A Single Intramuscular Injection of Peramivir Demonstrates Anti-influenza Activity Against Recently Isolated Pandemic Flu Virus H1N1 (A/CA/04/2009) in a mouse influenza model

Shanta Bantia[a], Debra Kellogg[a], Cynthia Parker[a], Ramanda Upshaw[a], Natalia A. Ilyushina[b], Mark Prichard[c], Y.S. Babu[a]

[a]BioCryst Pharmaceuticals, Inc., Birmingham, AL, USA, [b]St. Jude Children's Research Hospital, Memphis, TN, USA, [c]University of Alabama at Birmingham, Birmingham, AL, USA

INTRODUCTION

- The importance of influenza neuraminidase (NA) in viral replication and pathogenesis has led to the development of selective inhibitors of this enzyme. Inhaled zanamivir and oral oseltamivir are effective in both prophylaxis and treatment of influenza A and B viruses.[1-3]

- Peramivir (**Figure 1**) is a specific and potent inhibitor of influenza NA and has demonstrated in vitro activity that is comparable or better than oseltamivir carboxylate and zanamivir.[4] Peramivir is active when administered IM and IV for both prophylaxis and treatment in mouse influenza virus infection models.[5,6] Intravenous administration of peramivir is currently being evaluated in clinical trials for seasonal influenza and to treat influenza in hospitalized patients.[7,8]

- In this study, the efficacy of a single IM injection of peramivir given one hour prior to, or 24, 48 and 72 hrs after, infection was evaluated in mice infected with recently isolated pandemic flu virus H1N1 A/CA/04/09 (swine origin).

