A Phase II study of the purine nucleoside phosphorylase (PNP) inhibitor RO5092888 (BCX-4208) in patients with moderate to severe chronic plaque psoriasis: safety, tolerability and lymphocyte effects.

Joseph Gomes, PharmD,[1] Angela Georgy PharmD,[1] Sharon Passe, MS,[1] Shanta Bantia PhD,[2] Adrienne Farid PhD,[1] Gerhard Wolff MD,[1]

[1]Hoffmann-La Roche, Inc., Nutley, NJ; [2]BioCryst Pharmaceuticals, Inc., Birmingham, AL.

Abstract

PNP is a purine-metabolizing enzyme that catalyzes the phosphorolysis of 2'-deoxyguanosine [dGuo] to guanine and deoxyribose-1-phosphate. In T-cells, PNP inhibition leads to accumulation of deoxyguanosine triphosphate (dGTP), triggering apoptotic cell death. Chronic-plaque type psoriasis (psoriasis) is an autoimmune disorder, in which T-cells contribute, at least in part, to the manifestations and maintenance of the disease. Therefore, targeting T-cells may be a beneficial treatment strategy. Since for chronic inflammatory diseases the safety of potential immunosuppressive drugs is a major consideration, we conducted this study to investigate safety and tolerability of oral RO5092888 (BCX-4208) in patients with moderate to severe chronic plaque psoriasis.

This was a randomized, double-blind, placebo-controlled, dose-ranging study. Sixty six patients 18 to 70 years old were randomized into one of three groups: placebo, 20mg or 120mg. The study was conducted from August 2007 to June 2008. Patients received study drug over six weeks and were observed over an additional four weeks. Assessments for safety included tracking of adverse events (AEs) including infections; vital signs; ECGs; chemistry panel; LFTs; hematologic parameters including peripheral blood (PB) lymphocyte subsets $CD3^+$, $CD4^+$, $CD8^+$, $CD56^+$, $CD20^+$; and urinalysis.

Results: 65 of the 66 enrolled patients were analyzed. One serious AE was observed in the 20mg/d group, a deep vein thrombosis (DVT) in a patient with a history of DVTs, and was considered unrelated to study drug. The percentage of patients experiencing at least one adverse event (AE) of any grade was: placebo: 33% (7/21); 20mg: 41% (9/22) and 120mg: 59% (13/22). During treatment, three infections occurred in the placebo group (upper respiratory tract, influenza and sinusitis), one in the 20mg group (nasopharyngitis) and 4 in the 120mg group (2-upper respiratory tract infections, one bronchitis and one otitis externa). Reductions in PB lymphocytes and subsets were observed (Table 1). Nine patients showed decreased levels of CD4+ lymphocytes, below 350 cells/µL (n=20mg, 3; 120mg, n=6).

Conclusion: Daily oral administration of 20 mg or 120 mg of RO5092888 for up to 6 weeks demonstrated adequate safety and tolerability. Reductions in PB T cells, T cell subsets and B cells were observed (Table 1). Further investigation of RO5092888 is warranted in both T-cell and B-cell diseases.

Table1. Lymphocyte subpopulation	Mean Nadir (% change from baseline)		
	Placebo (n=21) Percent (SD)	20 mg (n=22) Percent (SD)	120 mg (n=22) Percent (SD)
CD3+	20.4 (17.1)	28.7 (17.0)	49.0 (16.3)
CD4+	20.4 (16.8)	27.4 (15.6)	45.7 (17.0)
CD8+	21.8 (19.6)	31.5 (17.9)	55.8 (15.8)
CD20+	24.9 (32.0)	36.5 (17.4)	63.3 (16.3)
CD56+	29.0 (27.5)	40.6 (29.7)	73.4 (15.2)

Introduction

• Low T cell numbers and defective T cell function characterized by severely compromised T cell responses to mitogenic stimuli have been reported in children born with PNP deficiency. Approximately one-third of patients have, in addition, defective B cell function[1]. This rare clinical condition therefore provides the rationale for the development of specific inhibitors of PNP and their evaluation in T-cell mediated autoimmune disease like psoriasis.

