10 µM) against A/Hong Kong/2369/2009.

Figures 9 and 13 show results for Zanamivir plus T-705. A possible region of synergy ranging over six concentrations was observed for Zanamivir (0.0032 to 1.0 µM) and three concentrations for T-705 (0.32 to 3.2 µM) against A/CA/07/2009; and three concentrations for Zanamivir (1.0 to 32 µM) and T-705 (1.0 to 10 µM) against A/Hong Kong/2369/2009.

Figures 10 and 14 show results for Oseltamivir plus Zanamivir. The combination of these drugs produced a possible region of synergy ranging over six concentrations (0.0032 to 1.0 µM) for Oseltamivir, but only when combined with 1.0 µM Zanamivir against A/CA/07/2009. Only the highest concentrations of both drugs showed a possible region of synergy: 10 or 32 µM Oseltamivir combined with 3.2 µM Zanamivir against A/Hong Kong/2369/2009.

CONCLUSIONS

- Efficacy of antivirals determined by Neutral Red dye uptake and virus yield reduction in MDCK cells indicate a range of sensitivity to the neuraminidase inhibitors for the H1N1 viruses tested.
 - However, significantly higher EC_{50} values for the neuraminidase inhibitors were only observed against the Oseltamivir-resistant A/Hong Kong/2369/2009 virus.
 - In addition, significantly higher EC_{50} values against the A/Hong Kong/2369/2009 virus were only observed for Oseltamivir and Peramivir (not Zanamivir).
- Dose response curves indicate the largest increase in EC_{90} for Oseltamivir against the A/Hong Kong/2369/2009 virus compared to the A/CA/07/2009 virus. The EC_{90} for Zanamivir increased the least.
- Synergistic drug combinations involving multiple doses of each drug were observed when T-705 was combined with a neuraminidase inhibitor: Oseltamivir, Peramivir, or Zanamivir.
 - The greatest number of drug concentrations showing synergy was observed for the Peramivir plus T-705 combination.
- Effects of drug combinations on oseltamivir-resistant influenza A/Hong Kong/2369/2009 virus showed:
 - Synergistic combinations involving three doses of T-705 (1.0, 3.2, and 10.0 µM) for all three neuraminidase inhibitors.
 - Only Oseltamivir and Zanamivir showed synergy at three concentrations (3.2, 10, and 32 µM for Oseltamivir, or 0.1, 0.32, and 1.0 µM for Zanamivir) when combined with T-705.
 - Peramivir showed synergy at two concentrations (0.32 and 1.0 µM) in combination with T-705.
 - A minimal combining effect was observed for Oseltamivir plus Zanamivir, and then only at the highest concentrations tested (10 or 32 µM Oseltamivir combined with 3.2 µM Zanamivir).

ACKNOWLEDGEMENTS

Financial support from the Virology Branch, National Institute of Allergy and Infectious Diseases, National Institutes of Health, USA [Contract N01-AI-30048].