Figure 1: Structure of Peramivir

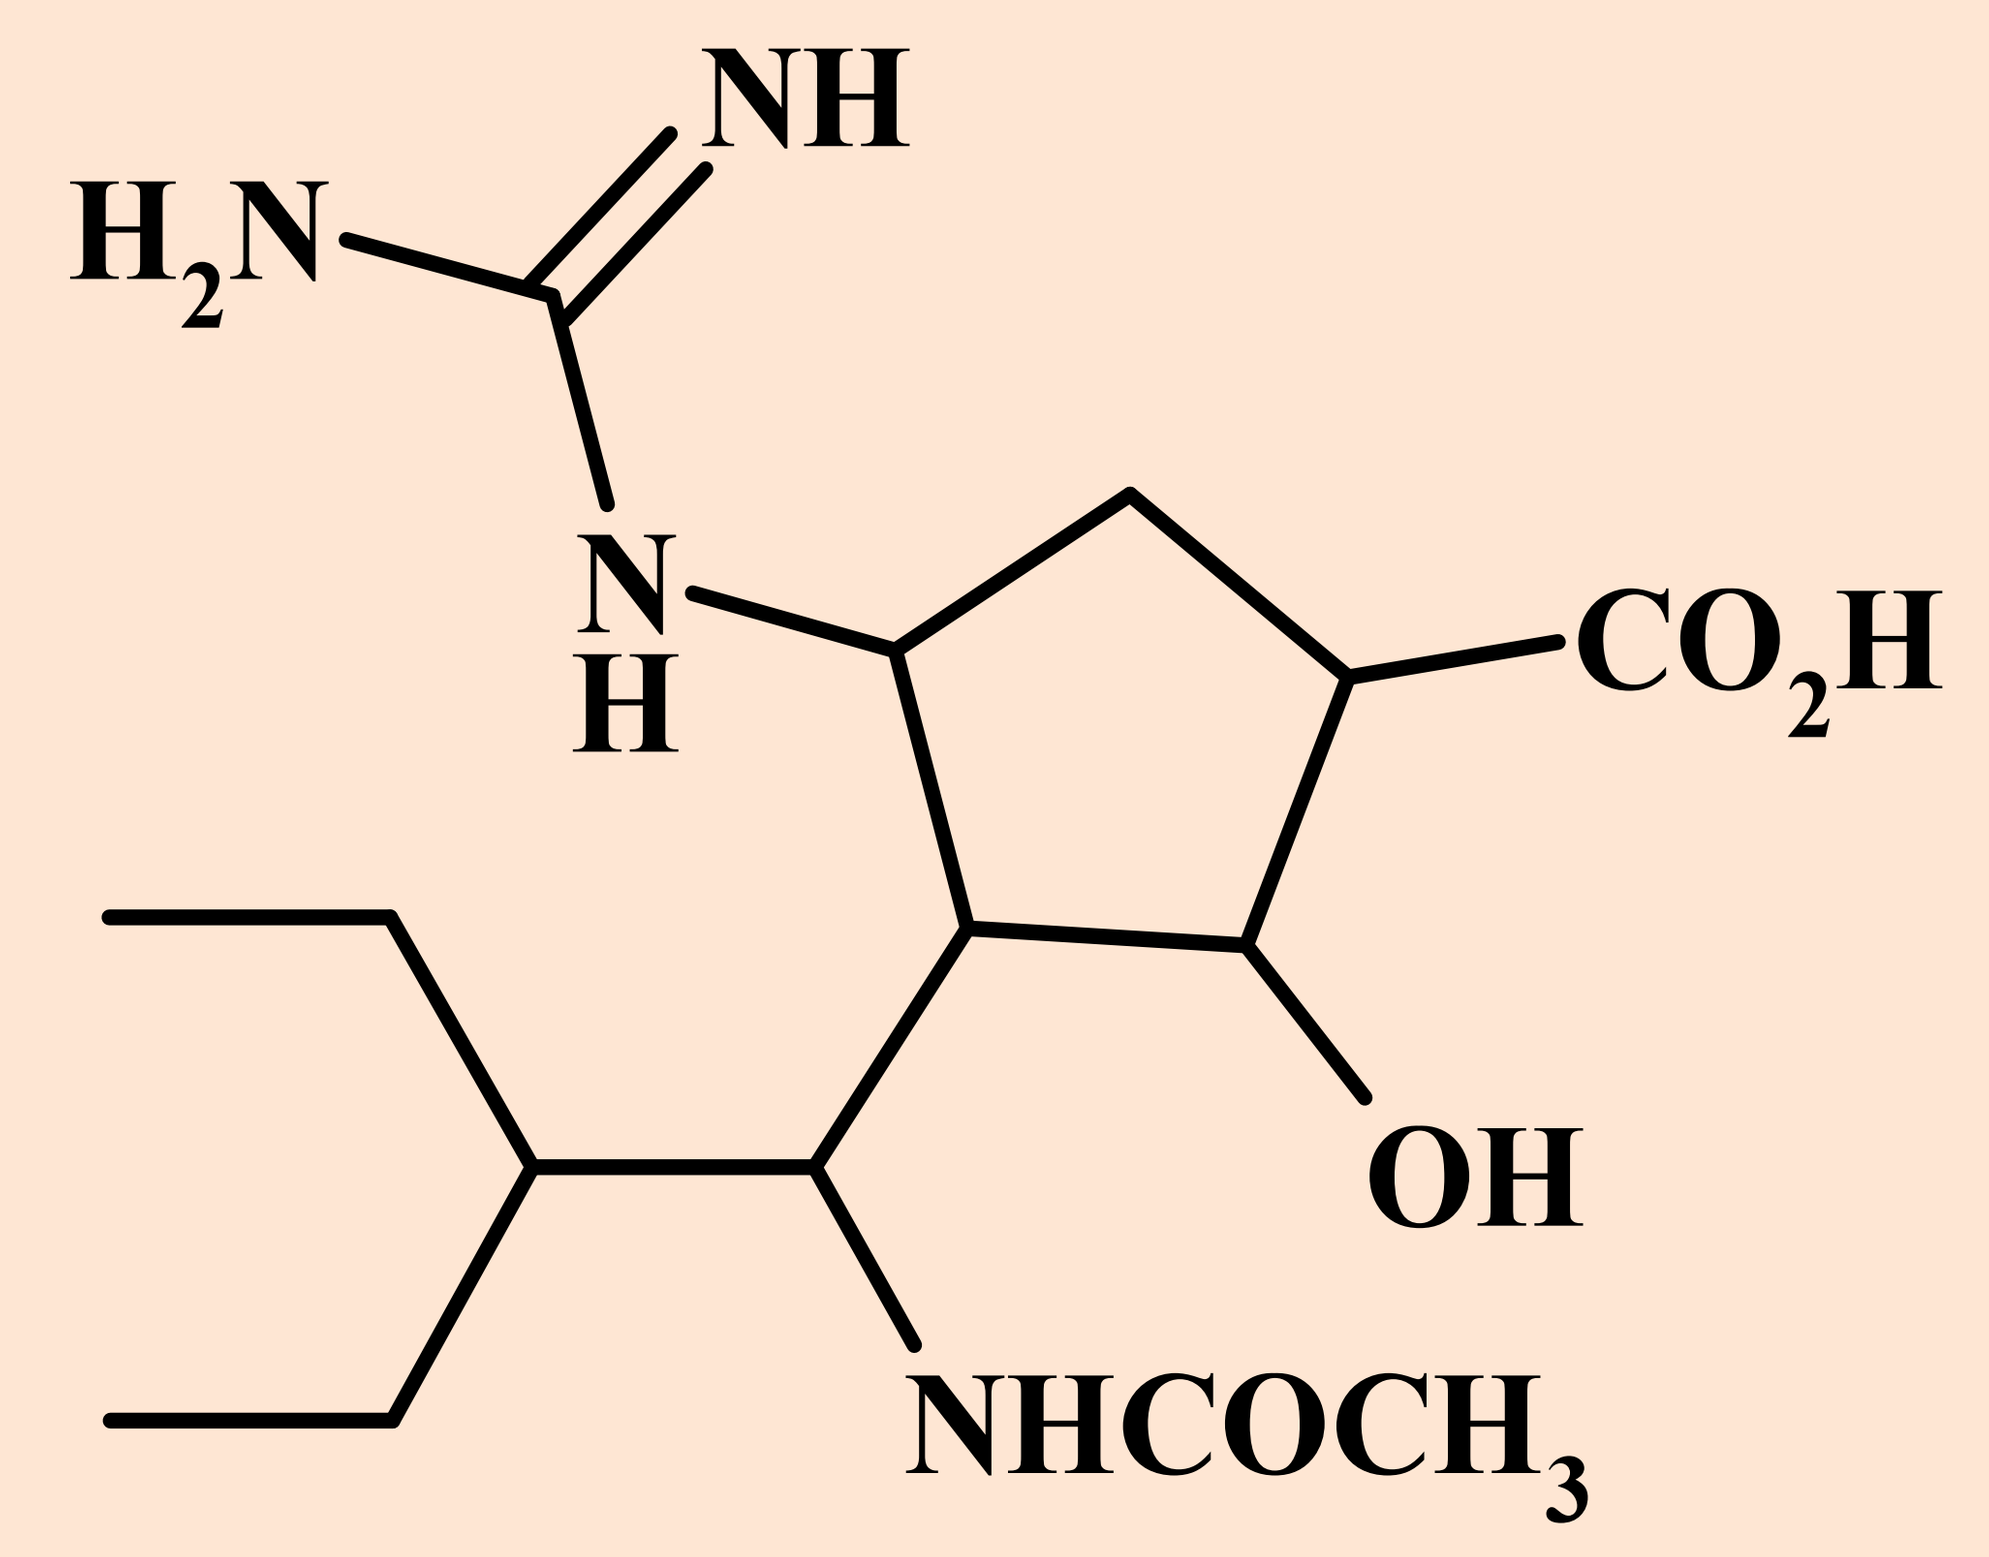
NH
H_2N
N
H
CO_2H
OH
$NHCOCH_3$

MATERIALS AND METHODS

- NA Activity: A fluorimetric assay was used to measure influenza virus NA activity.
- Animal Studies: Influenza virus: A/CA/04/09 (H1N1). Animals: Female BALB/c mice 6-8 weeks old. In the prophylaxis study peramivir was administered as a single intramuscular injection 1 hr prior to infection with virus. In the delayed treatment study peramivir was administered as single IM injection and oseltamivir was administered orally bid for 5 days starting at 24, 48 and 72 hrs after infection.
- Parameters for evaluation of antiviral activity included weight loss, reduction in mortality and/or increase in mean day to death (MDD) determined through 16-21 days.
- Statistical analysis: The t-test was used to evaluate differences in mean day to death. One way analysis of variance (ANOVA) was performed to evaluate differences in weight loss. Kaplan-Meier Survival analysis (log rank test) was applied to survival number differences.

SUMMARY

- Both peramivir and oseltamivir carboxylate are potent inhibitors of influenza A/CA/04/09 neuraminidase (**Table 1**).