• RO509288 (BCX-4208; Figure 2) is a potent inhibitor of PNP and demonstrated in vitro inhibition of proliferation of lymphocytes and apoptosis in T and B-lymphocyte subpopulations (See Abstract # 7315).

• Preclinical pharmacokinetic (PK) and pharmacodynamic (PD) data demonstrated that RO5092888 increases plasma dGuo (the desired metabolic profile) to the pharmacologic levels required for inhibition of T cell proliferation.

•This is the first study to assess safety of RO5092888 in patients with moderate to severe plaque psoriasis.

Figure 1: PNP Pathway

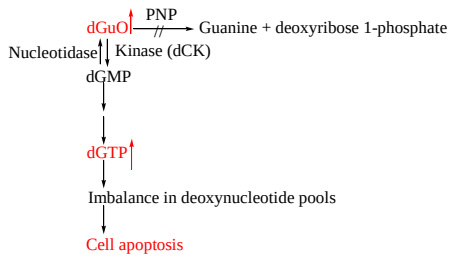
dGuO
PNP
Guanine + deoxyribose 1-phosphate
Nucleotidase
Kinase (dCK)
dGMP
dGTP
Imbalance in deoxynucleotide pools
Cell apoptosis

Figure 2: Structure of RO5092888 (BCX-4208):

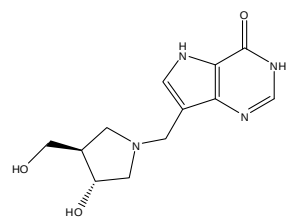
O
H
N
NH
N
N
HO
HO

Materials and Methods

This was a randomized, double-blind, placebo-controlled, dose-ranging study. Patients with medically stable moderate to severe chronic plaque psoriasis were randomized (1:1:1) to receive either placebo or one of two doses of RO5092888 administered as an oral QD (*quaque die*, once a day) regimen (Table 2). The study was conducted from August 2007 to June 2008. Patients received study drug over six weeks and were observed over an additional four weeks.

Group	Dosing Regimen	Number of Patients
1	20 mg QD x 6 wk	21
2	120 mg QD x 6 wk	22
3	Placebo QD x 6 wk	22

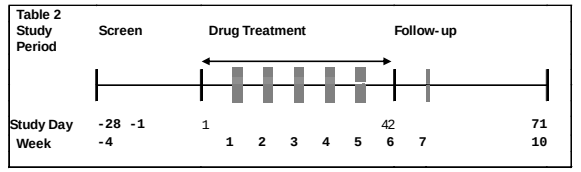
Table 2
Study
Period
Screen
Drug Treatment
Follow-up
Study Day
-28 -1
1
42
71
Week
-4
1 2 3 4 5 6 7
10

The primary analysis for safety occurred after all patients completed 10 weeks of treatment. The safety analysis population included all patients who received at least one dose and had a safety assessment performed post baseline. Assessments for safety included tracking of adverse events (AEs) including infections; vital signs; ECGs; chemistry panel; LFTs; hematologic parameters including peripheral blood (PB) lymphocyte subsets $CD3^+$, $CD4^+$, $CD8^+$, $CD56^+$, $CD20^+$; and urinalysis.

Inclusion Criteria

• Men or women between the ages of 18 and 70 years.

• Normal Glomerular Filtration Rate (GFR) (> 80 mL/min) estimated by creatinine clearance as calculated by the Cockcroft-Gault formula.

• Medically stable, clinically active moderate to severe chronic plaque psoriasis with body surface area involvement > 10%, with minimum PASI score of 10 at the time of screening and who have had psoriasis for more than 6 months.

• Females of non child-bearing potential and males must agree to use contraception, along with their partners.

Exclusion Criteria

• Erythrodermic, guttate, palmar, plantar pustular or generalized pustular psoriasis at the time of screening or any skin condition that may interfere with evaluation of the effects of the study medication on plaque lesions.

• Current or history of psoriatic arthritis

• CD4+ cells counts by flow cytometry and absolute lymphocyte counts outside normal range.

• Active or latent tuberculosis (TB) infection. These are to be ruled out with a Purified Protein Derivative (PPD) skin test plus a chest X-ray.

• Serious local or systemic bacterial, fungal, and/or viral infection (e.g., pneumonia, septicemia) within 3 months of receiving study medication.