- Peramivir as a single intramuscular injection given prophylactically demonstrates efficacy in mice infected with H1N1 (A/CA/04/09) influenza virus as demonstrated by survival, weight loss and mean day to death (**Table 2**).

- Peramivir as a single IM injection and Oseltamivir given orally bid for 5 days, 24 hrs post infection, demonstrates efficacy in mice infected with H1N1 (A/CA/04/09) influenza virus as demonstrated by survival and weight loss (**Table 3**).

Table 1. Inhibition of influenza A/CA/04/09 neuraminidase with Peramivir and Oseltamivir Carboxylate

Inhibitor	IC50 (nM)*
Peramivir	0.43 ± 0.07
Oseltamivir carboxylate	1.10 ± 0.19

*Mean of two independent measurements ± SEM

Table 2: Effect of a Single Intramuscular injection of Peramivir in Mice Infected with H1N1 A/CA/04/2009 Influenza virus

Treatment	Dose (mg/kg)	Survival/Total (%)	Mean day to death ± SEM	Weight change day 4[§] Mean ± SEM (gms)
Vehicle, uninfected	0	5/5 (100)	> 21	0.32 ± 0.22
Vehicle	0	3/15 (20)	5.8 ± 1.3	- 3.41 ± 0.17
Peramivir	1.0	6/10* (60)	9.5 ± 1.2*	- 2.11 ± 0.22**
Peramivir	3.0	6/10* (60)	8.5 ± 0.9*	- 1.51 ± 0.33**
Peramivir	10.0	9/10** (90)	7.0 ± 0.0*	- 1.84 ± 0.19**
Peramivir	30.0	10/10** (100)	>21*	- 1.83± 0.17**