• PCR-positivity for HBV-DNA, HCV-RNA, HIV-RNA, CMV-DNA.

• Treatment with the following drugs before receiving study medication: biologics within 3 months or 5x its half-life (whichever is shorter), systemic retinoids within 4 weeks; systemic immunosuppressants (e.g. methotrexate, cyclosporine) and phototherapy within 4 weeks; topical treatment for psoriasis within 2 weeks.

• Treatment with systemic antibiotics, anti-fungals or anti-infectives within 2 weeks before receiving study medication.

Patient Stopping Rule

• **CD4+ Cell Count**

• If CD4+ flow cytometer cell count values fall < 350 cells/mm^3, then stop study medication immediately, withdraw from the study and follow-up until resolution.

Results

• Sixty five of the 66 enrolled patients were analyzed. Treatment arms were well balanced with respect to all demographic characteristics (Table 3)

• One serious AE was observed in the 20mg group, a deep vein thrombosis (DVT) in a patient with a history of DVTs, and was considered unrelated to study drug. The percentage of patients experiencing at least one adverse event (AE) of any grade was: placebo: 33% (7/21); 20mg: 41% (9/22) and 120mg: 59% (13/22). The most common adverse events in the study were infection, low CD4+ count, anemia and diarrhea (Table 4). During treatment, three infections occurred in the placebo group (upper respiratory tract, influenza and sinusitis), one in the 20mg group (nasopharyngitis) and 4 in the 120mg group (2-upper respiratory tract infections, one bronchitis and one otitis externa).

• Reductions over time in PB lymphocytes and subsets were observed (Figures 3 -7). Nine patients showed decreased levels of CD4+ lymphocytes, below 350 cells/µL (20mg,n= 3; 120mg, n=6). Rates of treatment completion were slightly lower in the 20mg and 120mg group (Table 5).

•No evidence of clinical efficacy, a secondary objective, was observed in psoriasis patients with the doses and duration of administration tested.

Table 3. Baseline Demographics and Disease Characteristics

Demographic	Placebo N = 21	RO5092888 20mg N = 22	RO5092888 120mg N = 22
Age – yr			
Mean	43.2	41.4	46.6
Min-Max	28 – 57	18 – 70	22 - 66
Sex			
Male	3(14)	5(23)	2(9)
Female	18(86)	17(77)	20(91)
Race/Ethnic Group			
White	18(72)	19(68)	19(59)
Black	1(5)	2(9)	2(9)
Hispanic	3(4)	4(18)	6(27)
Asian	1(5)	0	0
American Indian	0	1(5)	0
Other	1(5)	0	0
Duration of Psoriasis – yr			
Mean	16.0	17.6	20.0
Range	4 – 30	2 – 40	4 - 60
PASI Score			
Mean	17.8	16.65	16.98
Range	9.9 – 43.5	10.1 – 45.2	10.7 – 36.5
PGA			
Mean	3.4	3.6	3.4
Range	2 – 6	2 – 6	2 - 6
TLS			
Mean	7.5	7.9	7.8
Range	5 – 12	3 – 10	4 - 10

Table 4. Adverse Events reported in greater than 5% of Patients in any group

Adverse Event	Placebo N = 21 No. (%)	RO5092888 20mg N = 22 No. (%)	RO5092888 120mg N = 22 No. (%)
Infection	3 (14)	1(5)	4 (18)
CD4+ cell count < 350	0	3 (14)	6 (27)
Anemia	0	1 (5)	2 (9)
Diarrhea	0	0	2 (9)

Table 5. Rates of Treatment Completion and Reasons for Not Completing Treatment

Reason	Placebo N = 21 No. (%)	RO5092888 20mg N = 22 No. (%)	RO5092888 120mg N = 22 No. (%)
Completion of treatment	18 (86)	16 (73)	16 (73)
Reasons not completing treatment	3 (14)	6 (27)	6 (27)
AE/Illness	0	4 (18)	4 (18)
Insufficient Response	2(10)	1 (5)	2 (9)
Refuse Treatment	1 (5)	1 (5)	0

Figure 3: CD4 (10**9/L) Mean Values

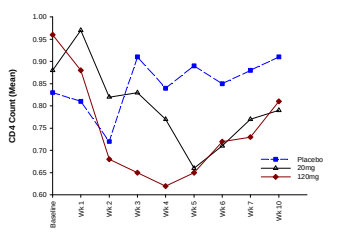
CD4 Count (Mean)
Baseline
Wk 1
Wk 2
Wk 3
Wk 4
Wk 5
Wk 6
Wk 7
Wk 10
Placebo
20mg
120mg

Figure 4: CD8 (10**9/L) Mean Values

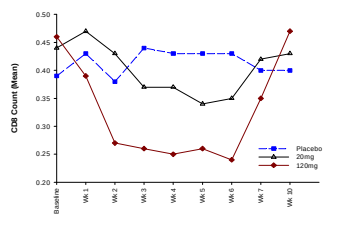
CD8 Count (Mean)
Placebo
20mg
120mg

Figure 5: CD3 (10**9/L) Mean Values

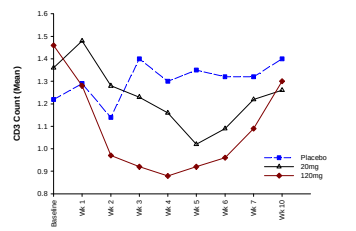
CD3 Count (Mean)
Placebo
20mg
120mg

Figure 6: CD16/CD56 (10**9/L) Mean Values

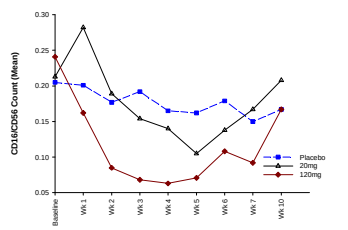
CD16/CD56 Count (Mean)
Placebo
20mg
120mg

Figure 7: CD20 (10**9/L) Mean Values

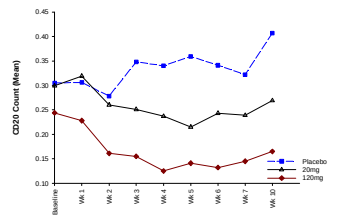
CD20 Count (Mean)
Placebo
20mg
120mg

Conclusions

Daily oral administration of 20 mg or 120 mg of RO5092888 for up to 6 weeks demonstrated adequate safety and tolerability. As expected from the phenotype of PNP-deficient patients, dose-dependent reduction of T cell numbers were observed. Pharmacological inhibition of PNP activity with BCX-4208 was however shown to dose-dependently affect other immune cell populations such as B cells and Natural Killer cells. These findings provide the rationale for investigating further the use of RO5092888 in T and B-cell mediated diseases

References

1. Markert ML. Purine nucleoside phosphorylase deficiency. Immunodefic Rev 1991;3:45-81.
2. Sircar JC and Gilbertsen RB. Purine nucleoside phosphorylase (PNP) inhibitors: Potentially selective immunosuppressive agents. Drugs of the Future 1988;13:653–668.
3. Kazmers IS, Mitchell BS, DaDonna PE, Wotring II, Townsend LB, et al. Inhibition of purine nucleoside phosphorylase by 8 aminoguanosine: selective toxicity for T lymphoblasts. Science 1981;214:1137–1139.
4. Bantia S, Miller PJ, Parker CD, Ananth SL, Horn LL, et al. Purine nucleoside phosphorylase inhibitor BCX 1777 (Immucillin H)—a novel potent and orally active immunosuppressive agent. Int Immunopharmacol 2001;1:1199 1210.
5. Bantia S, Miller PJ, Parker CD, Ananth SL, Horn LL, et al. Comparison of in vivo efficacy of BCX 1777 and cyclosporin in xenogeneic graft-vs.-host disease: The role of dGTP in antiproliferative action of BCX 1777. Int Immunopharmacol 2002;2:913 923.

Disclosures: Gomes: *Hoffmann-La Roche, Inc:* Employment. **Georgy:** *Hoffmann-La Roche, Inc.:* Employment. **Passe:** *Hoffmann-La Roche, Inc.:* Employment . **Farid:** *Hoffmann-La Roche, Inc.:* Employment. **Bantia:** *BioCryst Pharmaceuticals, Inc.:* Employment. **Wolff:** *Hoffmann-La Roche, Inc.:* Employment