*p<0.03, **p<0.001 vs. vehicle group;
[§]Day 4 maximum weight loss in the vehicle group with no deaths

Table 3: Effect of Delayed Treatment of Peramivir and Oseltamivir in Mice Infected with H1N1 A/CA/04/2009 Influenza virus

Treatment	Route of Admini-stration	Treatment Time Post infection	Survival / Total	Mean Day to Death ± SEM	Weight change Day 4[§] Mean ± SEM (gms)
Vehicle (uninfected)	IM	24 hr	5/5	>21 ± 0.0	0.6 ± 0.4
Vehicle	IM Single injection	24 hr	2/15	7.85 ± 0.45	-2.9 ± 0.2
Vehicle	Oral bid x 5 days	24hr	2/15	7.23 ± 0.26	-3.2 ± 0.4
Peramivir 50mg/kg	IM Single injection	24 hr 48 hr 72 hr	10/10[a] 4/10 5/10	>21 ± 0.0[a] 7.17 ± 0.48 8.0 ± 0.45	-1.7 ± 0.4[a] -3.4 ± 0.5 -3.0 ± 0.5
Oseltamivir 10mg/kg/d	Oral bid x 5 days	24 hr 48 hr 72 hr	9/10[b] 3/10 2/10	8.0 ± 0.0 7.86 ± 0.63 7.62 ± 0.59	-2.3 ±0.5[c] -2.8 ± 0.6 -3.5 ± 0.6

[a]p<0.001 vs. vehicle (IM); [b]p<0.001, [c]p<0.01 vs. vehicle (oral); [§] Maximum weight loss in the vehicle group with no deaths

CONCLUSION

- Peramivir given as a single IM injection demonstrates efficacy in mice infected with recently isolated pandemic flu virus H1N1 A/CA/04/09 (swine origin).

REFERENCES

1. Hayden FG, Atmar RL, Schilling M, Johnson C, Poretz D, Paar D, Huson L, Ward P, Mills, RG. Use of the selective oral neuraminidase inhibitor oseltamivir to prevent influenza. *N. Engl. J. Med.* 1999; 341:1336-1343.

2. Hayden FG, Osterhaus AD, Treanor JJ, et al. Efficacy and safety of the neuraminidase inhibitor zanamivir in the treatment of influenza virus infections. *N Engl J Med* 1997; 337:874-880.

3. Kaiser L, Wat C, Mills T, Mahoney P, Ward P, Hayden F. Impact of oseltamivir treatment on influenza-related lower respiratory tract complications and hospitalizations. *Arch. Intern. Med.* 2003; 163:1667-1672.

4. Bantia S, Parker CD, Ananth SL, Horn LL, Andries K, Chand P, Kotian PL, Dehghani A, El-Kattan Y, Lin T, Hutchison TL, Montgomery JA, Kellogg DL, Babu YS. Comparison of the anti-influenza virus activity of RWJ-270201 with those of oseltamivir and zanamivir. *Antimicrob Agents Chemother*. 2001; 45: 1162-1167.

5. Bantia S, Arnold CS, Parker CD, Upshaw R, Pooran C. Anti influenza virus activity of peramivir in mice with single intramuscular injection. *Antiviral Research* 2006; 69(1):39-45.

6. Kobayashi M, Isoda N, Kodama M, Yoshida R, Sato A, Yamano Y, Sakoda Y, Kida H. Inhibitory Effect of Peramivir (S-021812, BCX-1812) against Highly Pathogenic Avian Influenza Viruses. 49th Interscience Conference on Antimicrobial Agents and Chemotherapy (*ICAAC* 2009). San Francisco. September 12-15, 2009. Abstract V-1064.

7. Kohno S, Yen MY, Cheong HJ, and others. V-537a - Single-Intravenous Peramivir vs. Oral Oseltamivir to Treat Acute, Uncomplicated Influenza in the Outpatient Setting: A Phase III Randomized, Double-Blind Trial. (*ICAAC* 2009). Abstract V-537a.

8. Ison MG, McGeer AJ, Hui DS, Clezy K, O'Neil B, Flynt A, Elder J, Simon TJ, Alexander WJ. Safety and Efficacy of Multiple-Day Treatment with Intravenous Peramivir or Oral Oseltamivir in Hospitalized Adults with Acute Influenza. XI International Symposium on Respiratory Viral Infections. 2009. Oral abstract.